INTESTINOTROPHIC EFFECT OF A NOVEL LONG-ACTING GLP-2 ANALOG, HM15912, IN ANIMAL MODEL OF SHORT BOWEL SYNDROME AND POTENTIAL AS MONTHLY ADMINISTRATION

P-249 / Nutrition and chronic disease

J. H. Choi*, [1], J. B. Lee [1], E. J. Park [1], S. M. Bae [1], D. J. Kim [1], S. H. Lee [1], I. Y. Choi [1]
[1]HANMI PHARM. CO., LTD., Hwaseong-si, Korea, Republic Of

INTRODUCTION

HM15912, a long-acting GLP-2 analog, provided significant morphological and functional improvement in small intestine compared to daily or weekly GLP-2 medications via substantially extended half-life and systemic exposure

HM15912 is conjugated with a human IgG4 Fc *via* flexible PEG linker, and has a same biological mode of action with human GLP-2 via IGF-1

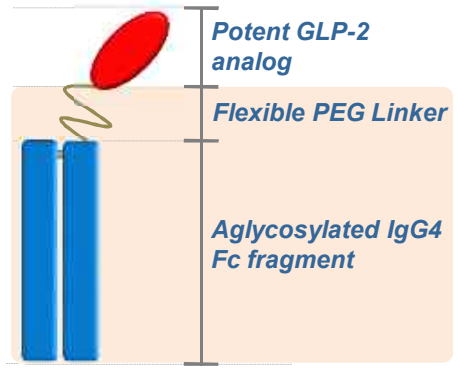
Potent GLP-2 analog
Flexible PEG Linker
Aglycosylated IgG4 Fc fragment

[General profile]

- **Rationally designed GLP-2 analog to have a more potent intestinotrophic action *vs* human GLP-2**
- **Extended half-life allows once-monthly dosing in human**
- **Ready-to-inject with soluble formation**

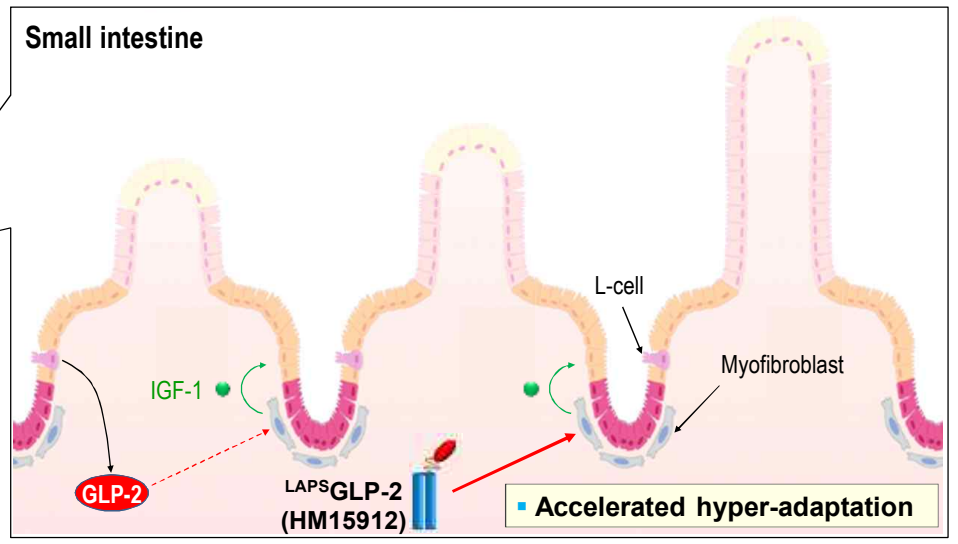
Small intestine
L-cell
Myofibroblast
IGF-1
GLP-2
LAPSGLP-2 (HM15912)
Accelerated hyper-adaptation

LAPSCOVERY : Long Acting Peptide/Protein DiSCOVERY Technology

This figure was modified from the following reference, *AJP-GI 307: G1147–G1168 (2014)*

OBJECTIVES

- **Short bowel syndrome (SBS) is characterized by reduced intestinal length and insufficient absorptive surface area, leading to malabsorption of essential nutrients. In consequence, parenteral nutrition (PN) by intravenous supplementation is required. To reduce PN dependency, the only approved GLP-2 agonism-associated drug for SBS is Revestive® (teduglutide), daily GLP-2 analog. Nevertheless, the widespread use of teduglutide may be still limited due to insufficient efficacy, frequent daily administration and complicated preparation steps for its reconstitution.**
- **Hence, there is a medical unmet needs for more effective and longer lasting GLP-2 analog drugs. We have developed a long-acting GLP-2 analog, HM15912, to provide a novel therapeutic option. Here, we investigated the potential therapeutic effect of HM15912 in small bowel resected animal model and intestinotrophic effect in normal mice.**

METHODS

Experimental scheme

Study #1 : Intestinotrophic efficacy in SBS model rats

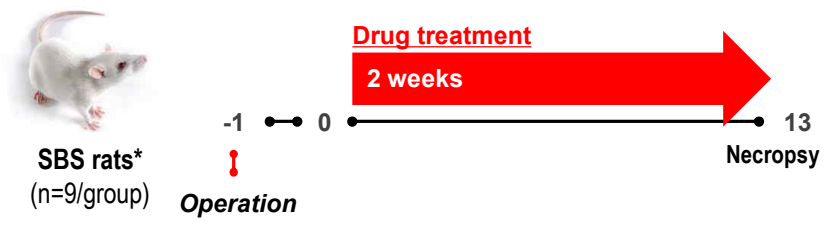
Drug treatment
2 weeks
-1
0
13
Necropsy
SBS rats*
(n=9/group)
Operation

*80% intestinal resection

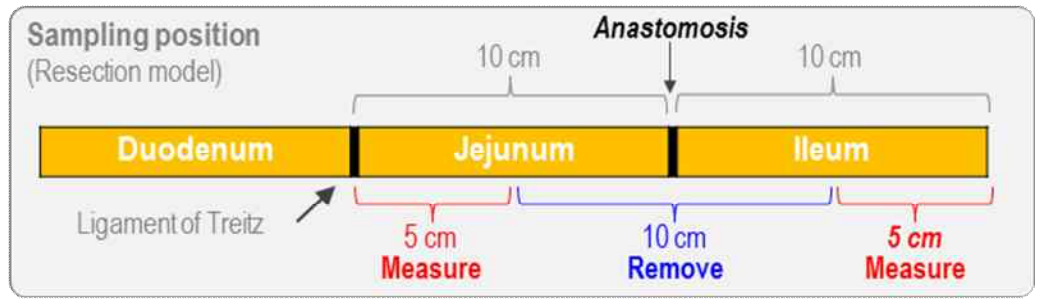
Sampling position
(Resection model)
Anastomosis
10 cm
10 cm
Duodenum
Jejunum
Ileum
Ligament of Treitz
5 cm
Measure
10 cm
Remove
5 cm
Measure

Study #2 : Intestinotrophic efficacy according to various dosing regimens in mice

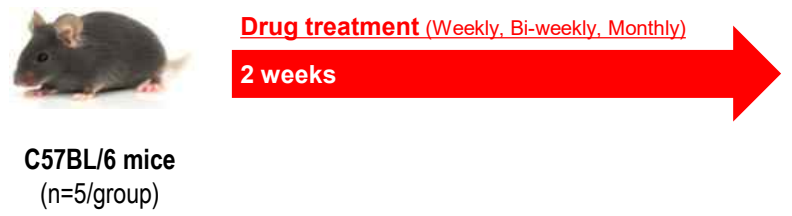
Drug treatment (Weekly, Bi-weekly, Monthly)
2 weeks
C57BL/6 mice
(n=5/group)

- In study #1, to induce short bowel syndrome, SD rats were resected 80% of small intestine. In this jejuno-ileal resected rats, tedulgutide (s.c., BID) or HM15912 (s.c., Q2D) were subcutaneously administered for 2 weeks. Non-operation and sham operation rats were used as negative control. At the end of treatment, wet weight of jejunum and ileum were measured, and histological analysis such as villus height, crypt depth, and mucosal area were also evaluated. Additionally, serum D-xylose concentrations were measured to evaluate absorption capacity (D-xylose absorption test).
- In study #2, to further investigate benefit of extended dosing interval supporting once monthly administration in human, C57BL/6 mice were treated with teduglutide or weekly GLP-2 analogs by their respective treatment regimen considering the typical treatment regimen in human, or HM15912 by various administration regimens, every other day or once weekly for 2 weeks. Small intestine mass and serum D-xylose concentrations were measured at the end of treatment.

RESULTS

Intestinotrophic efficacy in SBS model rats

Figure 1. Significant intestinotrophic efficacy of HM15912 compared to teduglutide in SBS model rats (Study #1)

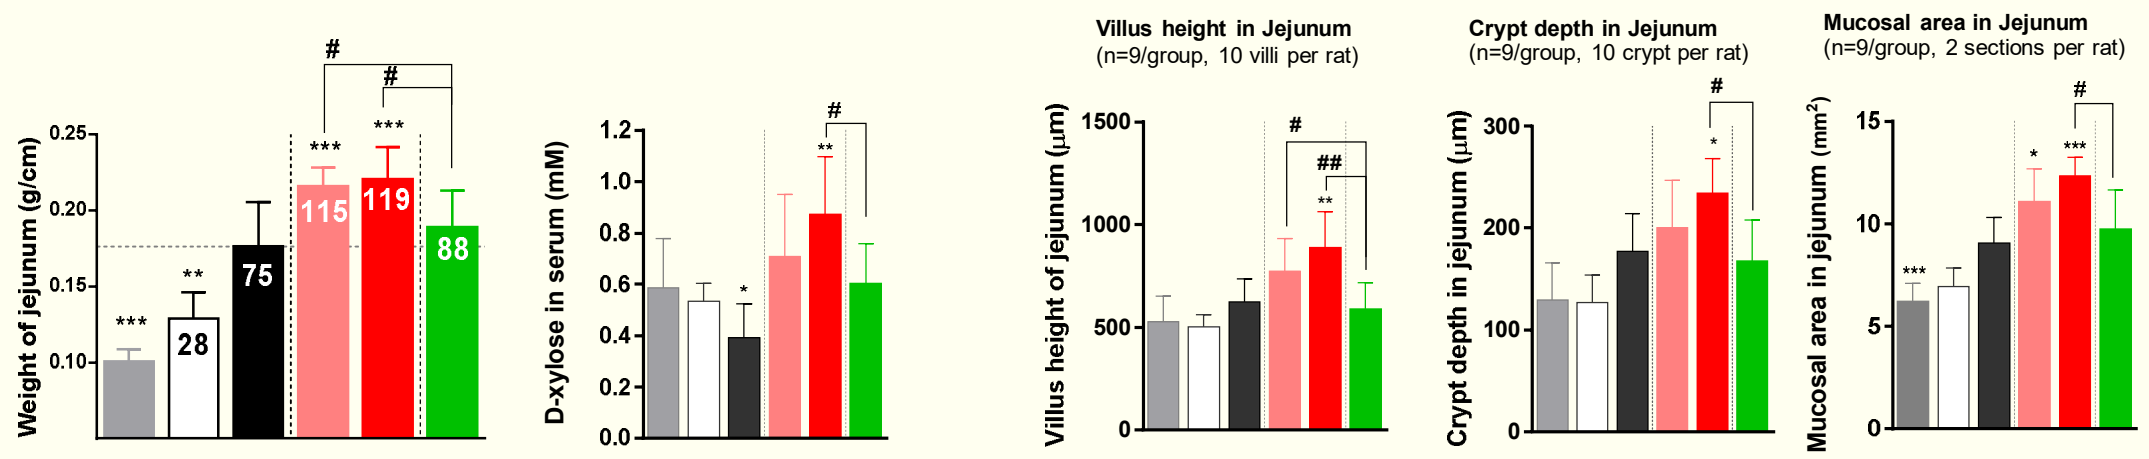
(a) Weight of jejunum
(b) Serum D-xylose level
(c) Histological analysis
Weight of jejunum (g/cm)
115
119
88
75
28
D-xylose in serum (mM)
Villus height in Jejunum
(n=9/group, 10 villi per rat)
Villus height of jejunum (µm)
Crypt depth in Jejunum
(n=9/group, 10 crypt per rat)
Crypt depth in jejunum (µm)
Mucosal area in Jejunum
(n=9/group, 2 sections per rat)
Mucosal area in jejunum (mm²)

Non-operation vehicle	Teduglutide,	30.0 nmol/kg/BID, SC **[0.05 mg/kg/day HED]**
Sham-operation vehicle	HM15912,	4.5 nmol/kg/Q2D, SC **[0.06 mg/kg/week HED]**
Resection vehicle	HM15912,	30.0 nmol/kg/Q2D, SC **[1.0 mg/kg/week HED]**

*, Significantly differ. vs. Non-operation by one way ANOVA test
#, Significantly differ. vs. teduglutide by one way ANOVA test

➢In 80% jejuno-ileal resected rats, HM15912 treatment significantly increased wet weight of jejunum compared to resection vehicle or teduglutide treated group, but this is not the case of ileum. This results were well-correlated with histological analysis such as villus height, crypt depth, and mucosal area. In addition, HM15912 treated group was associated with a significant increase in absorption capacity of small intestine based on serum D-xylose concentrations. *HED= Human equivalent dose considering body surface area*

Intestinotrophic efficacy according to various dosing regimens in mice

Figure 2. Significant intestinotrophic efficacy of HM15912 compared to daily and weekly GLP-2 analogs in C57BL/6 mice (Study #2)

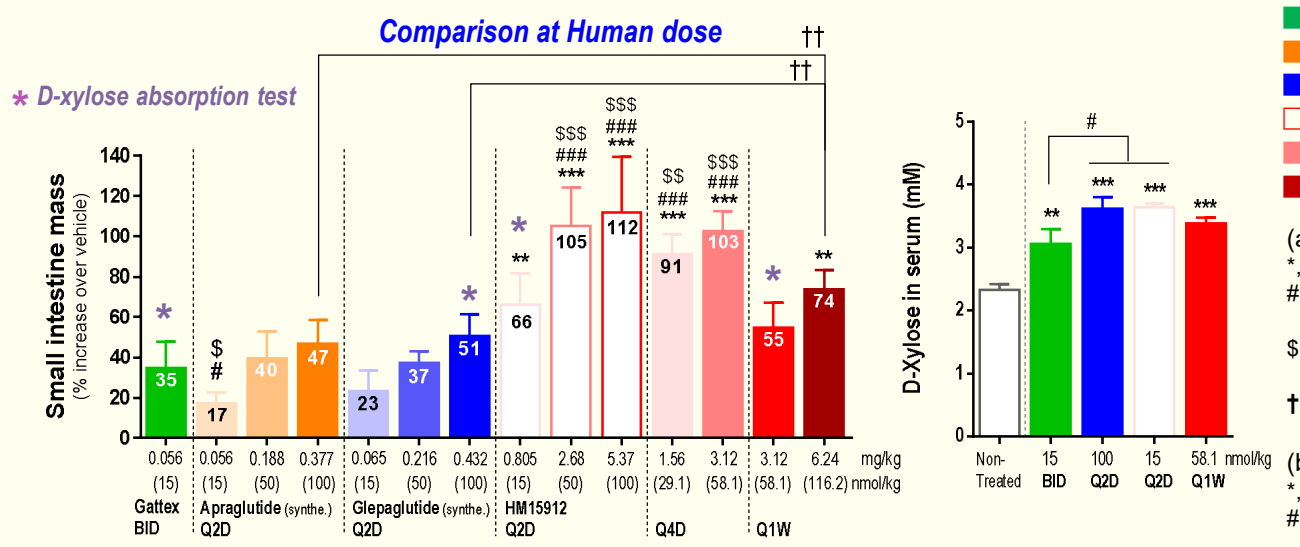
(a) Small intestine mass
(b) Serum D-xylose level
Comparison at Human dose
* D-xylose absorption test
Small intestine mass
(% increase over vehicle)
35
17
40
47
23
37
51
66
105
112
91
103
55
74
0.056 (15) Gattex BID
0.056 (15) 0.188 (50) 0.377 (100) Apraglutide (synthe.) Q2D
0.065 (15) 0.216 (50) 0.432 (100) Glepaglutide (synthe.) Q2D
0.805 (15) 2.68 (50) 5.37 (100) HM15912 Q2D
1.56 (29.1) 3.12 (58.1) Q4D
3.12 (58.1) 6.24 (116.2) Q1W
mg/kg
nmol/kg
D-Xylose in serum (mM)
Non-Treated
15 BID
100 Q2D
15 Q2D
58.1 Q1W
nmol/kg

Teduglutide	15.0 nmol/kg/BID, SC **[E_{max}, EPAR]**
Apraglutide (Synthesized)	~100 nmol/kg/Q2D, SC **[~10 mg/week HED]**
Glepaglutide (Synthesized)	~100 nmol/kg/Q2D, SC **[~10 mg/week HED]**
HM15912 (weekly)	~100 nmol/kg/Q2D, SC **[~1.5 mg/kg/week HED]**
HM15912 (Biweekly)	~58.1 nmol/kg/Q4D, SC **[~0.9 mg/kg/bi-week HED]**
HM15912 (Monthly)	~116.2 nmol/kg/QW, SC **[~2 mg/kg/month HED]**

(a) Small intestine mass
*, Significantly differ. vs. Teduglutide by one way ANOVA test
#, Significantly differ. vs. Apraglutide (Synthesized) 100 nmol/kg/Q2D by one way ANOVA test
$, Significantly differ. vs. Glepaglutide (Synthesized) 100 nmol/kg/Q2D by one way ANOVA test
†, Significantly differ. by unpaired T-test

(b) D-xylose absorption at week 2
*, Significantly differ. vs. vehicle by one way ANOVA
#, Significantly differ. vs. Teduglutide by one way ANOVA

➢In normal mice, all administration regimens of HM15912 significantly increased small intestine (SI) mass than tedulgutide and weekly GLP-2 analogs. At equimolar dose with Q2D, HM15912 significantly increased SI mass than weekly GLP-2 analogs (112% vs 47% and 51% over vehicle). Even after weekly administration of HM15912 mimicking once a month in human, it also significantly increased SI mass (74% over vehicle) than weekly GLP-2 analogs (47% and 51% over vehicle). *HED= Human equivalent dose considering body surface area*

CONCLUSIONS

- **HM15912, novel long-acting GLP-2 analog, significantly improved not only small intestinal growth but also nutrition absorbing capacity than tedugltuide as well as weekly GLP-2 analogs, which are currently under clinical development, even with a less dosing frequency, supporting that HM15912 will provide a better treatment option to SBS patients in terms of remarkable small bowel tropic effect and more extended administration interval.**

REFERENCES

1) *AJP-GI 307: G1147–G1168 (2014), 2) Teduglutide EMEA/H/C/002345, 3) ClinicalTrials.gov Identifier: NCT03905707 (Glepaglutide)*

Hanmi Pharm. Co., Ltd.