A First-in-Human, Double-blinded, Randomized, Placebo-controlled, Single Ascending Dose Study to Assess Safety, Tolerability, Pharmacokinetics and Pharmacodynamics of HM15912 in Healthy Subjects

Minsoo Kang[1], JaeDuk Choi[1], Sujin Seo[1], Jiyoung Sin[1], Hyeong-Seok Lim[2], Seungjae Baek[1],
[1]Hanmi Pharm. Co., Ltd, Seoul, Korea. [2]Clinical Trial Center at Asan Medical Center, Seoul, Korea.

Introduction

HM15912 is a novel long-acting glucagon like peptide-2 (GLP-2) agonist with extended half-life through reduced renal clearance and FcRn-mediated vascular endothelial recycling. In the rat model of short bowel syndrome (SBS), HM15912 has shown the intestinotrophic effects and induced intestinal growth with absorption capacity improvement (Abstract No. #939208).

This is a first-in-human (FiH), randomized, double-blinded, placebo-controlled, single ascending dose study to assess the safety, tolerability, pharmacokinetics (PK) and pharmacodynamics (PD) of HM15912 in adult healthy volunteers. The study evaluates 5 sequential doses (0.05, 0.1, 0.5, 1.0 and 1.5 mg/Kg, respectively) with enrolling 8 subjects at each dose. The subjects randomly assigned to HM15912 and placebo by 3:1 ratio (6 on HM15912, 2 on placebo). To minimize the risk to the subjects, the sentinel dosing approach was employed with each cohort. The subject should be hospitalized from the day before administration until D7. After discharge from the hospital, the subject should have two outpatient visits on D10 and D17, and safety follow-up visit on D30. For cohort 4 and 5 (1.0 mg/Kg and 1.5 mg/Kg), more frequent and longer visits (D8, 10, 17 and 30) with longer safety follow up (D44) was conducted. The DEM (Dose Escalation Meeting) was conducted when at least 6 subjects in each cohort have passed D17 with available blind safety, PK and PD data for the next dose escalation.

Primary objective in this study was to assess the safety and tolerability of HM15912 after single subcutaneous (SC) administration.

STUDY DESIGN

Figure 1. Study Scheme

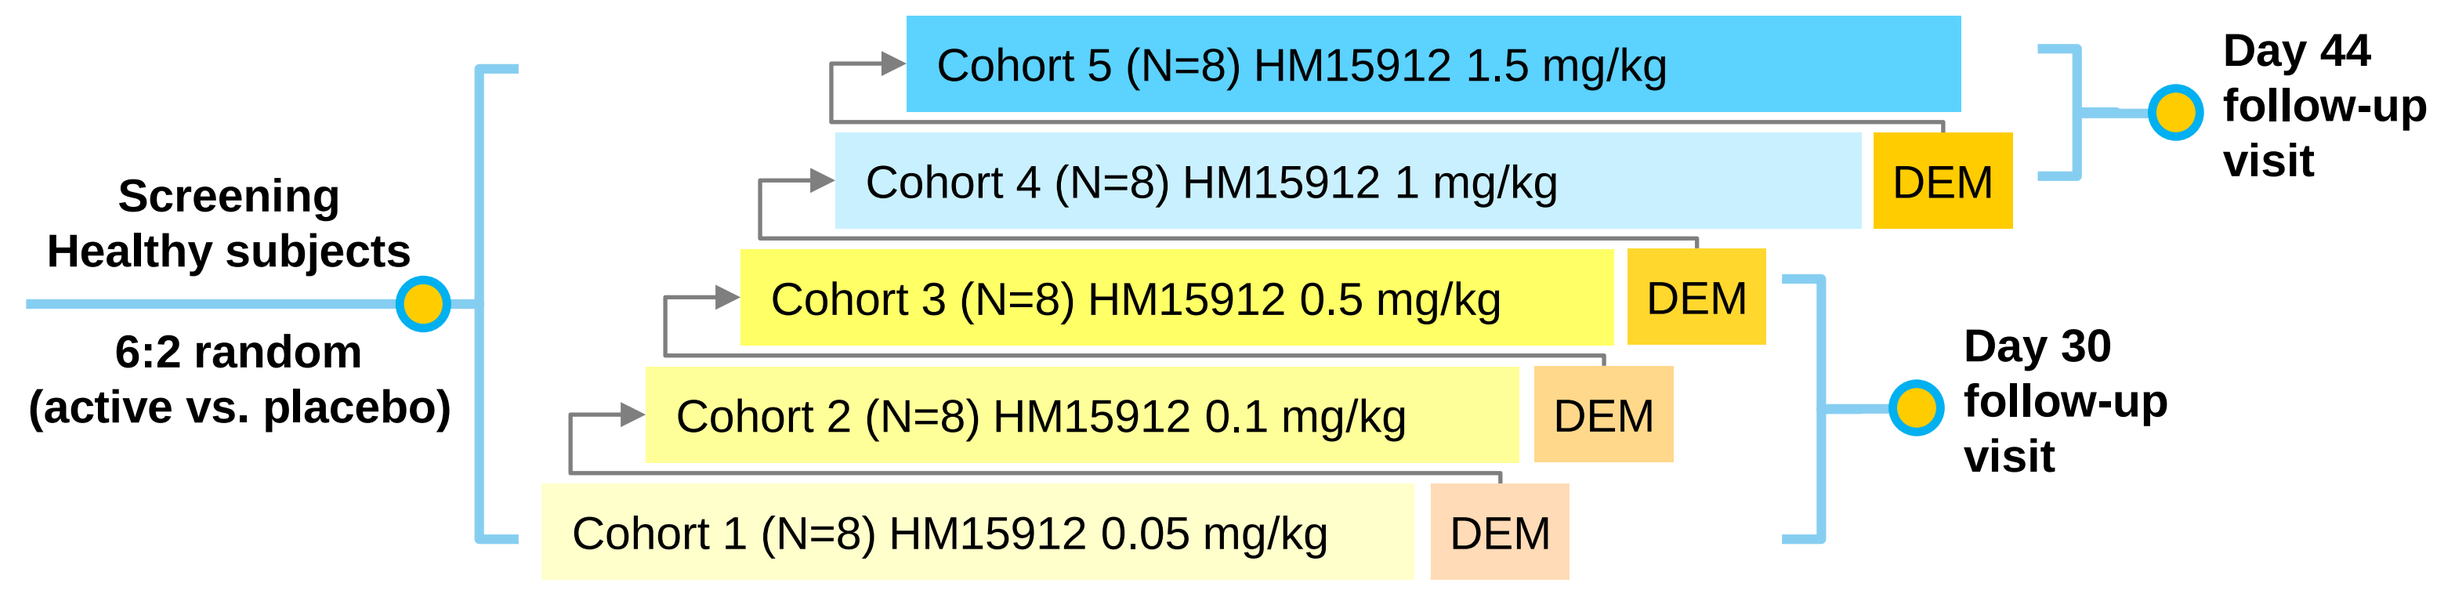
Screening
Healthy subjects
6:2 random
(active vs. placebo)
Cohort 5 (N=8) HM15912 1.5 mg/kg
Cohort 4 (N=8) HM15912 1 mg/kg
Cohort 3 (N=8) HM15912 0.5 mg/kg
Cohort 2 (N=8) HM15912 0.1 mg/kg
Cohort 1 (N=8) HM15912 0.05 mg/kg
DEM
Day 44 follow-up visit
Day 30 follow-up visit

*DEM: Dose-escalation meeting

- **Primary objective:** To assess safety and tolerability of HM15912
- **Secondary objective:** To assess PK profile of HM15912
- **Exploratory objective:** To assess PD properties of HM15912

BACKGROUND

HM15912 is a GLP-2 analogue, conjugated with a human IgG Fc fragment *via* a flexible PEG linker. GLP-2 analog component is synthesized based on human GLP-2 and it is responsible for the desired pharmaceutical effects. The Fc portion of HM15912 is designated to have an extended half-life without any immune-mediated effector function.

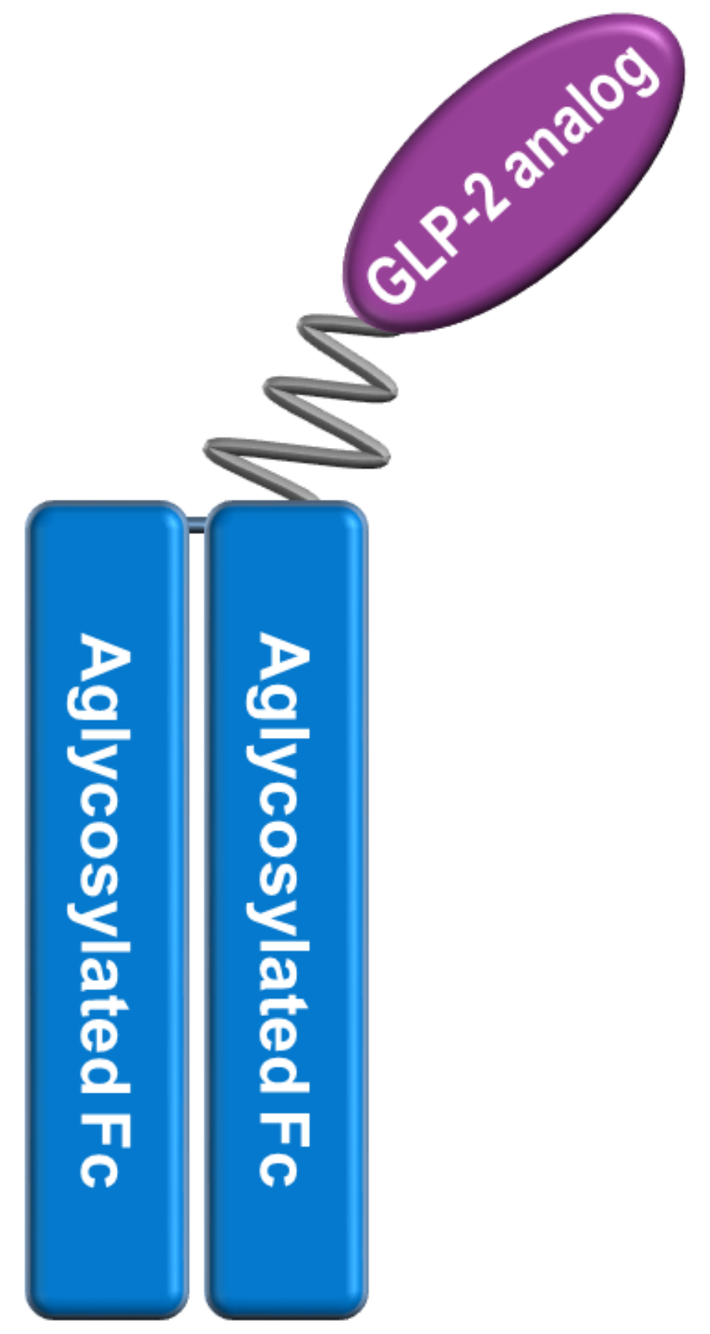
GLP-2 analog
Aglycosylated Fc
Aglycosylated Fc

[General Profile of HM15912]

- HM15912 is formulated as an aqueous solution (ready-to-inject).
- HM15912 induced the expression of IGF-1 in mouse primary intestinal subepithelial myofibroblasts (ISEMFs) to a similar level as native human GLP-2.
- In vivo study, HM15912 had a higher potency in not only morphological improvement (increase in small intestine weight) but also functional improvement (increase in absorption of D-xylose) than the commercially available GLP-2 analog in 80% jejuno-ileal resection rat model
- During the 26 weeks toxicity studies, the rats and monkeys showed generally well tolerated safety profiles and showed the hypertrophy of small intestine

Results

- **Demographics characteristics**

Table 1. Demographics Characteristics

Characteristics	Category	HM15912					Placebo (N=10)
		0.05 mg/kg (N=6)	0.1 mg/kg (N=6)	0.5 mg/kg (N=6)	1.0 mg/kg (N=6)	1.5 mg/kg (N=6)	
Sex, n (%)	Male	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	10 (100.0)
Age, (years)	N (%)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	10 (100.0)
	Mean (SD)	31.0 (5.18)	28.3 (8.87)	26.0 (5.83)	25.0 (3.52)	31.7 (7.39)	27.2 (2.39)
Weight, (kg)	N (%)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	10 (100.0)
	Mean (SD)	68.10 (7.76)	69.90 (4.52)	71.78 (7.51)	62.45 (5.55)	71.37 (5.77)	69.40 (7.78)
Height, (cm)	N (%)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	10 (100.0)
	Mean (SD)	168.8 (5.23)	171.7 (3.33)	174.0 (5.02)	169.0 (3.22)	174.3 (4.55)	175.6 (4.97)
BMI, (kg/m²)	N (%)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	6 (100.0)	10 (100.0)
	Mean (SD)	23.88 (2.19)	23.70 (0.87)	23.67 (1.33)	21.83 (1.68)	23.52 (2.28)	22.54 (2.67)

- **Pharmacokinetics**

a. Serum concentration of HM15912 reached its peak levels (mean t_{max}) in 71.8 to 142.9 hours. Mean terminal elimination $t_{1/2}$ of HM15912 was estimated to approximately 108 to 167.4 hours (Figure 2 and Table 2).

Figure 2. Serum HM15912 concentration

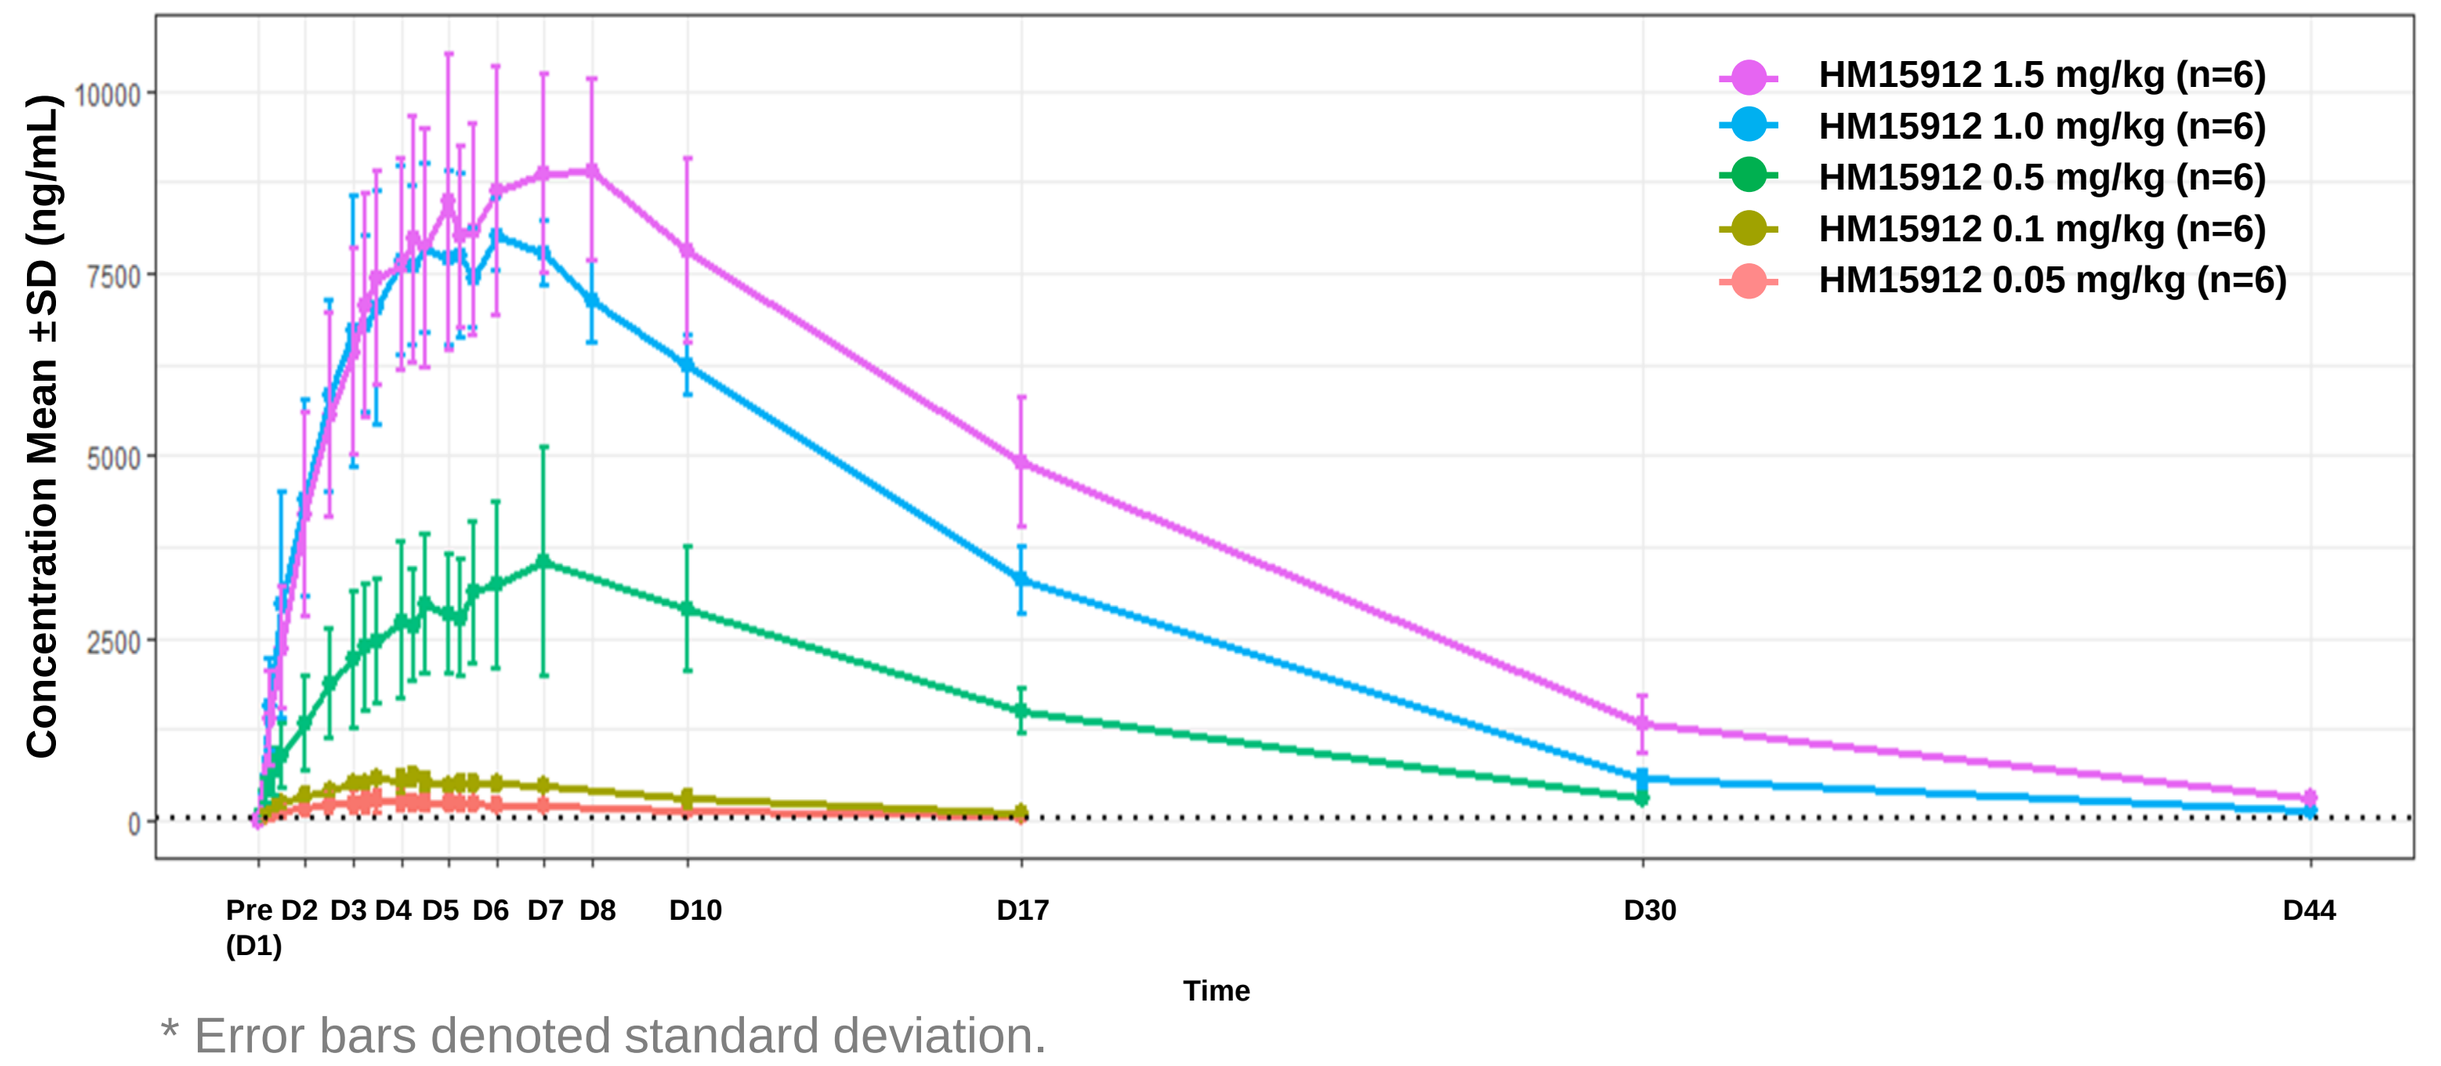
HM15912 1.5 mg/kg (n=6)
HM15912 1.0 mg/kg (n=6)
HM15912 0.5 mg/kg (n=6)
HM15912 0.1 mg/kg (n=6)
HM15912 0.05 mg/kg (n=6)
Concentration Mean ±SD (ng/mL)
10000
7500
5000
2500
0
Pre (D1) D2 D3 D4 D5 D6 D7 D8 D10 D17 D30 D44
Time

* Error bars denoted standard deviation.

Table 2. Summary of PK parameters of HM15912

Mean (SD)	HM15912				
	0.05 mg/kg (N=6)	0.1 mg/kg (N=6)	0.5 mg/kg (N=6)	1.0 mg/kg (N=6)	1.5 mg/kg (N=6)
C_{max} (ng/mL)	275.5 (135.51)	627.8 (95.05)	3740.0 (1331.50)	8385.0 (878.74)	9463.3 (1304.43)
t_{max} $(hour)$	91.92 (32.78)	71.84 (21.06)	142.92 (40.65)	100.82 (31.92)	137.27 (31.84)
$t_{1/2}$ $(hour)$	107.95 (19.12)	110.81 (26.87)	147.16 (17.80)	138.34 (10.39)	167.39 (14.01)
AUC_{last} $(ng \cdot h/mL)$	56473.66 (15420.71)	129202.85 (20388.32)	1139559.12 (288280.56)	2741811.90 (87787.75)	3533094.11 (429715.92)
AUC_{inf} $(ng \cdot h/mL)$	66236.99 (15445.99)	142276.41 (28246.98)	1237890.30 (307305.37)	2765271.37 (84076.12)	3609893.96 (450941.89)

- **Safety and tolerability**

a. All of the TEAEs reported in Study HM-GLP2-101 were mild and moderate in severity. No serious AEs (SAEs) or deaths were reported in the study. no subjects have discontinued study due to TEAEs (Table 3). The most frequent TEAEs was injection site bruising (17.5%; 7/40), nasopharyngitis (3/40; 7.5%) and headache (3/40; 7.5%).

b. For the TRAE (Treatment Related AE), there are three moderate TRAEs during the study. Two moderate TRAEs (Presyncope and Gastroenteritis) were developed at 0.1 mg/kg cohort and resolved without intervention. One moderate drug eruption was developed and resolved at the placebo cohort (Table 4)

c. Two (2) out of 30 (6.7%) HM15912 treated subjects had anti-drug antibody (ADA) response. One (1) subject in 0.1 mg/kg dose cohort have treatment-induced ADA response on D30, that was specific to Fc domain of HM15912 and cross-reactive to endogenous GLP-2. One (1) subject in 1.0 mg/kg dose cohort had positive ADA response on baseline (pre-existing antibody before treatment), D17 and D44, but titer was not increased at all, not specific to Fc domain nor API portion of HM15912, and not cross-reactive to endogenous GLP-2. None of these ADA have neutralizing activity (Table 5). There were no specific antibodies to PEG portion of HM15912.

Table 3. Overall reported TEAEs

TEAE Category, Subject(%)	HM15912					Placebo (N=10)	All (N=40)
	0.05 mg/kg (N=6)	0.1 mg/kg (N=6)	0.5 mg/kg (N=6)	1.0 mg/kg (N=6)	1.5 mg/kg (N=6)		
Any TEAE	3 (50.0%)	3 (50.0%)	3 (50.0%)	4 (66.7%)	6 (100.0%)	6 (60.0%)	25 (62.5%)
Maximum Severity of TEAE							
Mild	2 (33.3%)	1 (16.7%)	3 (50.0%)	4 (66.7%)	5 (83.3%)	5 (50.0%)	20 (50.0%)
Moderate	1 (16.7%)	2 (33.3%)	0	0	1 (16.7%)	1 (10.0%)	5 (12.5%)
Severe	0	0	0	0	0	0	0
Any Serious TEAE	0	0	0	0	0	0	0
Any TEAE Related to Study Drug	3 (50.0%)	3 (50.0%)	2 (33.3%)	4 (66.7%)	4 (66.7%)	6 (60.0%)	22 (55.0%)
Maximum Severity of related TEAE							
Mild	3 (50.0%)	1 (16.7%)	2 (33.3%)	4 (66.7%)	4 (66.7%)	5 (50.0%)	19 (47.5%)
Moderate	0	2 (33.3%)	0	0	0	1 (10.0%)	3 (7.5%)
Severe	0	0	0	0	0	0	0
Any Serious related TEAE	0	0	0	0	0	0	0

Table 4. TRAEs by System Organ Class (SOC) and Preferred Terms (PT)

System Organ Class Preferred Term, Subject (%)	HM15912					Placebo (N=10)	All (N=40)
	0.05mg/kg (N=6)	0.1mg/kg (N=6)	0.5mg/kg (N=6)	1.0mg/kg (N=6)	1.5mg/kg (N=6)		
General disorders and administration site conditions	0	0	1 (16.7%)	2 (33.3%)	3 (50.0%)	2 (20.0%)	8 (20.0%)
Injection site bruising	0	0	0	2 (33.3%)	3 (50.0%)	2 (20.0%)	7 (17.5%)
Pyrexia	0	0	1 (16.7%)	0	0	0	1 (2.5%)
Investigations	0	0	1 (16.7%)	1 (16.7%)	2 (33.3%)	1 (10.0%)	5 (12.5%)
Neutrophil count decreased	0	0	1 (16.7%)	0	0	1 (10.0%)	2 (5.0%)
Aspartate aminotransferase increased	0	0	0	1 (16.7%)	0	0	1 (2.5%)
Blood bilirubin increased	0	0	0	0	1 (16.7%)	0	1 (2.5%)
Blood creatine phosphokinase increased	0	0	0	1 (16.7%)	0	0	1 (2.5%)
White blood cells urine positive	0	0	0	0	1 (16.7%)	0	1 (2.5%)
Nervous system disorders	2 (33.3%)	1 (16.7%)	0	0	0	1 (10.0%)	4 (10.0%)
Headache	2 (33.3%)	0	0	0	0	1 (10.0%)	3 (7.5%)
Presyncope	0	1 (16.7%)[a]	0	0	0	0	1 (2.5%)
Gastrointestinal disorders	0	0	0	3 (50.0%)	0	0	3 (7.5%)
Abdominal pain upper	0	0	0	1 (16.7%)	0	0	1 (2.5%)
Paraesthesia oral	0	0	0	1 (16.7%)	0	0	1 (2.5%)
Retching	0	0	0	1 (16.7%)	0	0	1 (2.5%)
Infections and infestations	0	2 (33.3%)	0	0	0	1 (10.0%)	3 (7.5%)
Nasopharyngitis	0	2 (33.3%)	0	0	0	1 (10.0%)	3 (7.5%)
Gastroenteritis	0	1 (16.7%)[a]	0	0	0	0	1 (2.5%)
Musculoskeletal and connective tissue disorders	1 (16.7%)	0	0	0	0	1 (10.0%)	2 (5.0%)
Back pain	1 (16.7%)	0	0	0	0	0	1 (2.5%)
Myalgia	0	0	0	0	0	1 (10.0%)	1 (2.5%)
Metabolism and nutrition disorders	0	0	0	1 (16.7%)	0	0	1 (2.5%)
Decreased appetite	0	0	0	1 (16.7%)	0	0	1 (2.5%)
Skin and subcutaneous tissue disorders	0	0	0	0	0	1 (10.0%)	1 (2.5%)
Fixed eruption	0	0	0	0	0	1 (10.0%)[a]	1 (2.5%)

a) Moderate TRAEs were developed and resolved at 0.1 mg/kg (Presyncope, Gastroenteritis) and at Placebo (Fixed eruption)

Table 5. Summary of ADA

Visit[a] Tier, Subjects (%) Result, Subjects (%)	HM15912, n (%)					Placebo (N=10)
	0.05 mg/kg (N=6)	0.1 mg/kg (N=6)	0.5 mg/kg (N=6)	1.0 mg/kg (N=6)	1.5 mg/kg (N=6)	
Baseline						
Tier 1 Results (Screening)						
Negative	6 (100.0)	5 (83.3)	6 (100.0)	5 (83.3)	4 (66.7)	8 (80.0)
Positive	0	1 (16.7)	0	1 (16.7)	2 (33.3)	2 (20.0)
Tier 2 Results (Confirmatory)						
Negative	0	1 (100)	0	0	2 (100)	2 (100)
Positive	0	0	0	1 (100)	0	0
Day 10						
Tier 1 Results (Screening)						
Negative	6 (100.0)	5 (83.3)	6 (100.0)	-	-	6 (60.0)
Positive	0	1 (16.7)	0	-	-	0
Tier 2 Results (Confirmatory)						
Negative	0	1 (100)	0	-	-	0
Positive	0	0	0	-	-	0

Table 5. Summary of ADA (Cont'd)

Visit[a] Tier, Subjects (%) Result, Subjects (%)	HM15912, n (%)					Placebo (N=10)
	0.05 mg/kg (N=6)	0.1 mg/kg (N=6)	0.5 mg/kg (N=6)	1.0 mg/kg (N=6)	1.5 mg/kg (N=6)	
Day 17						
Tier 1 Results (Screening)						
Negative	-	-	-	5 (83.3)	4 (66.7)	3 (30.0)
Positive	-	-	-	1 (16.7)	2 (33.3)	1 (10.0)
Tier 2 Results (Confirmatory)						
Negative	-	-	-	0	2 (100)	1 (100)
Positive	-	-	-	1 (100)	0	0
Day 30						
Tier 1 Results (Screening)						
Negative	6 (100.0)	5 (83.3)	6 (100.0)	-	-	6 (60.0)
Positive	0	1 (16.7)	0	-	-	0
Tier 2 Results (Confirmatory)						
Negative	0	0	0	-	-	0
Positive	0	1 (100)	0	-	-	0
Day 44						
Tier 1 Results (Screening)						
Negative	-	-	-	4 (66.7)	5 (83.3)	2 (20.0)
Positive	-	-	-	2 (33.3)	1 (16.7)	2 (20.0)
Tier 2 Results (Confirmatory)						
Negative	-	-	-	1 (50.0)	1 (100)	2 (100.0)
Positive	-	-	-	1 (50.0)	0	0

a) Immunogenicity sampling was performed at baseline, D10 and D30 in cohort 1 to 3. In cohort 4 to 5, immunogenicity sampling was performed at baseline, D17 and D44.

- **Serum Surrogate Biomarker**

a. The Fasting Plasma Citrulline levels gradually increased up to D17 from 0.5 mg/Kg. Between 0.5 mg/Kg and 1.5 mg/Kg, the Fasting Plasma Citrulline levels are crowded and overlapped. From 0.5 mg/Kg, the Fasting Plasma Citrulline was detected after D30 (Figure 3).

Figure 3. Change from baseline of Fasting Plasma Citrulline

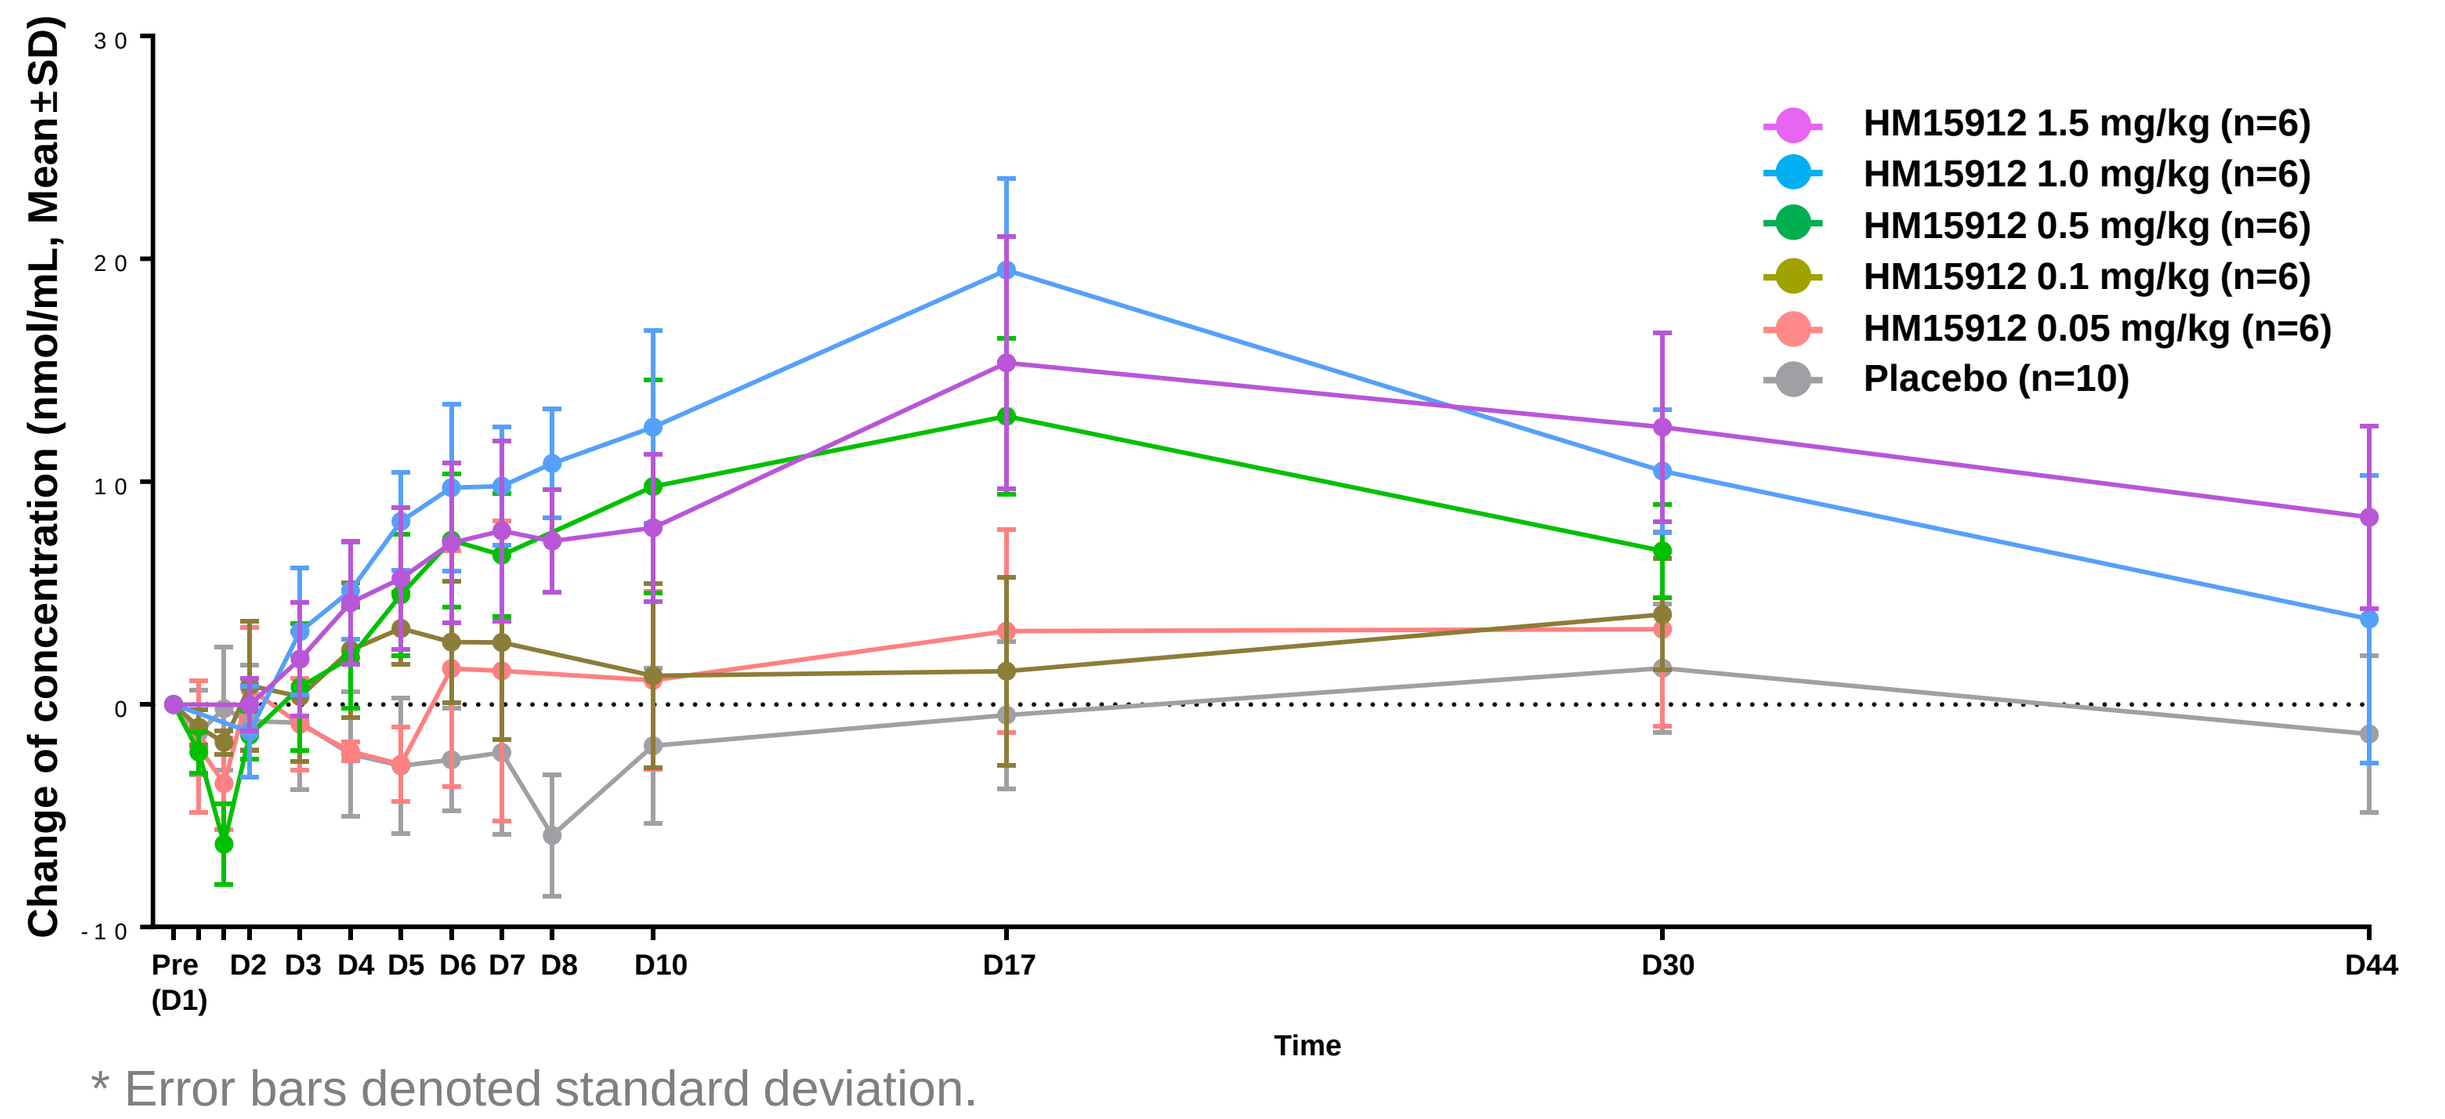
HM15912 1.5 mg/kg (n=6)
HM15912 1.0 mg/kg (n=6)
HM15912 0.5 mg/kg (n=6)
HM15912 0.1 mg/kg (n=6)
HM15912 0.05 mg/kg (n=6)
Placebo (n=10)
Change of concentration (nmol/mL, Mean±SD)
Pre (D1) D2 D3 D4 D5 D6 D7 D8 D10 D17 D30 D44
Time

* Error bars denoted standard deviation.

CONCLUSIONS

Single administration of HM15912 showed a favorable and tolerable safety profile with the healthy volunteers in wide dose range. The fasting plasma Citrulline level was elevated from the 0.5 mg/Kg. The pharmacokinetic profile, over up to 167 hours of mean $t_{1/2}$, supports a potential for every 2-weeks or 4-weeks dosing frequency.

Disclosures: None.

ASPEN 2021 NUTRITION SCIENCE & PRACTICE CONFERENCE
ASPEN 2021 Nutrition Science & Practice Conference; March 20-23, 2021.

Contact Information: Seungjae Baek, Chief Medical Officer, Hanmi Pharm. Co., Ltd.
E-mail: seungjae.baek@hanmi.co.kr
Phone: +82-2-410-9039

Hanmi Pharm. Co., Ltd.