Strong Synergistic Activity of an EZH1/2 Dual Inhibitor, HM97662, in Combination with Standard Chemotherapy in Preclinical Models of Advanced Solid Tumors

#1681

Seung Hyun Jung, Seon Yeong Han, Gunwoo Lee, Aran Park, Jooyun Byun, and Young Gil Ahn
Hanmi Pharmaceutical Co., Ltd., Seoul, Republic of Korea

Introduction

Many cancer patients eventually develop resistance to chemotherapy drugs after treatment. For instance, platinum-based drugs are commonly used as first-line treatments for ovarian cancer. While many patients initially respond well to this therapy, approximately 80% experience tumor recurrence within two years and ultimately acquire platinum-resistance. Recent studies suggest that EZH2 plays a key role in regulating cisplatin resistance in ovarian cancer.[1)]

Similarly, small cell lung cancer (SCLC) is an aggressive malignancy with a poor prognosis, particularly in relapsed or refractory cases. Topoisomerase 1 inhibitors are commonly used as a second-line therapy due to its manageable toxicity and efficacy. however, its effectiveness as a monotherapy is limited by chemotherapy resistance. Preclinical studies indicate that combining EZH2 inhibitors with chemotherapy may enhance anticancer efficacy by modulating the epigenetic environment and disrupting DNA repair mechanisms. This combination strategy holds promise for overcoming resistance and improving outcomes in SCLC and other cancers.[2)]

EZH2, an enzymatic subunit of the polycomb repressive complex 2 (PRC2), catalyzes the tri-methylation of histone 3 at lysine 27 (H3K27me3), repressing target genes involved in cell cycle regulation, differentiation, and tumor suppression.[3)] PRC2 and SWI/SNF have opposing roles in chromatin regulation, and EZH2 overexpression is linked to SWI/SNF mutations. This interaction affects cancer cell survival, highlighting EZH2 as a critical therapeutic target. Dual inhibition of EZH1 and EZH2 may offer greater anti-tumor efficacy, as EZH1 can compensate for the loss of EZH2 activity.

Here, we propose the anti-tumor potency of HM97662, an EZH1/2 dual inhibitor, in diverse solid cancer cell lines. We further evaluated its potential in combination with standard-of-care chemotherapy in preclinical models, including an advanced solid tumor xenograft mouse model.

Combined Effects of the EZH1/2 Inhibitor, HM97662

Dual Inhibition of EZH1/2 Combined with Chemotherapy for Cancer Treatment

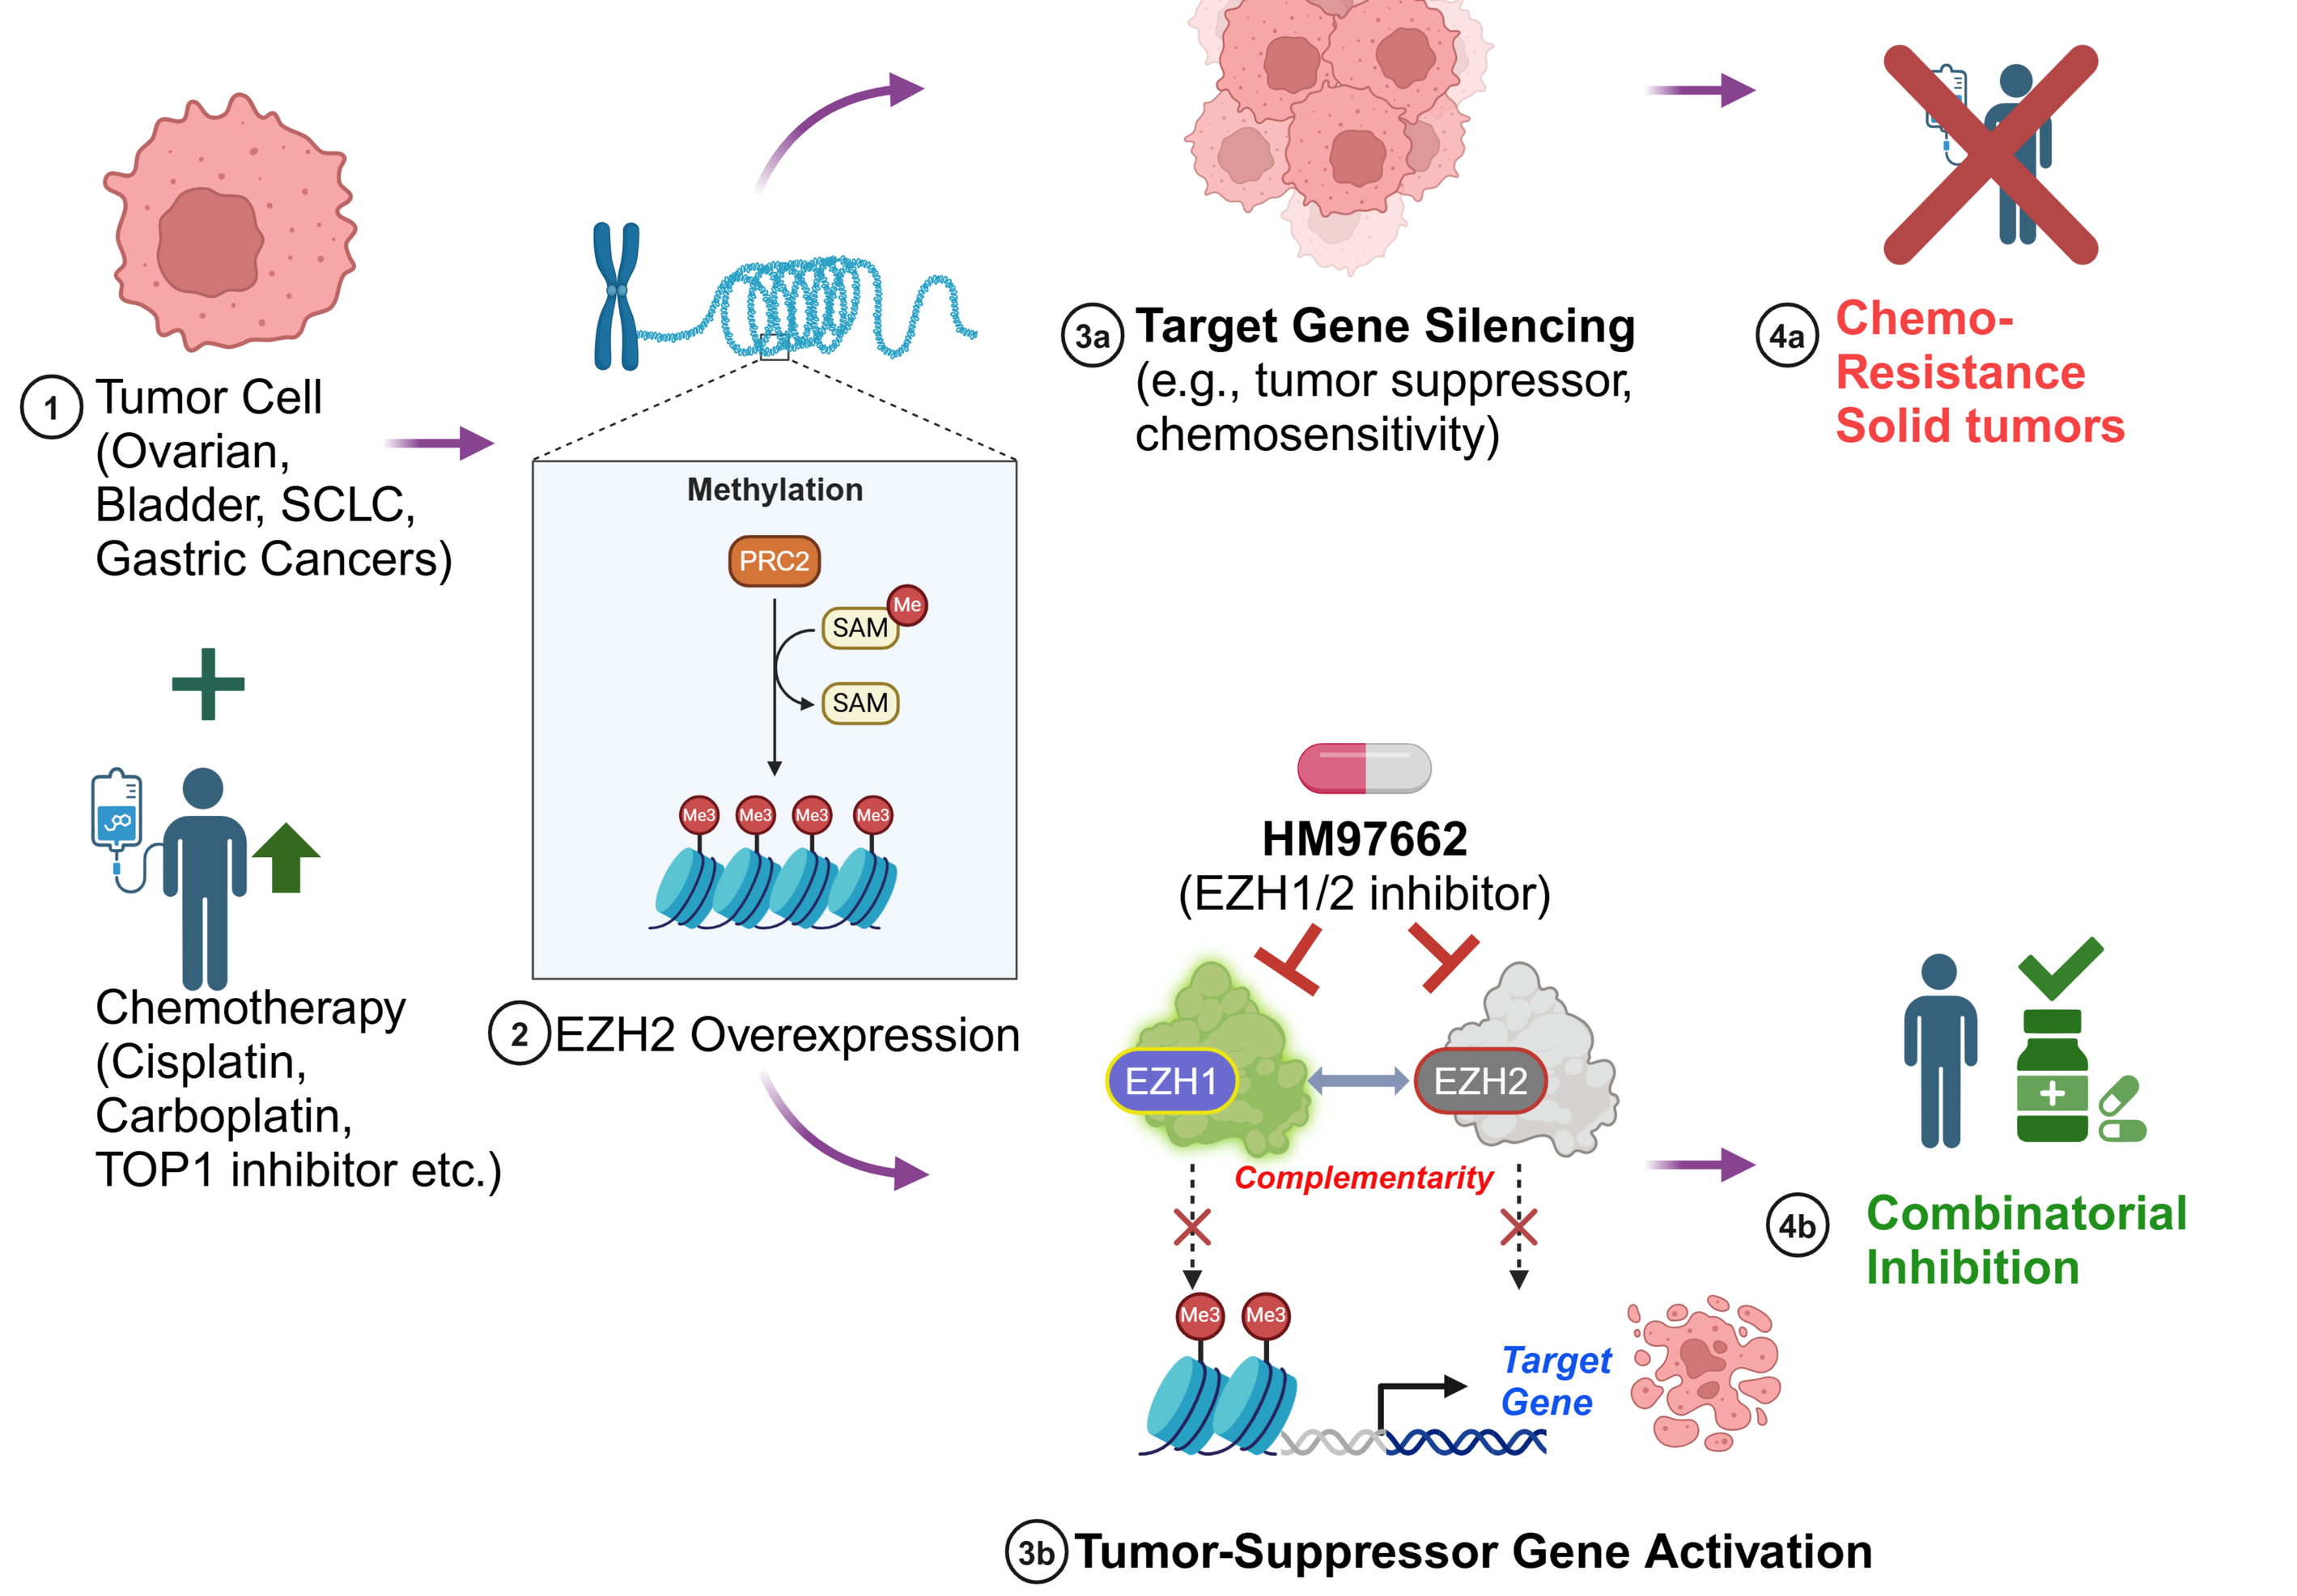
1 Tumor Cell (Ovarian, Bladder, SCLC, Gastric Cancers)
Chemotherapy (Cisplatin, Carboplatin, TOP1 inhibitor etc.)
Methylation
PRC2
SAM
Me
Me3
2 EZH2 Overexpression
3a Target Gene Silencing (e.g., tumor suppressor, chemosensitivity)
4a Chemo-Resistance Solid tumors
HM97662 (EZH1/2 inhibitor)
EZH1
EZH2
Complementarity
Target Gene
3b Tumor-Suppressor Gene Activation
4b Combinatorial Inhibition

Schematic illustration was created with BioRender.com.

Enhanced Inhibitory Activity Toward EZH1

A. Biochemical activity of EZH1/2

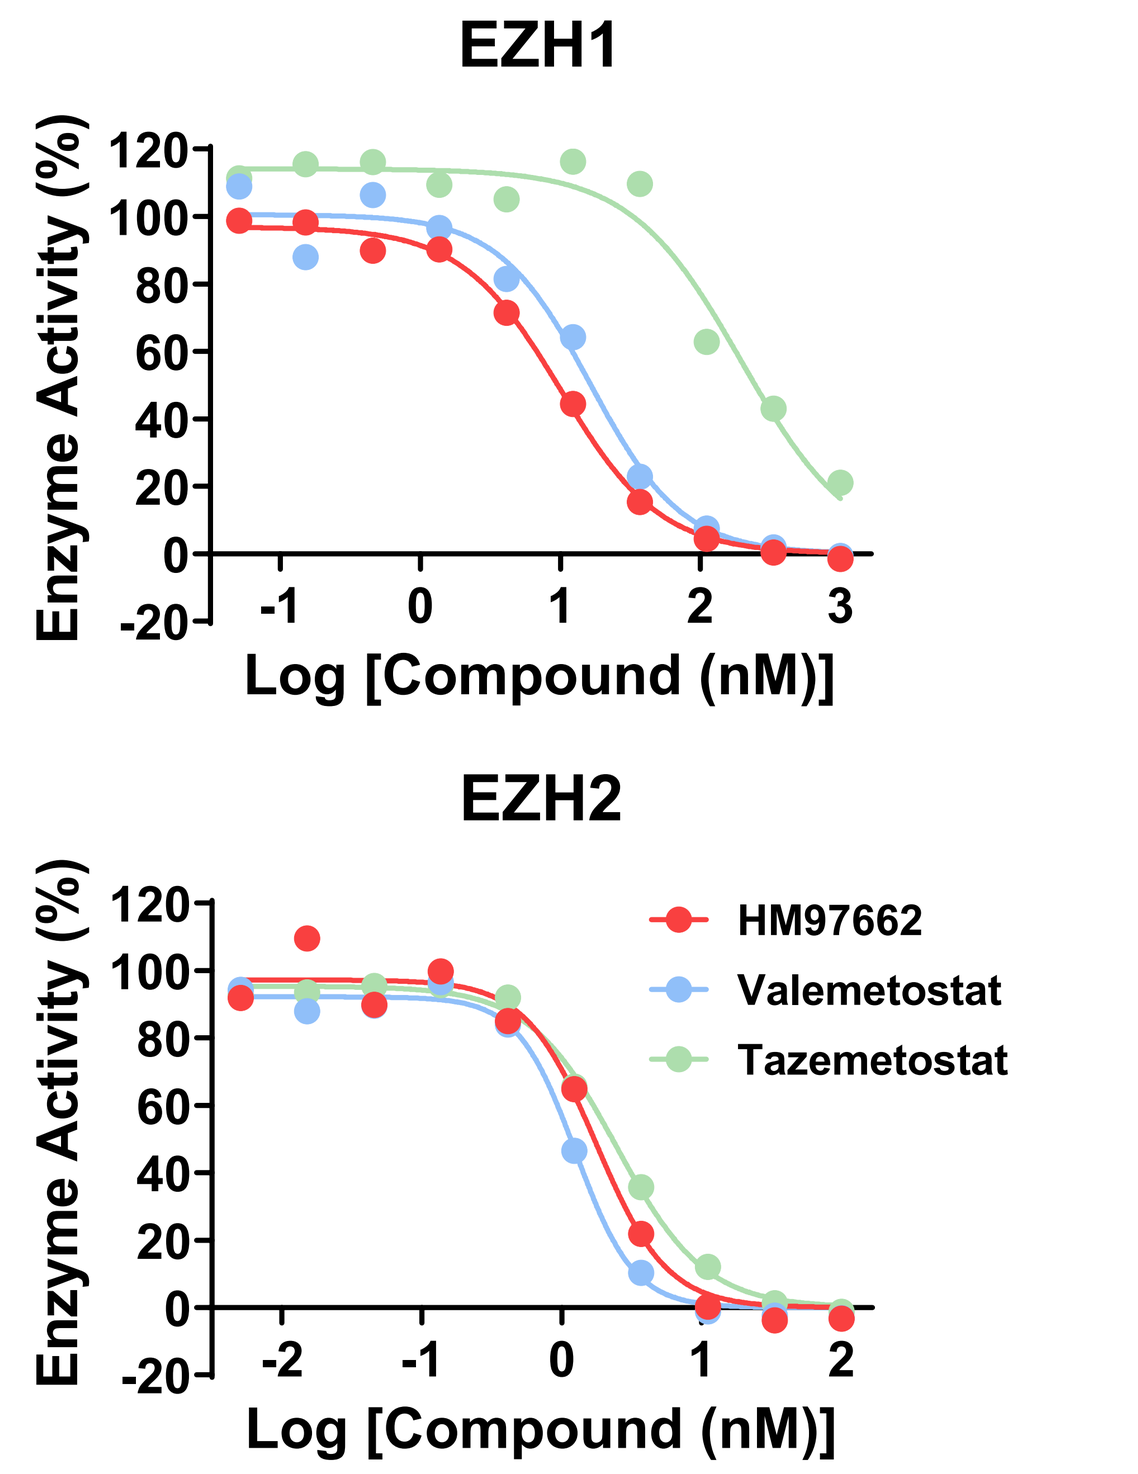
EZH1
EZH2
Enzyme Activity (%)
Log [Compound (nM)]
HM97662
Valemetostat
Tazemetostat

B. Comparison of EZH1 and EZH2 inhibition profiles with competitors

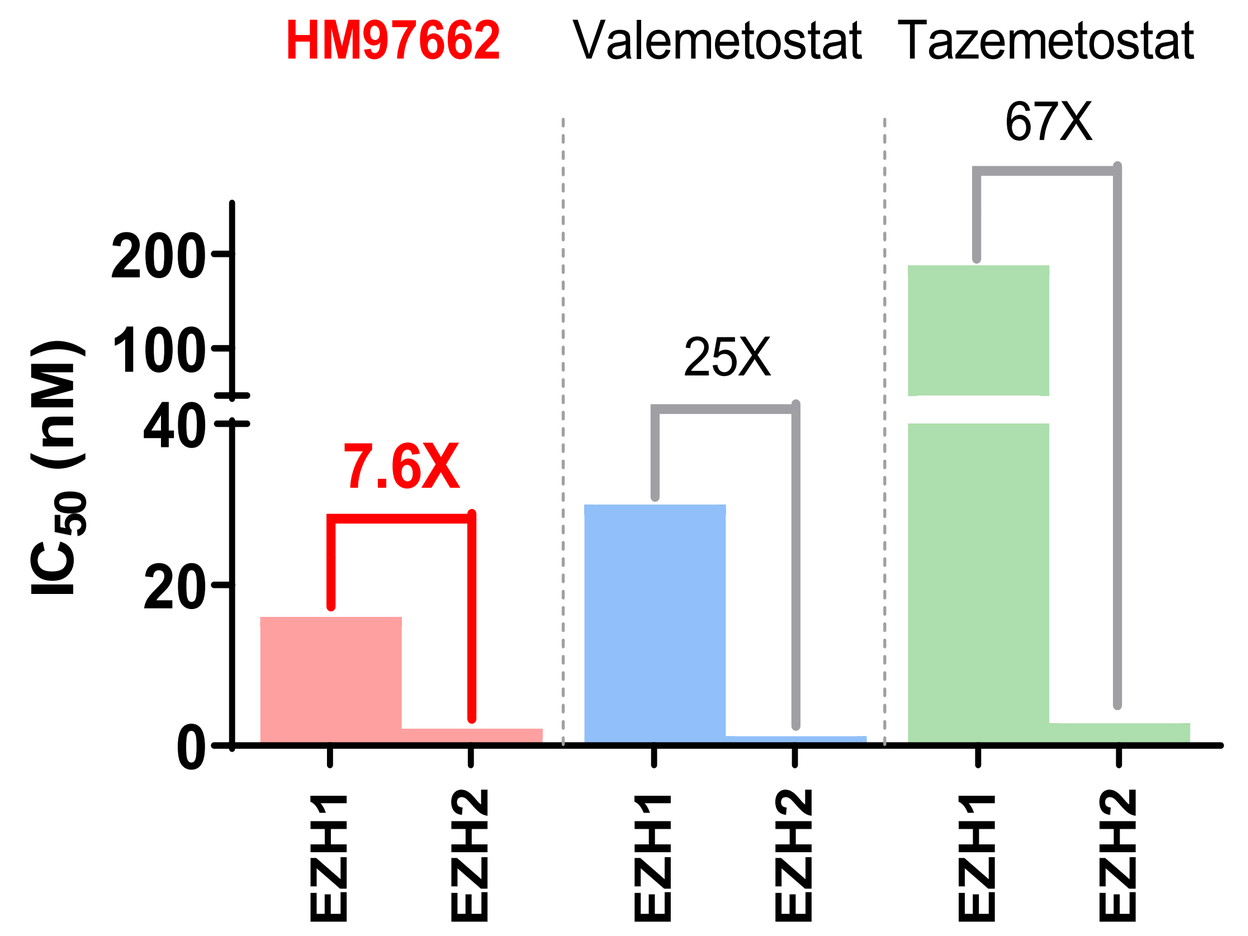
HM97662
Valemetostat
Tazemetostat
7.6X
25X
67X
IC50 (nM)
EZH1
EZH2

Anti-proliferative Effects

A. Biomarker-positive cancer cell lines

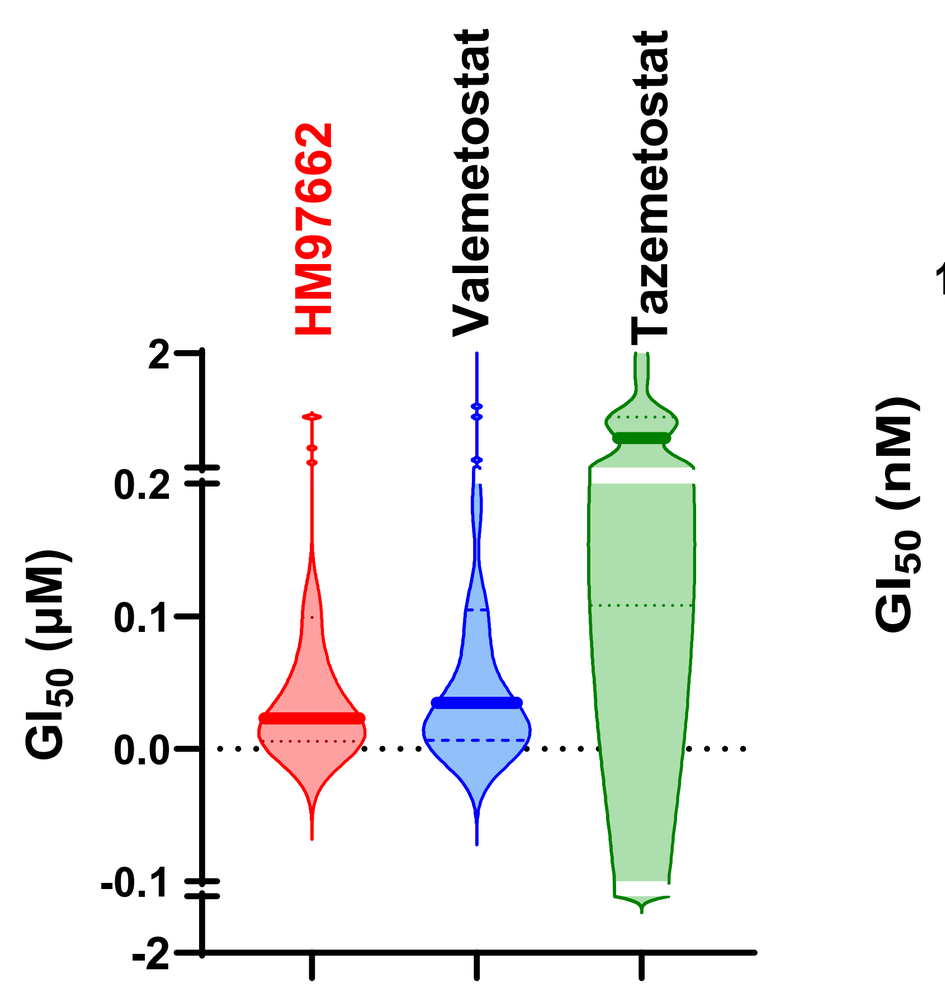
HM97662
Valemetostat
Tazemetostat
GI50 (μM)

Biomarker: EZH2 mutation, SWI/SNF mutation, ARID1A LOF etc.

B. SWI/SNF-mutated solid cancer cell lines

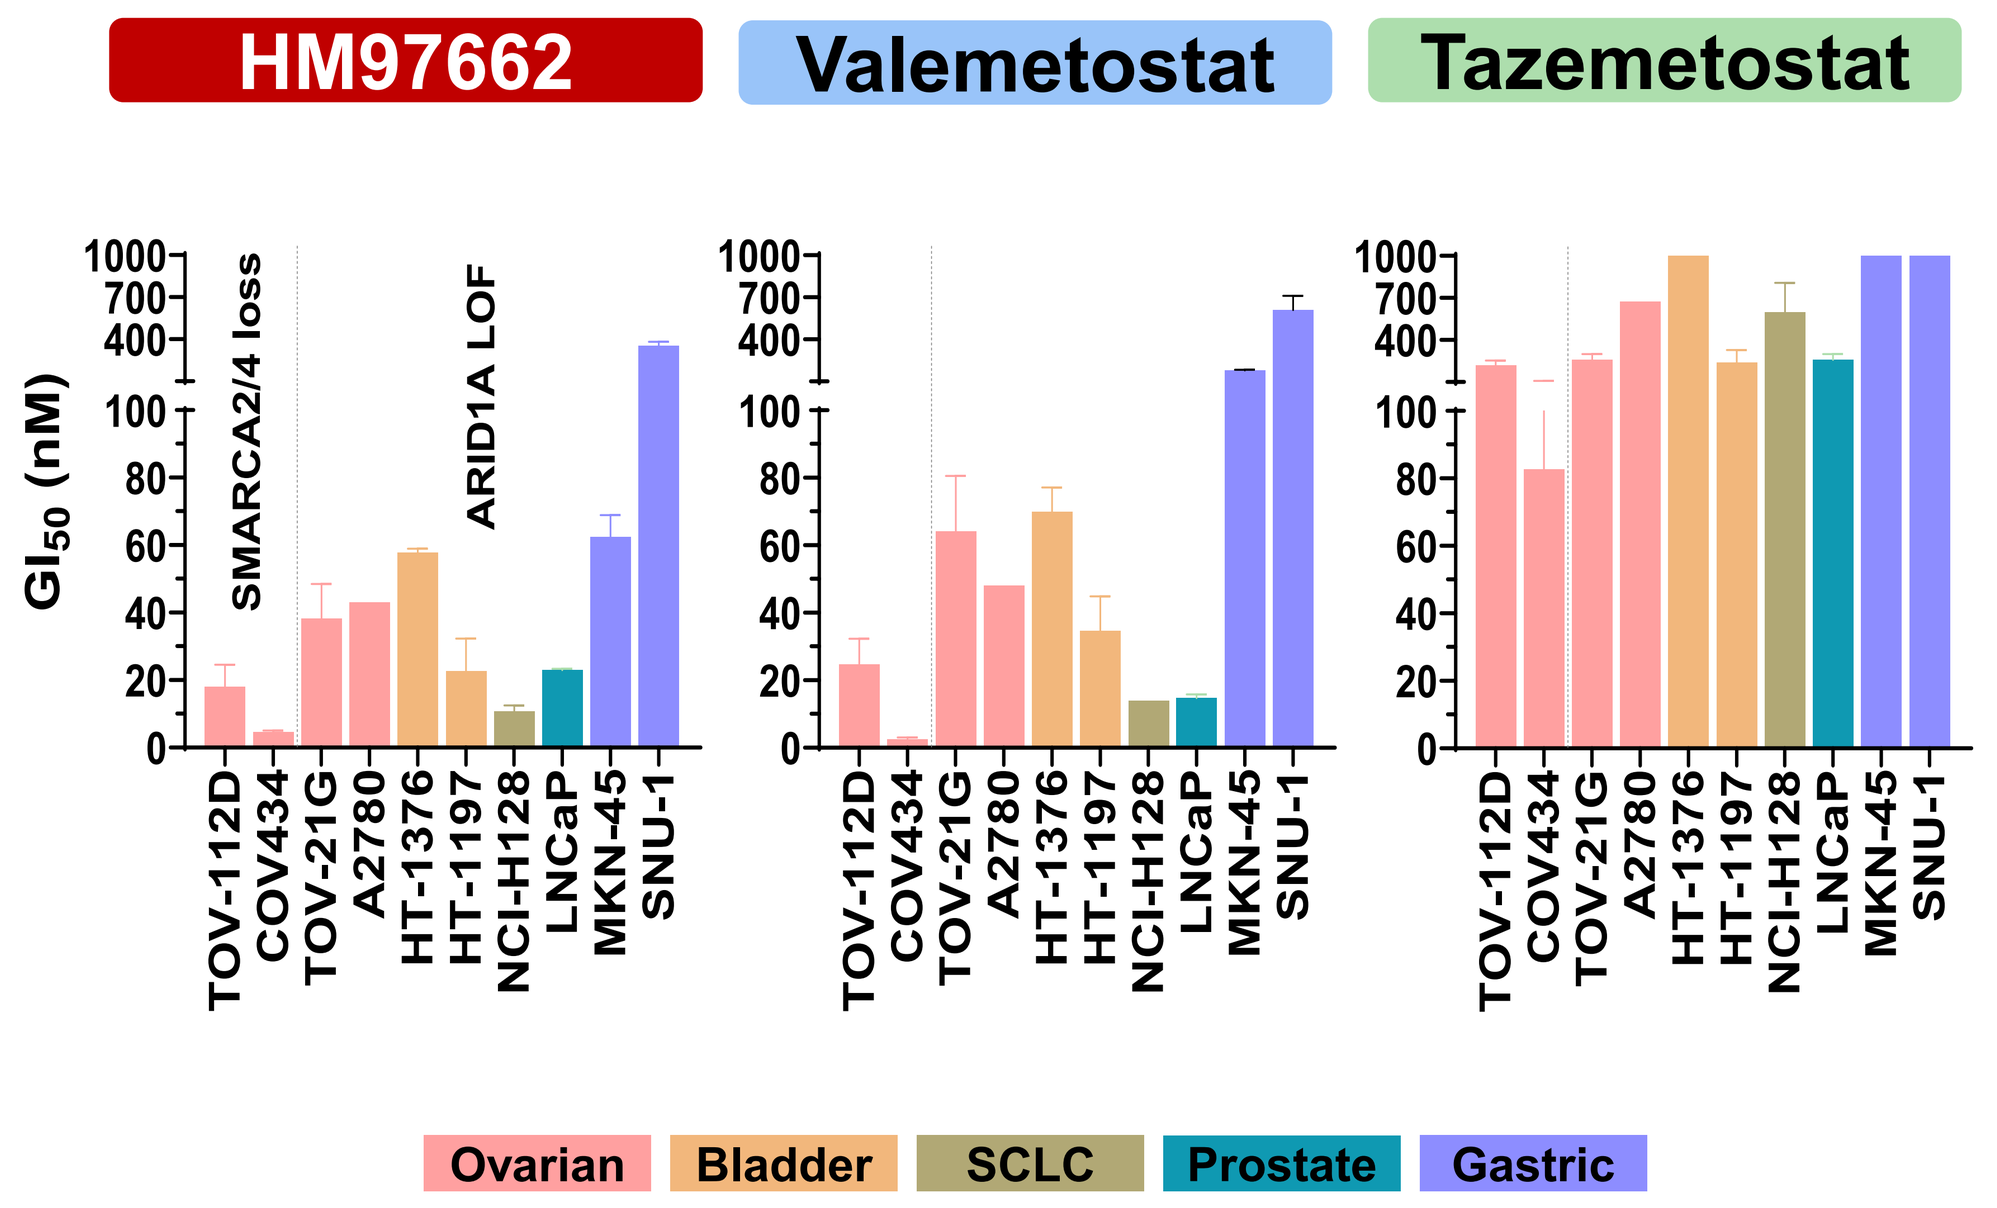
HM97662
Valemetostat
Tazemetostat
GI50 (nM)
SMARCA2/4 loss
ARID1A LOF
TOV-112D
COV434
TOV-21G
A2780
HT-1376
HT-1197
NCI-H128
LNCaP
MKN-45
SNU-1
Ovarian
Bladder
SCLC
Prostate
Gastric

Target Modulation of H3K27me3

A. HT-1376

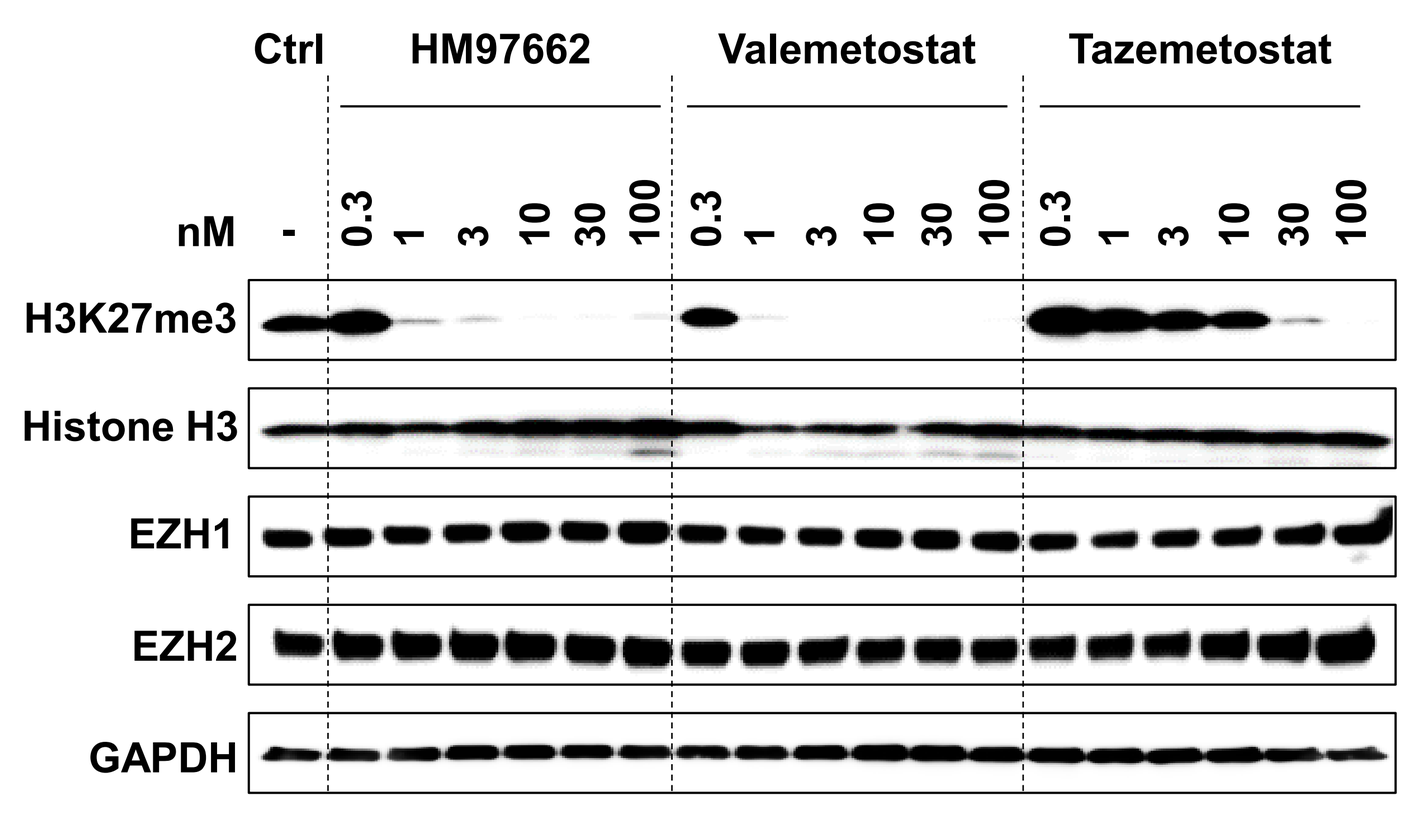
Ctrl
HM97662
Valemetostat
Tazemetostat
nM
H3K27me3
Histone H3
EZH1
EZH2
GAPDH

B. TOV-21G

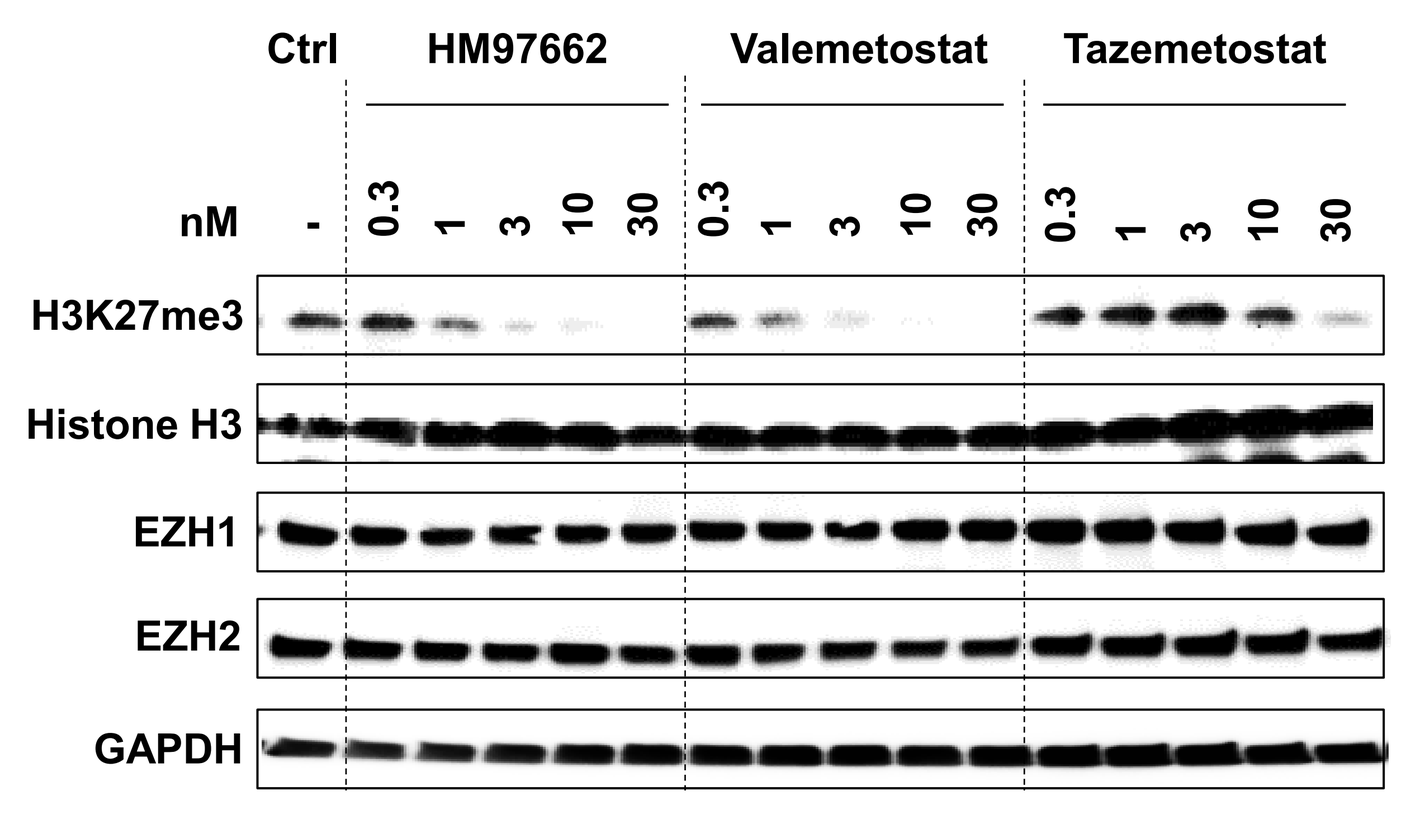
Ctrl
HM97662
Valemetostat
Tazemetostat
nM
H3K27me3
Histone H3
EZH1
EZH2
GAPDH

Cancer type	Cell line	Genetic alteration	H3K27me3 Inhibition (IC_{50}, nM)		
			HM97662	Valemetostat	Tazemetostat
Bladder	HT-1376	ARID1A LOF	0.9	0.5	12
Ovarian	TOV-21G	ARID1A LOF	1.2	1.6	27

Enhanced Sensitivity of Chemotherapy by HM97662

In vitro combination study with carboplatin in TOV-21G cells

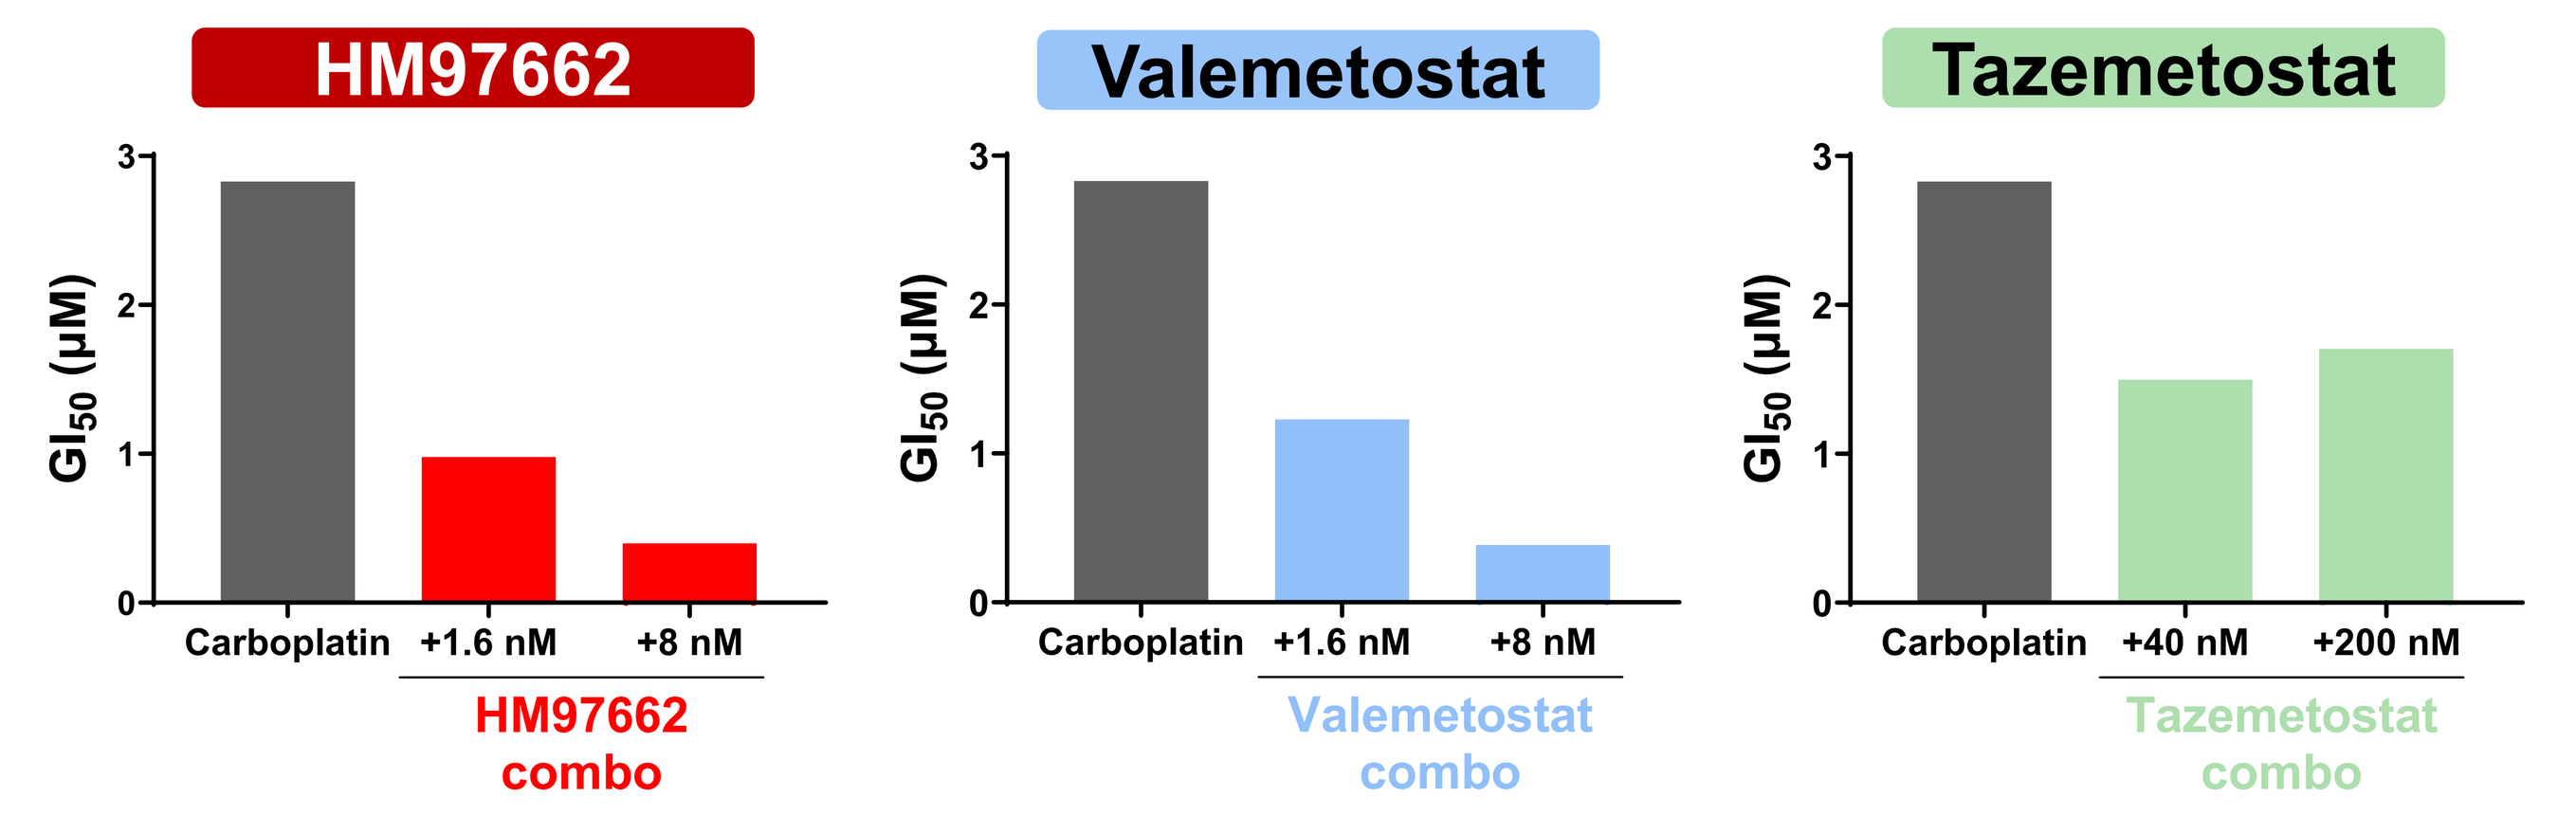
HM97662
Valemetostat
Tazemetostat
GI50 (μM)
Carboplatin
+1.6 nM
+8 nM
+40 nM
+200 nM
HM97662 combo
Valemetostat combo
Tazemetostat combo

Compounds	TOV-21G cells		
	Growth Inhibition (GI_{50}, nM)	Synergy Score (HSA)	Effect
HM97662	38	10.1	Synergism
Valemetostat	64	6.3	Moderate synergism
Tazemetostat	694	-4.5	Moderate antagonism

The synergy score was calculated using the HSA model, and the tool used was Combenefit.
Combenefit: an interactive platform for the analysis and visualization of drug combinations.

Synergistic effects of the HM97662 with platinum-based SoC

A. Bladder HT-1376

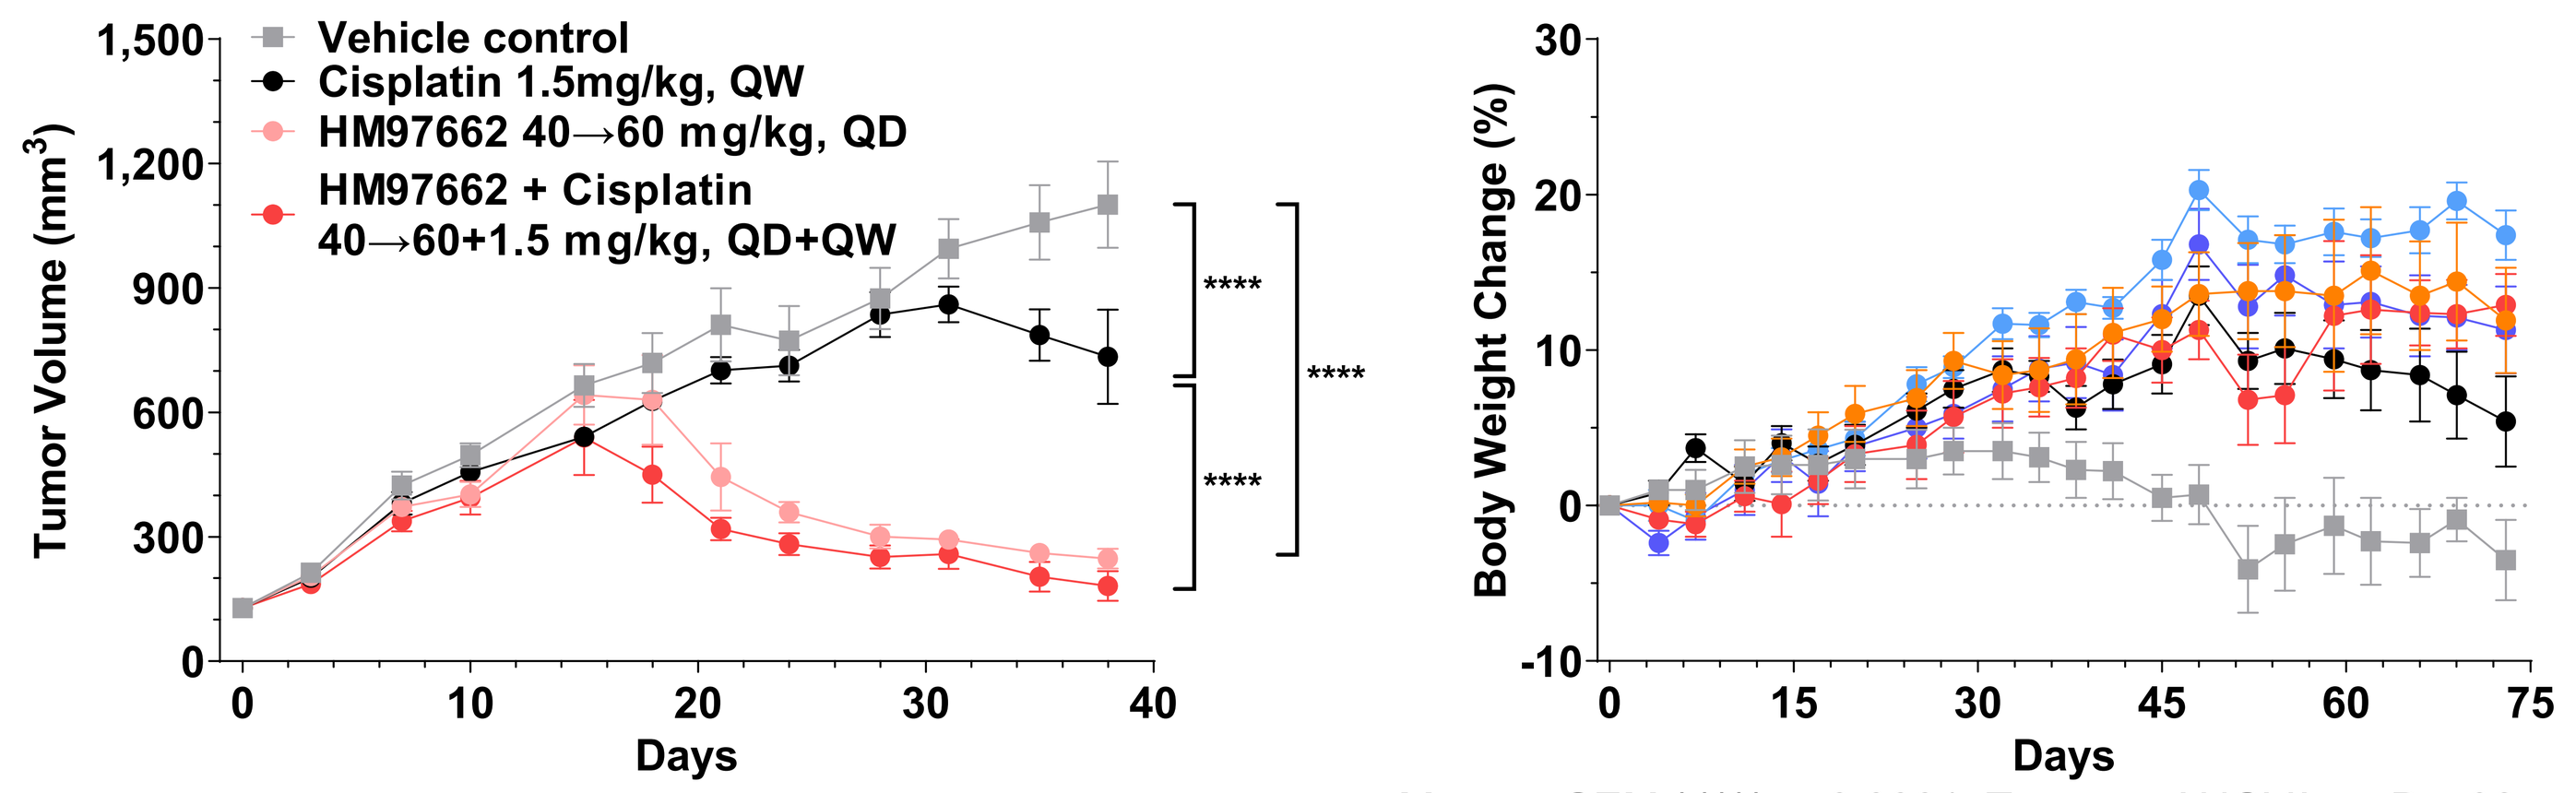
Vehicle control
Cisplatin 1.5mg/kg, QW
HM97662 40→60 mg/kg, QD
HM97662 + Cisplatin 40→60+1.5 mg/kg, QD+QW
Tumor Volume (mm3)
Body Weight Change (%)
Days

Mean ± SEM / **** p<0.0001, Two-way ANOVA on Day38

B. Bladder HT-1197

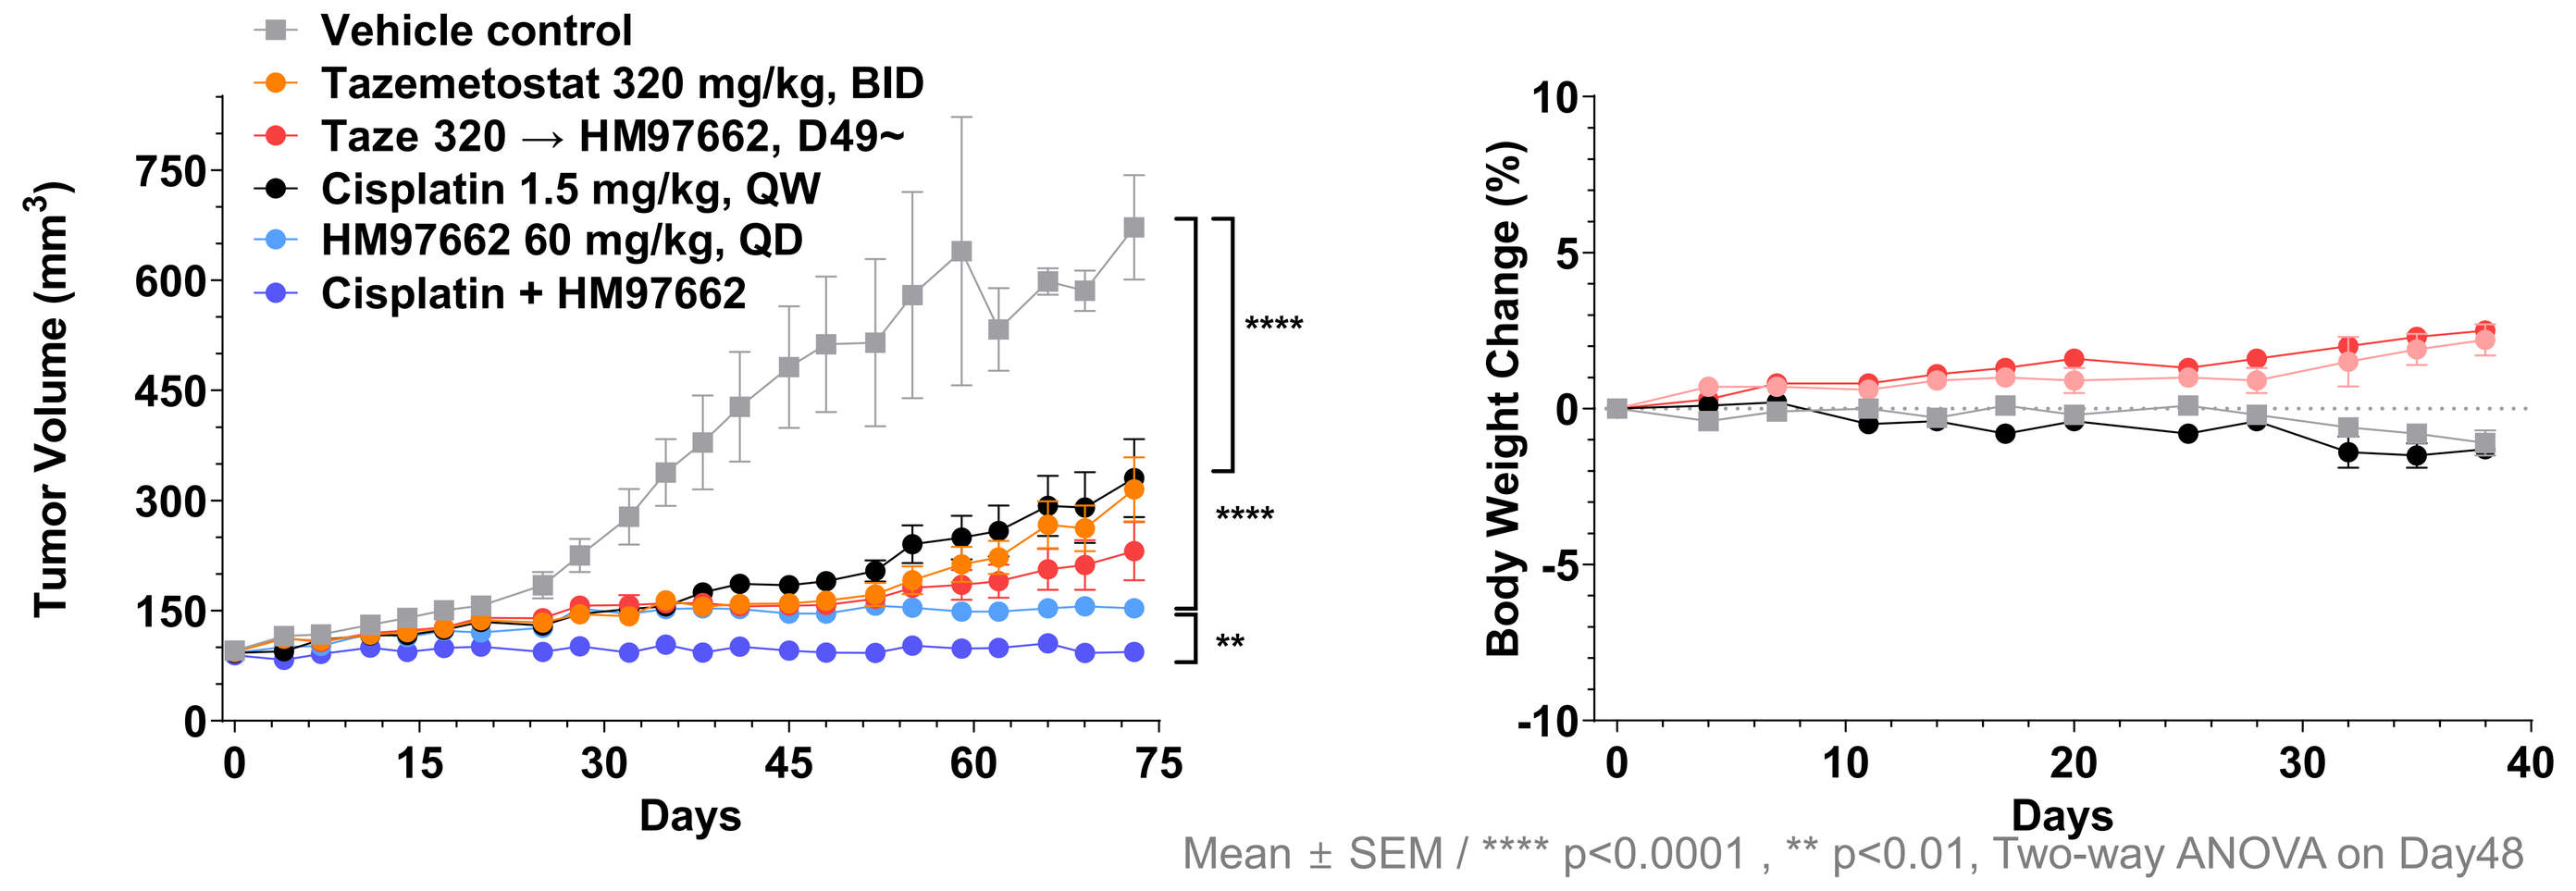
Vehicle control
Tazemetostat 320 mg/kg, BID
Taze 320 → HM97662, D49~
Cisplatin 1.5 mg/kg, QW
HM97662 60 mg/kg, QD
Cisplatin + HM97662
Tumor Volume (mm3)
Body Weight Change (%)
Days

Mean ± SEM / **** p<0.0001 , ** p<0.01, Two-way ANOVA on Day48

C. Ovarian TOV-21G

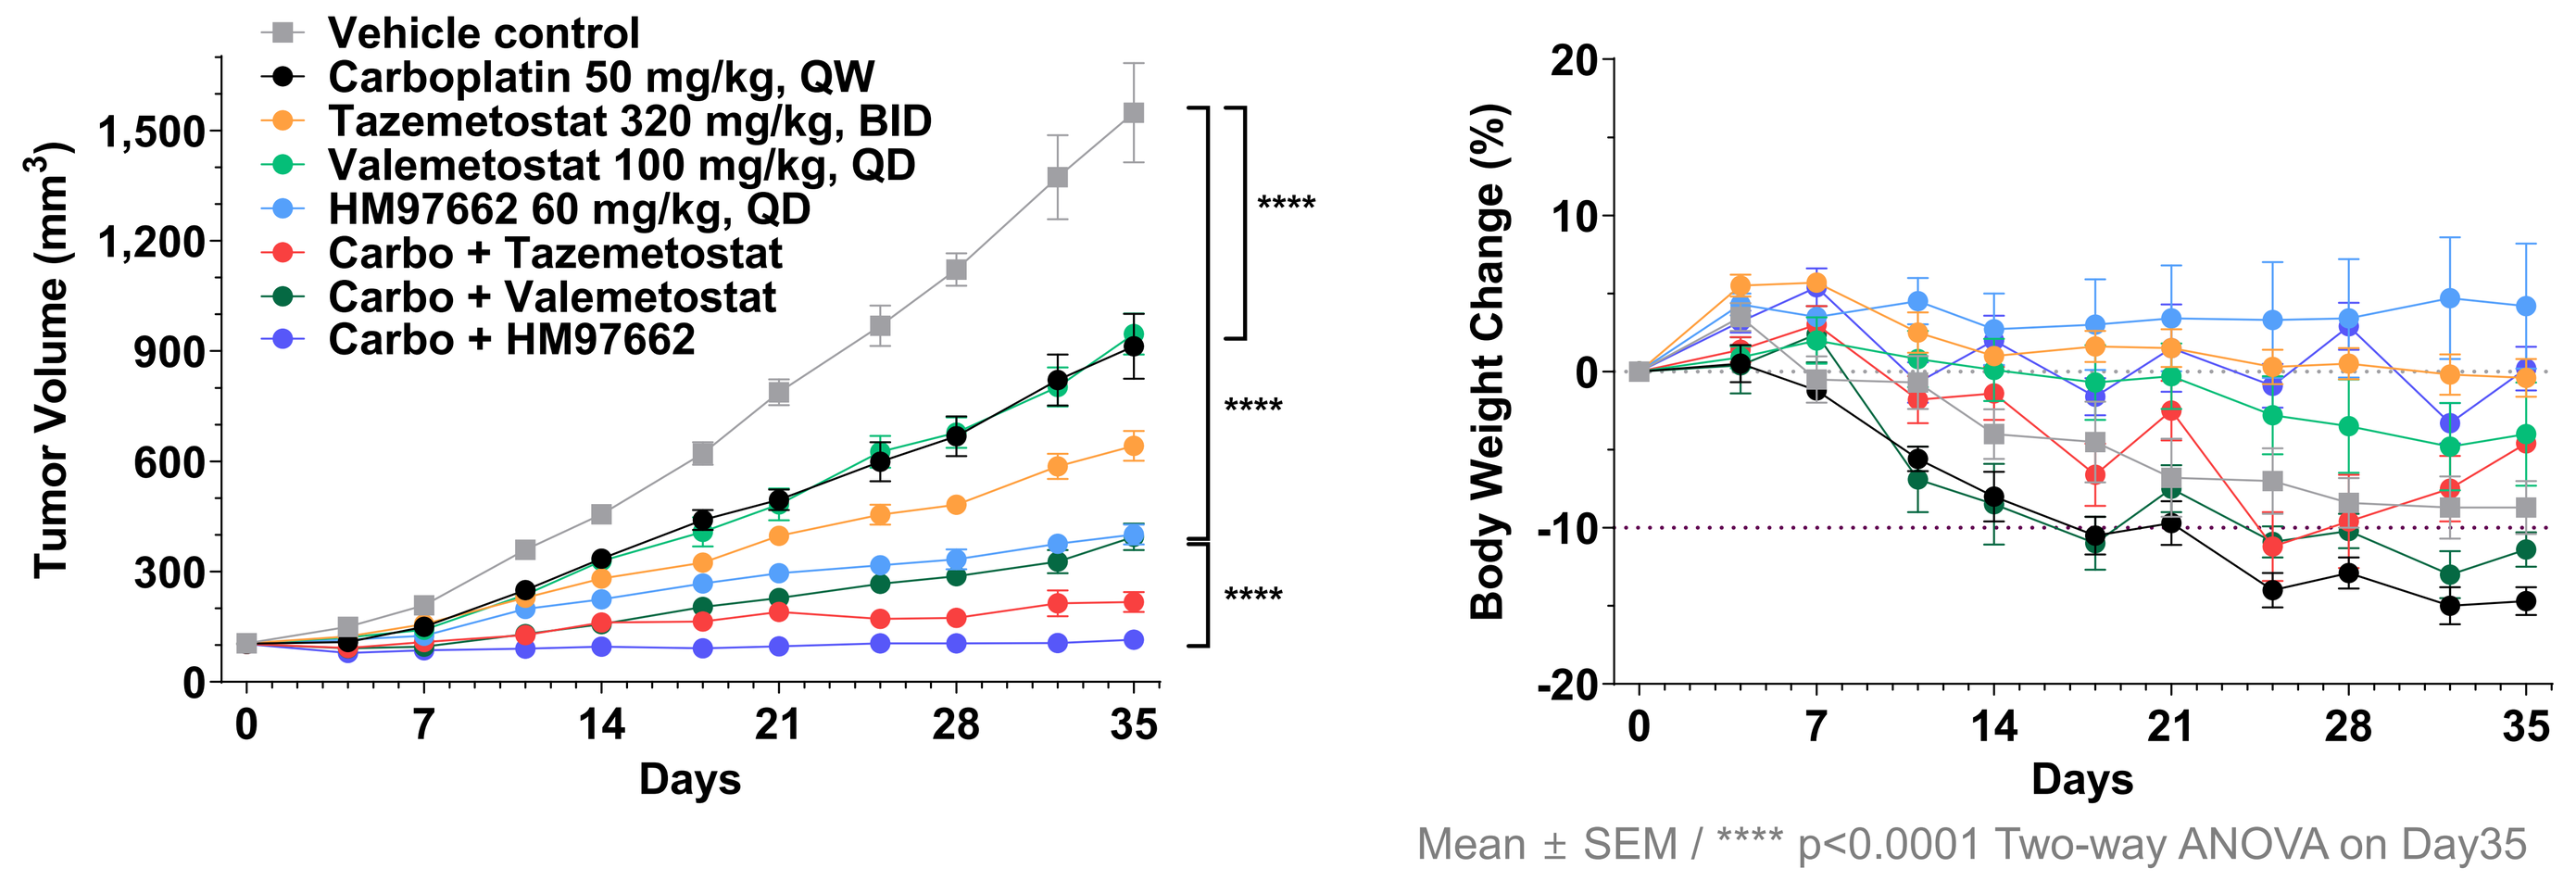
Vehicle control
Carboplatin 50 mg/kg, QW
Tazemetostat 320 mg/kg, BID
Valemetostat 100 mg/kg, QD
HM97662 60 mg/kg, QD
Carbo + Tazemetostat
Carbo + Valemetostat
Carbo + HM97662
Tumor Volume (mm3)
Body Weight Change (%)
Days

Mean ± SEM / **** p<0.0001 Two-way ANOVA on Day35

Synergistic effects of the HM97662 with TOP1 inhibitor

D. SCLC NCI-H82 (A related poster (abs no. 2405) will also be presented)

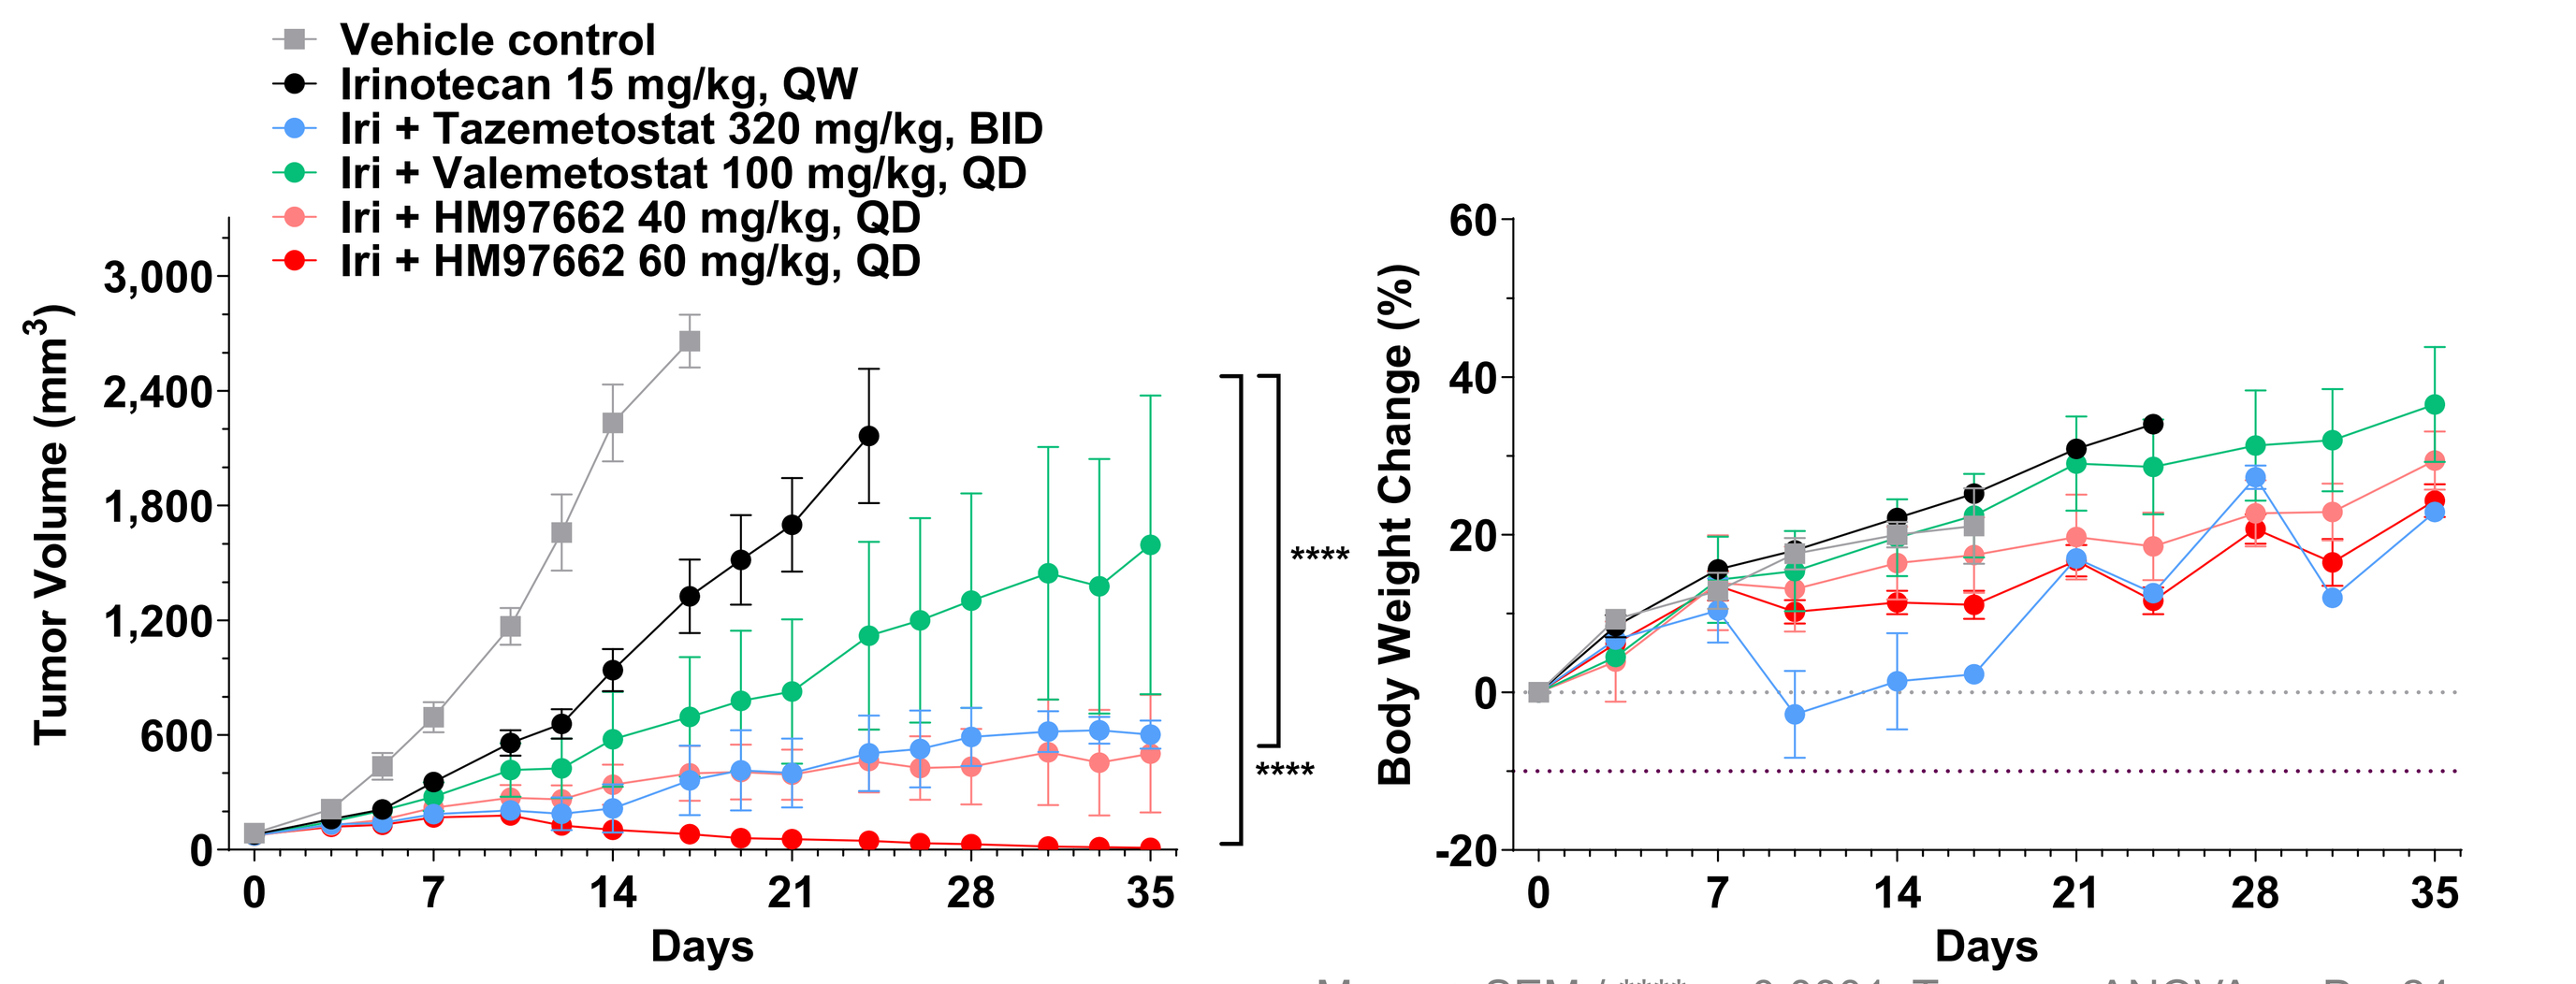
Vehicle control
Irinotecan 15 mg/kg, QW
Iri + Tazemetostat 320 mg/kg, BID
Iri + Valemetostat 100 mg/kg, QD
Iri + HM97662 40 mg/kg, QD
Iri + HM97662 60 mg/kg, QD
Tumor Volume (mm3)
Body Weight Change (%)
Days

Mean ± SEM / **** p<0.0001, Two-way ANOVA on Day24

E. Gastric MKN45

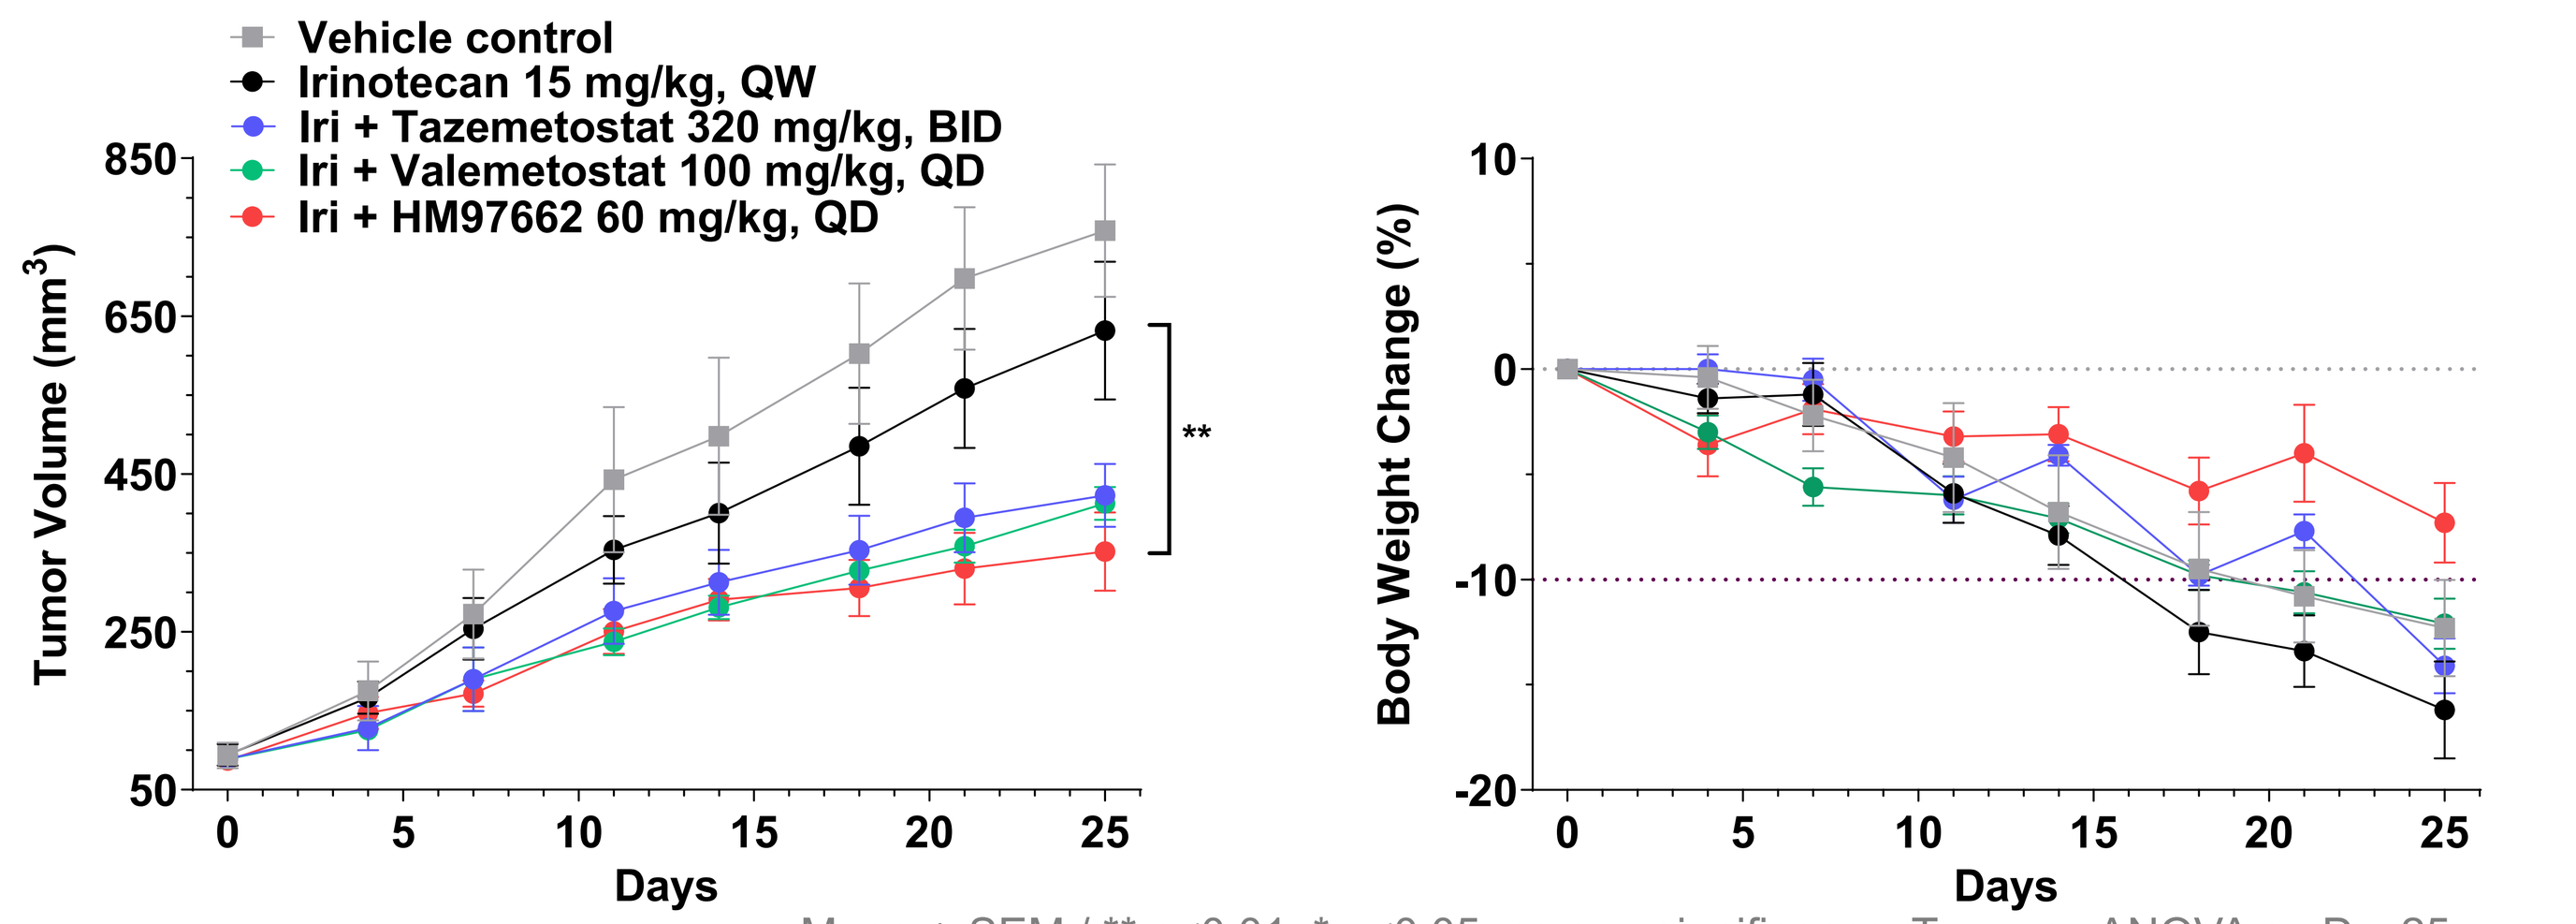
Vehicle control
Irinotecan 15 mg/kg, QW
Iri + Tazemetostat 320 mg/kg, BID
Iri + Valemetostat 100 mg/kg, QD
Iri + HM97662 60 mg/kg, QD
Tumor Volume (mm3)
Body Weight Change (%)
Days

Mean ± SEM / ** p<0.01, * p<0.05, ns non-significance, Two-way ANOVA on Day25

Concluding Remarks

- HM97662, an EZH1/2 dual inhibitor, showed a wide and strong growth inhibitory effect in various solid cancer cell lines.
- Moreover, HM97662 exhibited potent antitumor activities in xenograft mouse models with various solid cancer cells including ovarian, bladder, small cell lung cancer, and gastric cancers.
- Furthermore, HM97662 showed synergistic effects with chemotherapies such as topoisomerase 1 inhibitors and platinum-based standard treatment in associated tumor xenograft models.
- Taken together, HM97662 enhances the sensitivity of standard chemotherapy and maintains tolerability, as demonstrated by preclinical studies showing that its combination with chemotherapy promotes tumor regression at well-tolerated doses.
- Currently, a first-in-human phase 1 dose escalation study of HM97662 in advanced or metastatic solid tumors is underway in KR/AU (NCT05598151).

References

1) Duan R. et al., *Journal of Hematology & Oncology.* **2020,** *13,* 104;
2) Stefania C. et al., *Curr Oncol Rep.* **2023**, *25(11)*, 1277;
3) Huang K. et al., *Cell Cycle.* **2020**, *19(7)*, 758.

Acknowledgements

This research was supported by Korea Drug Development Fund funded by Ministry of Science and ICT, Ministry of Trade, and Energy, and Ministry of Health and Welfare (HN21C1077, Republic of Korea).

American Association for Cancer Research (AACR), April 25-30, 2025

Hanmi Pharm. Co., Ltd.