Robust Anti-obesity Effect and Mechanistic Insights of HM15275, A Novel Long-acting GLP-1/GIP/Glucagon Triple Agonist in Animal Model of Obesity

Sang Hyun Park, Yohan Kim, Jung Kuk Kim, Seon Myeong Lee, Eunjin Park, Dahae Lee, Jeong A. Kim, SungHee Hong, Sungmin Bae, Sang Hyun Lee, In Young Choi
Hanmi Pharmaceutical Co., Ltd., Seoul, Republic of Korea

Hanmi

Poster
774-P

Introduction

Introduction and Objective: Obesity has reached epidemic proportions globally, posing a critical public health crisis. To address this, HM15275, a novel long-acting GLP-1/GIP/Glucagon triple agonist was developed, demonstrating potent weight loss and glycemic control in prior preclinical studies. The present study further investigates the effects of HM15275 on body weight and body composition over an extended period and its underlying mechanisms.

Methods: The effect of HM15275 on body weight and body composition were assessed in DIO mice. To investigate the weight loss mechanism of HM15275, pair-fed and energy balance studies were conducted, and mesenteric white adipose tissue (mWAT) was subjected to immunohistochemistry. Tirzepatide (TZP) and retatrutide (RETA, in-house synthesis) served as comparative controls.

Results: In DIO mice, HM15275 treatment for 3 weeks resulted in a greater weight loss (-39.9% vs. D0) than TZP (-25.3% vs. D0). Switching from TZP to HM15275 at week 2 led to further weight loss. Mechanistically, a pair-fed study revealed that HM15275 induced both food intake-dependent and independent weight loss in which, latter was driven by increased energy expenditure. HM15275 also enhanced expression of PGC-1α and UCP-1 in mWAT, being more pronounced than TZP, which indicates stronger fat browning effect. Furthermore, longer treatment of HM15275 for 6 weeks significantly reduced more body weight than RETA. Notably, greater fat mass reduction and no difference in lean mass by HM15275 contributed to more improved weight loss quality compared to RETA.

Conclusion: In conclusion, HM15275 demonstrates potent weight loss with improved weight loss quality in DIO mice. Also, comprehensive elucidation of the underlying mechanism of action has been demonstrated. Clinical translation of such findings had been evaluated in phase 1, 4-week multiple ascending dose (MAD) study in obese patients.

Background

HM15275 is a novel long-acting GLP-1/GIP/Glucagon triple agonist conjugated with fatty acid moiety, optimally designed for treatment of obesity and relative complications.

HM15275

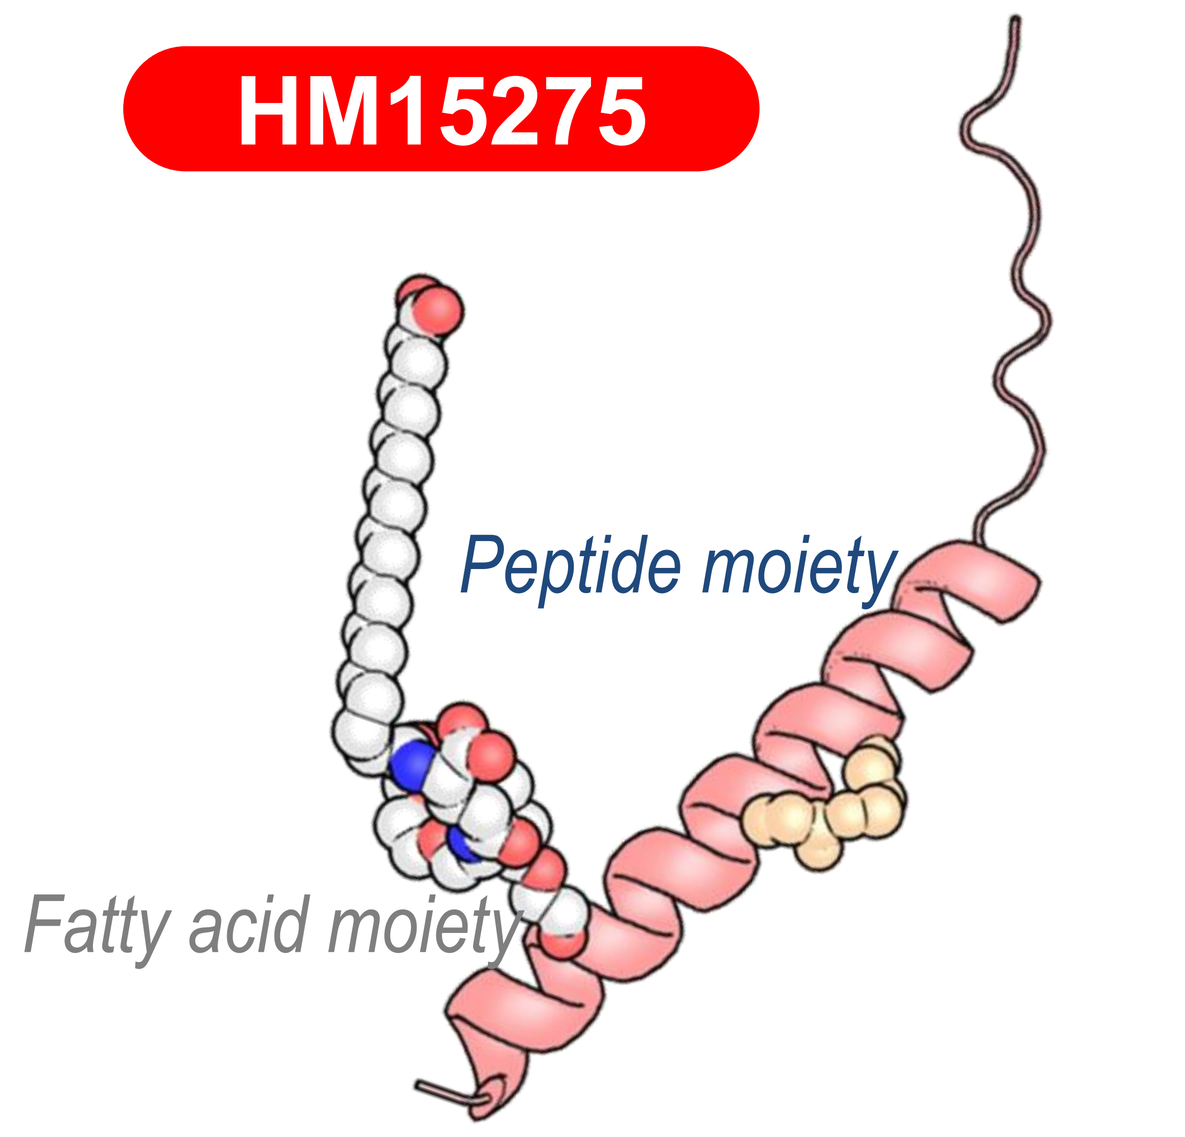
Peptide moiety
Fatty acid moiety

- Designed and optimized to maximize body weight reduction (activity balance)
- The extended half-life is sufficient for weekly dosing
- Additional CVRM benefits expected by proper utilization of glucagon
- P1 study in United States completed

GLP-1 **GIP**
➢Weight loss by appetite regulation
➢Indirect effects from BW loss and BG control for CVRM benefits

\+

Glucagon
➢Weight loss by energy expenditure
➢Direct tissue effect for CVRM benefits

Experimental methods

(a) Body weight efficacy study design

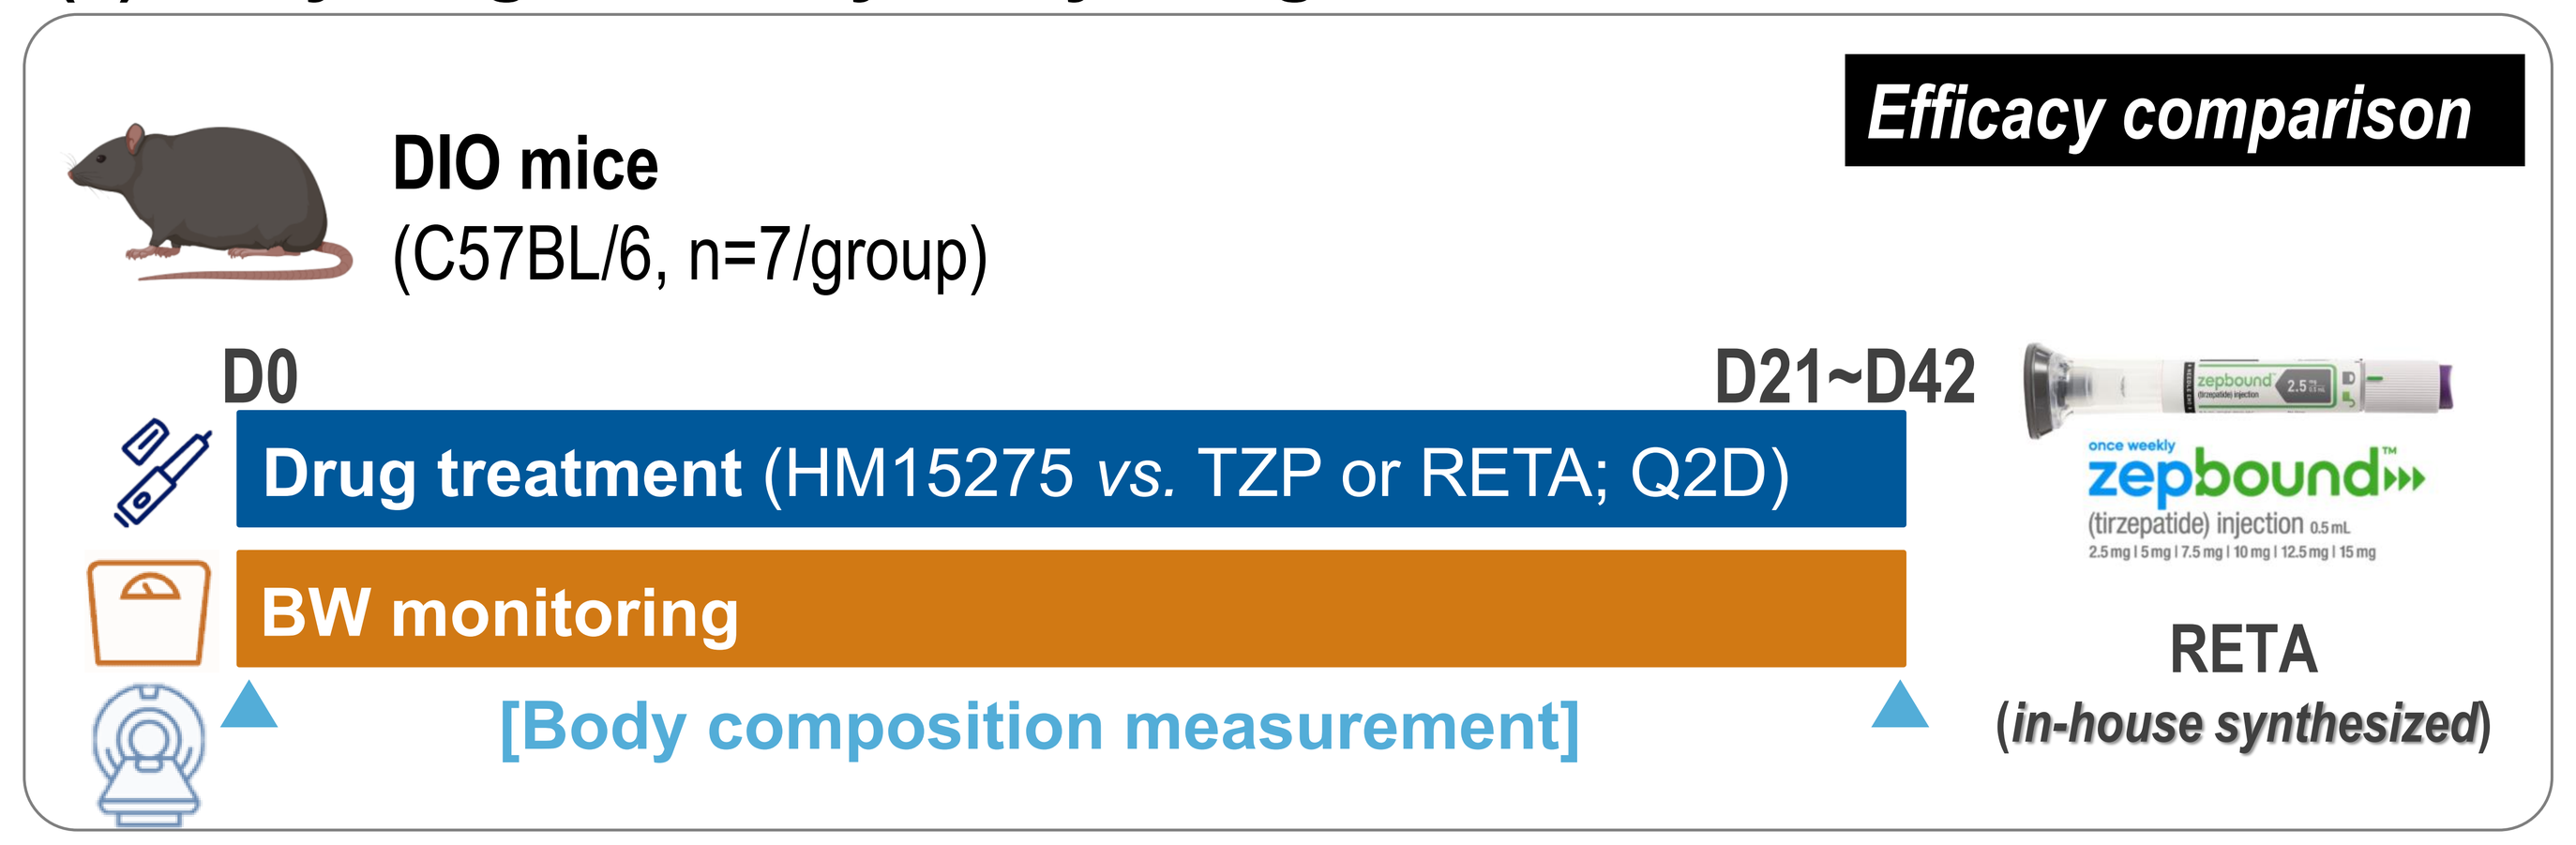
DIO mice
(C57BL/6, n=7/group)
Efficacy comparison
D0
D21~D42
Drug treatment (HM15275 vs. TZP or RETA; Q2D)
BW monitoring
[Body composition measurement]
zepbound
RETA
(in-house synthesized)

(b) Switching study design

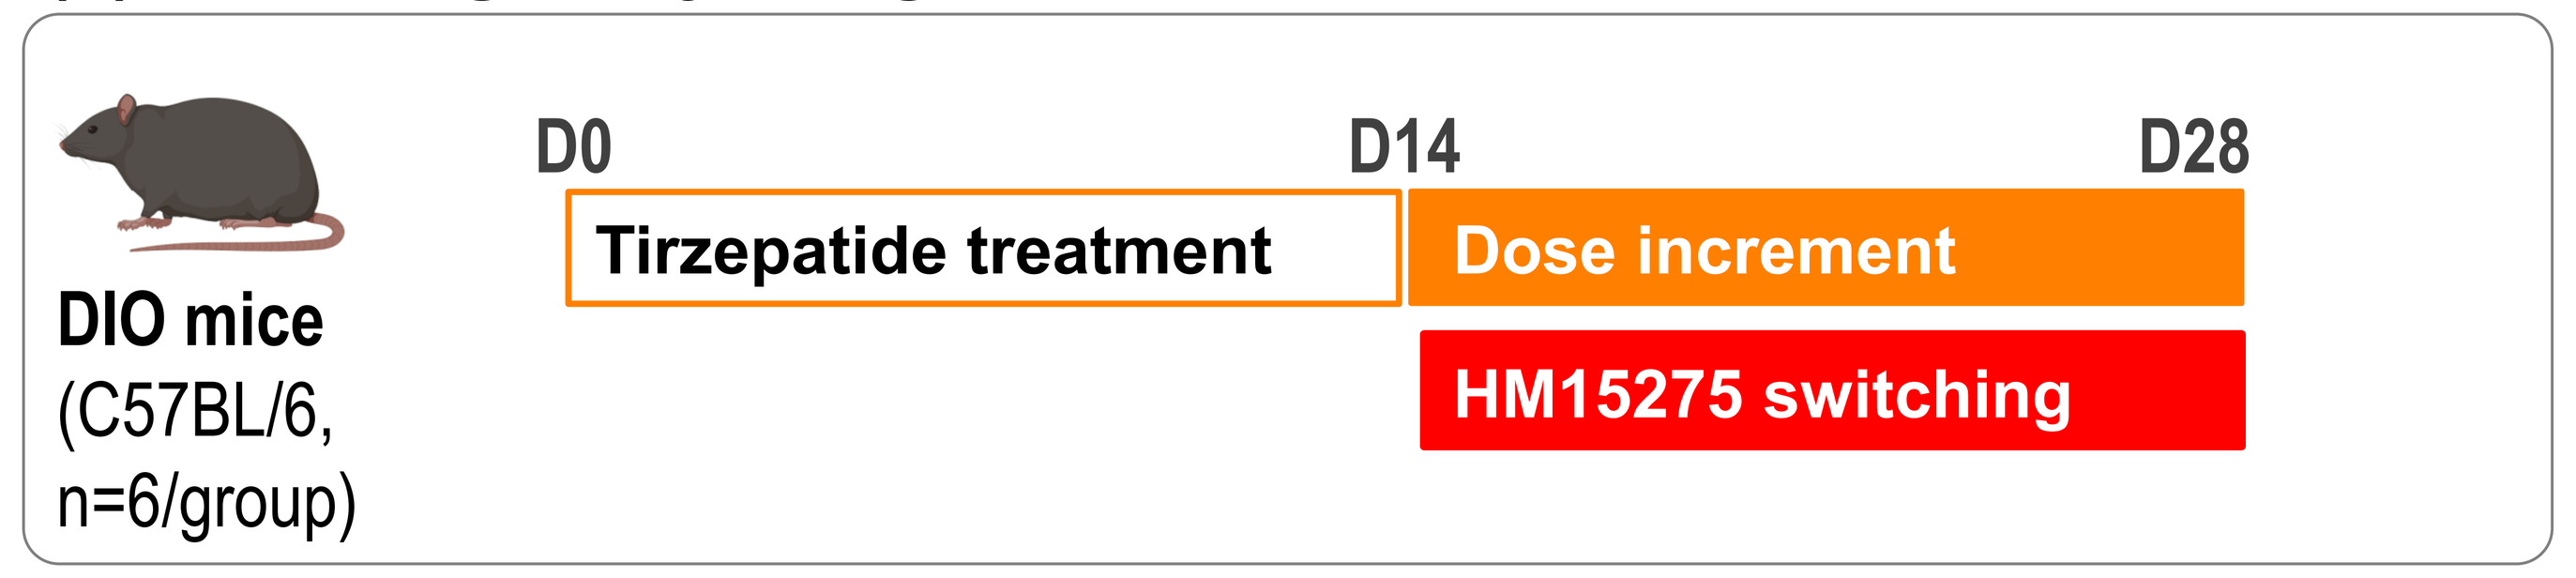
DIO mice
(C57BL/6,
n=6/group)
D0
D14
D28
Tirzepatide treatment
Dose increment
HM15275 switching

(c) Energy expenditure (EE) study design

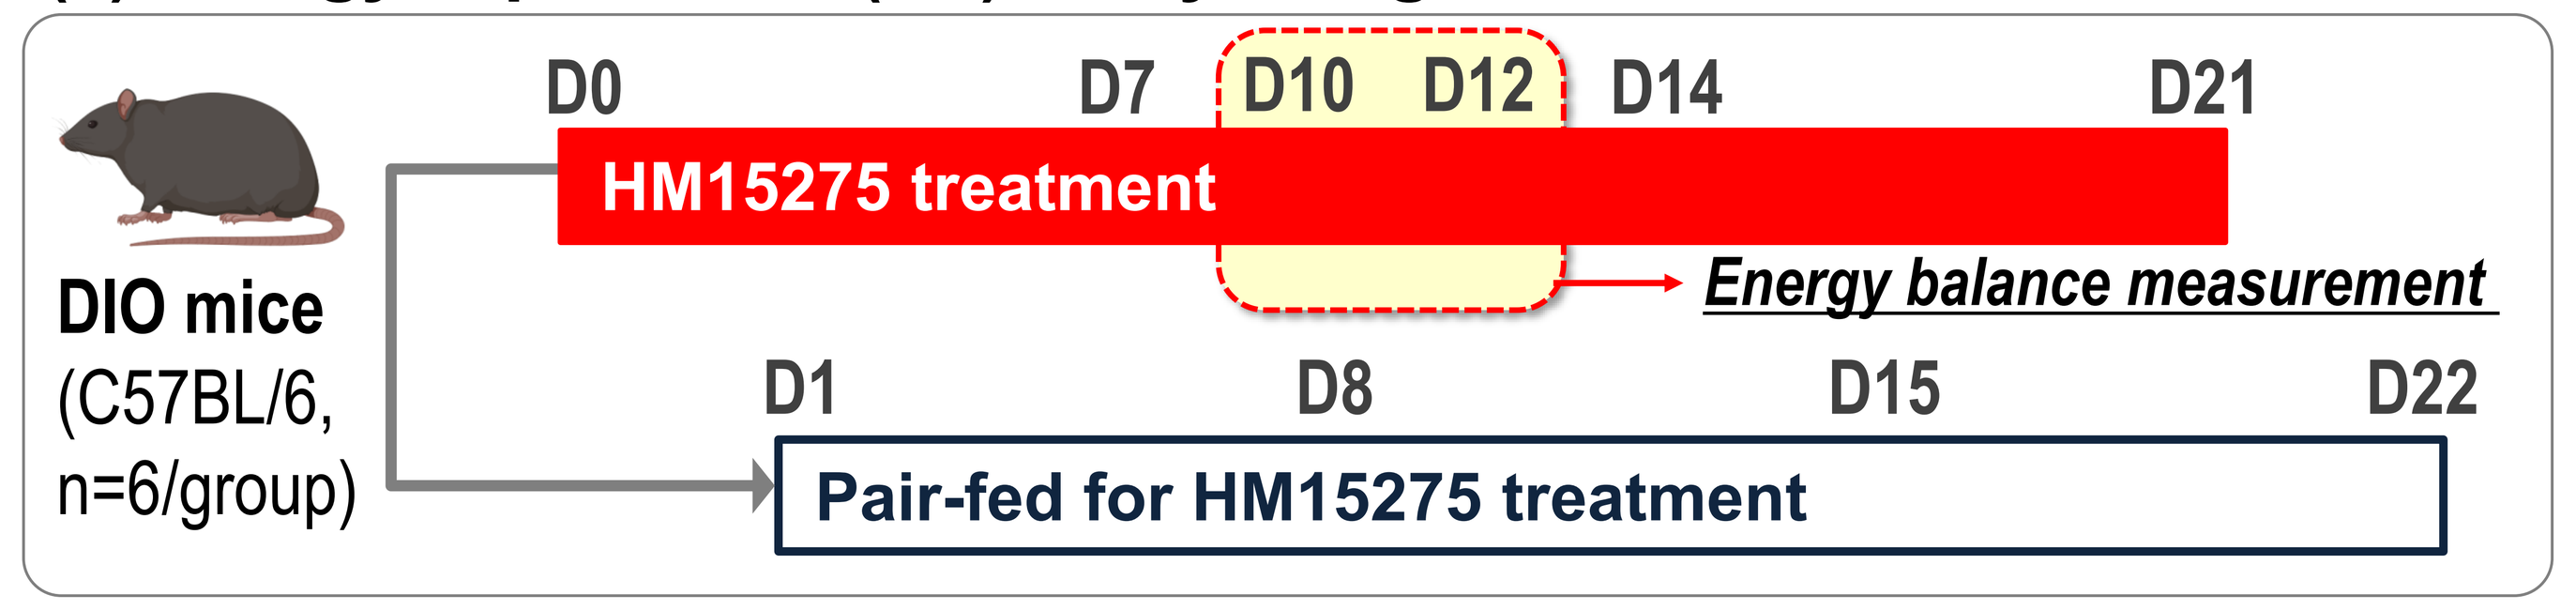
DIO mice
(C57BL/6,
n=6/group)
D0
D7
D10
D12
D14
D21
HM15275 treatment
Energy balance measurement
D1
D8
D15
D22
Pair-fed for HM15275 treatment

Superior weight loss and quality in DIO mice

Figure 1. Changes in body weight and body composition in DIO mice

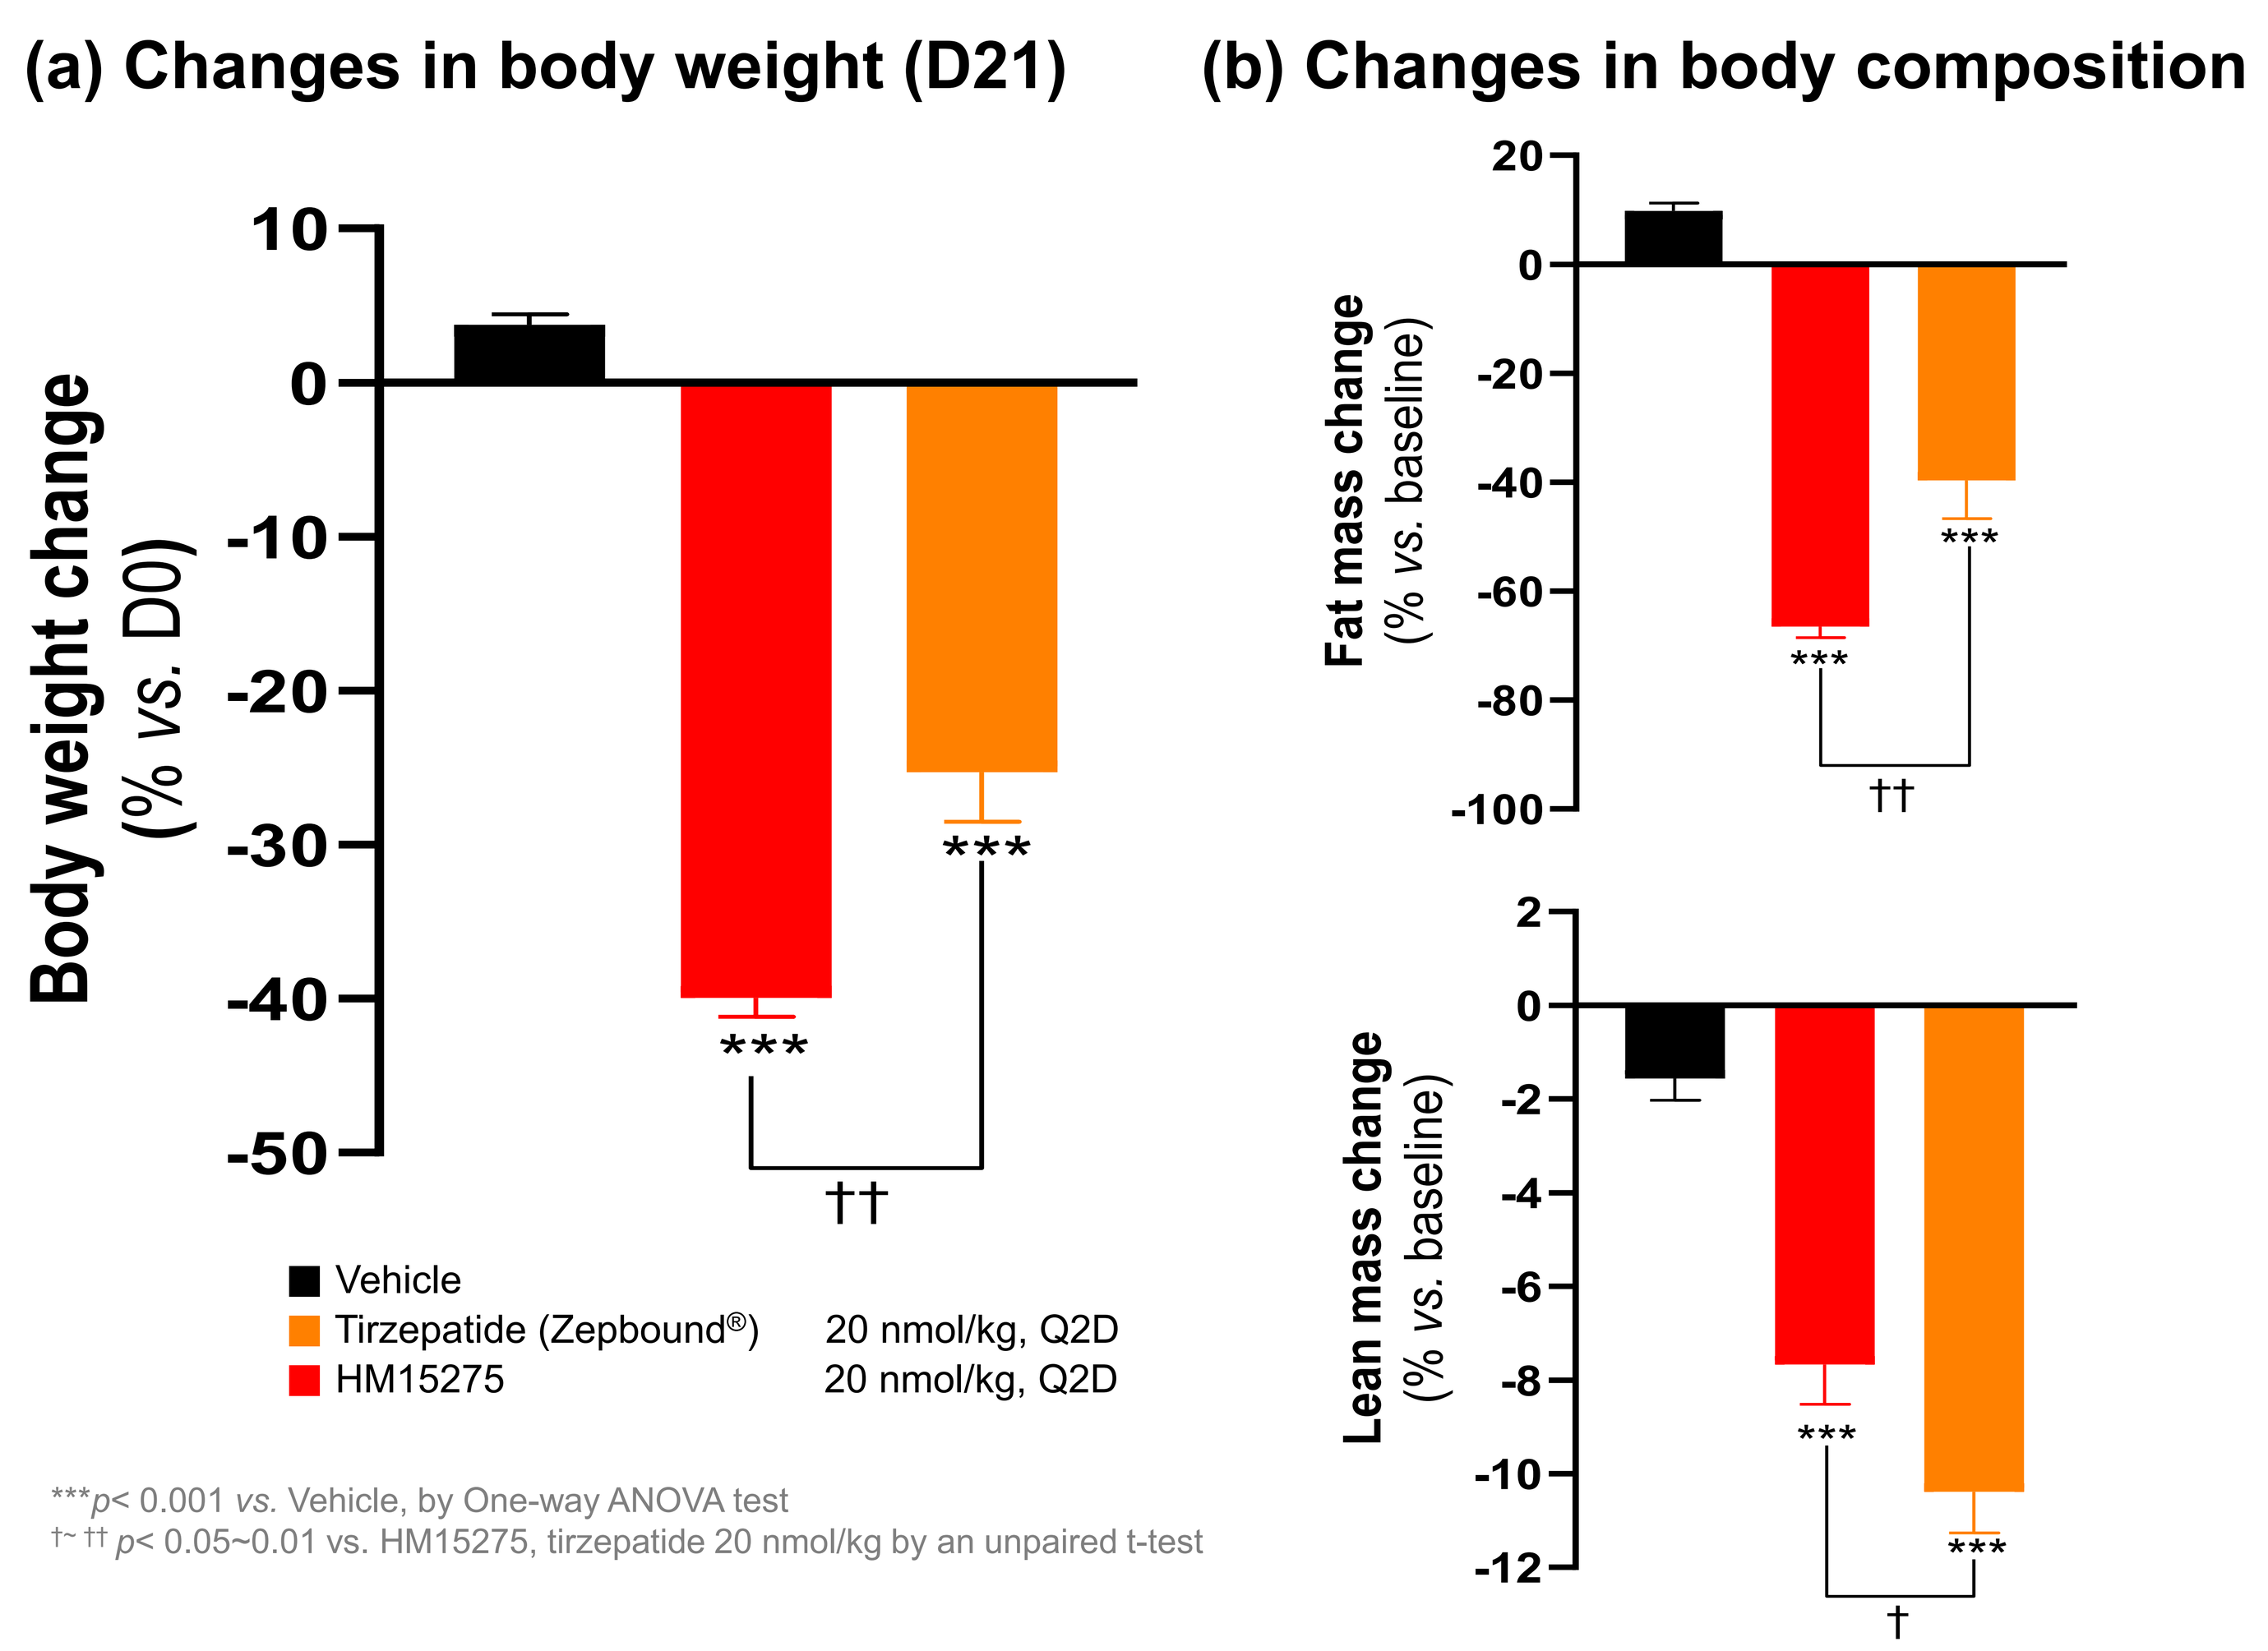
(a) Changes in body weight (D21)
(b) Changes in body composition
Body weight change (% vs. D0)
Fat mass change (% vs. baseline)
Lean mass change (% vs. baseline)
Vehicle
Tirzepatide (Zepbound®) 20 nmol/kg, Q2D
HM15275 20 nmol/kg, Q2D
***p< 0.001 vs. Vehicle, by One-way ANOVA test
†~††p< 0.05~0.01 vs. HM15275, tirzepatide 20 nmol/kg by an unpaired t-test

➢In DIO mice, HM15275 mainly attributed to more fat mass reduction and notably, despite greater body weight loss than tirzepatide, more favorable weight loss quality (WLQ) was demonstrated by HM15275.

Switching promoted additional weight loss benefits

Figure 2. Changes in body weight after switching in DIO mice

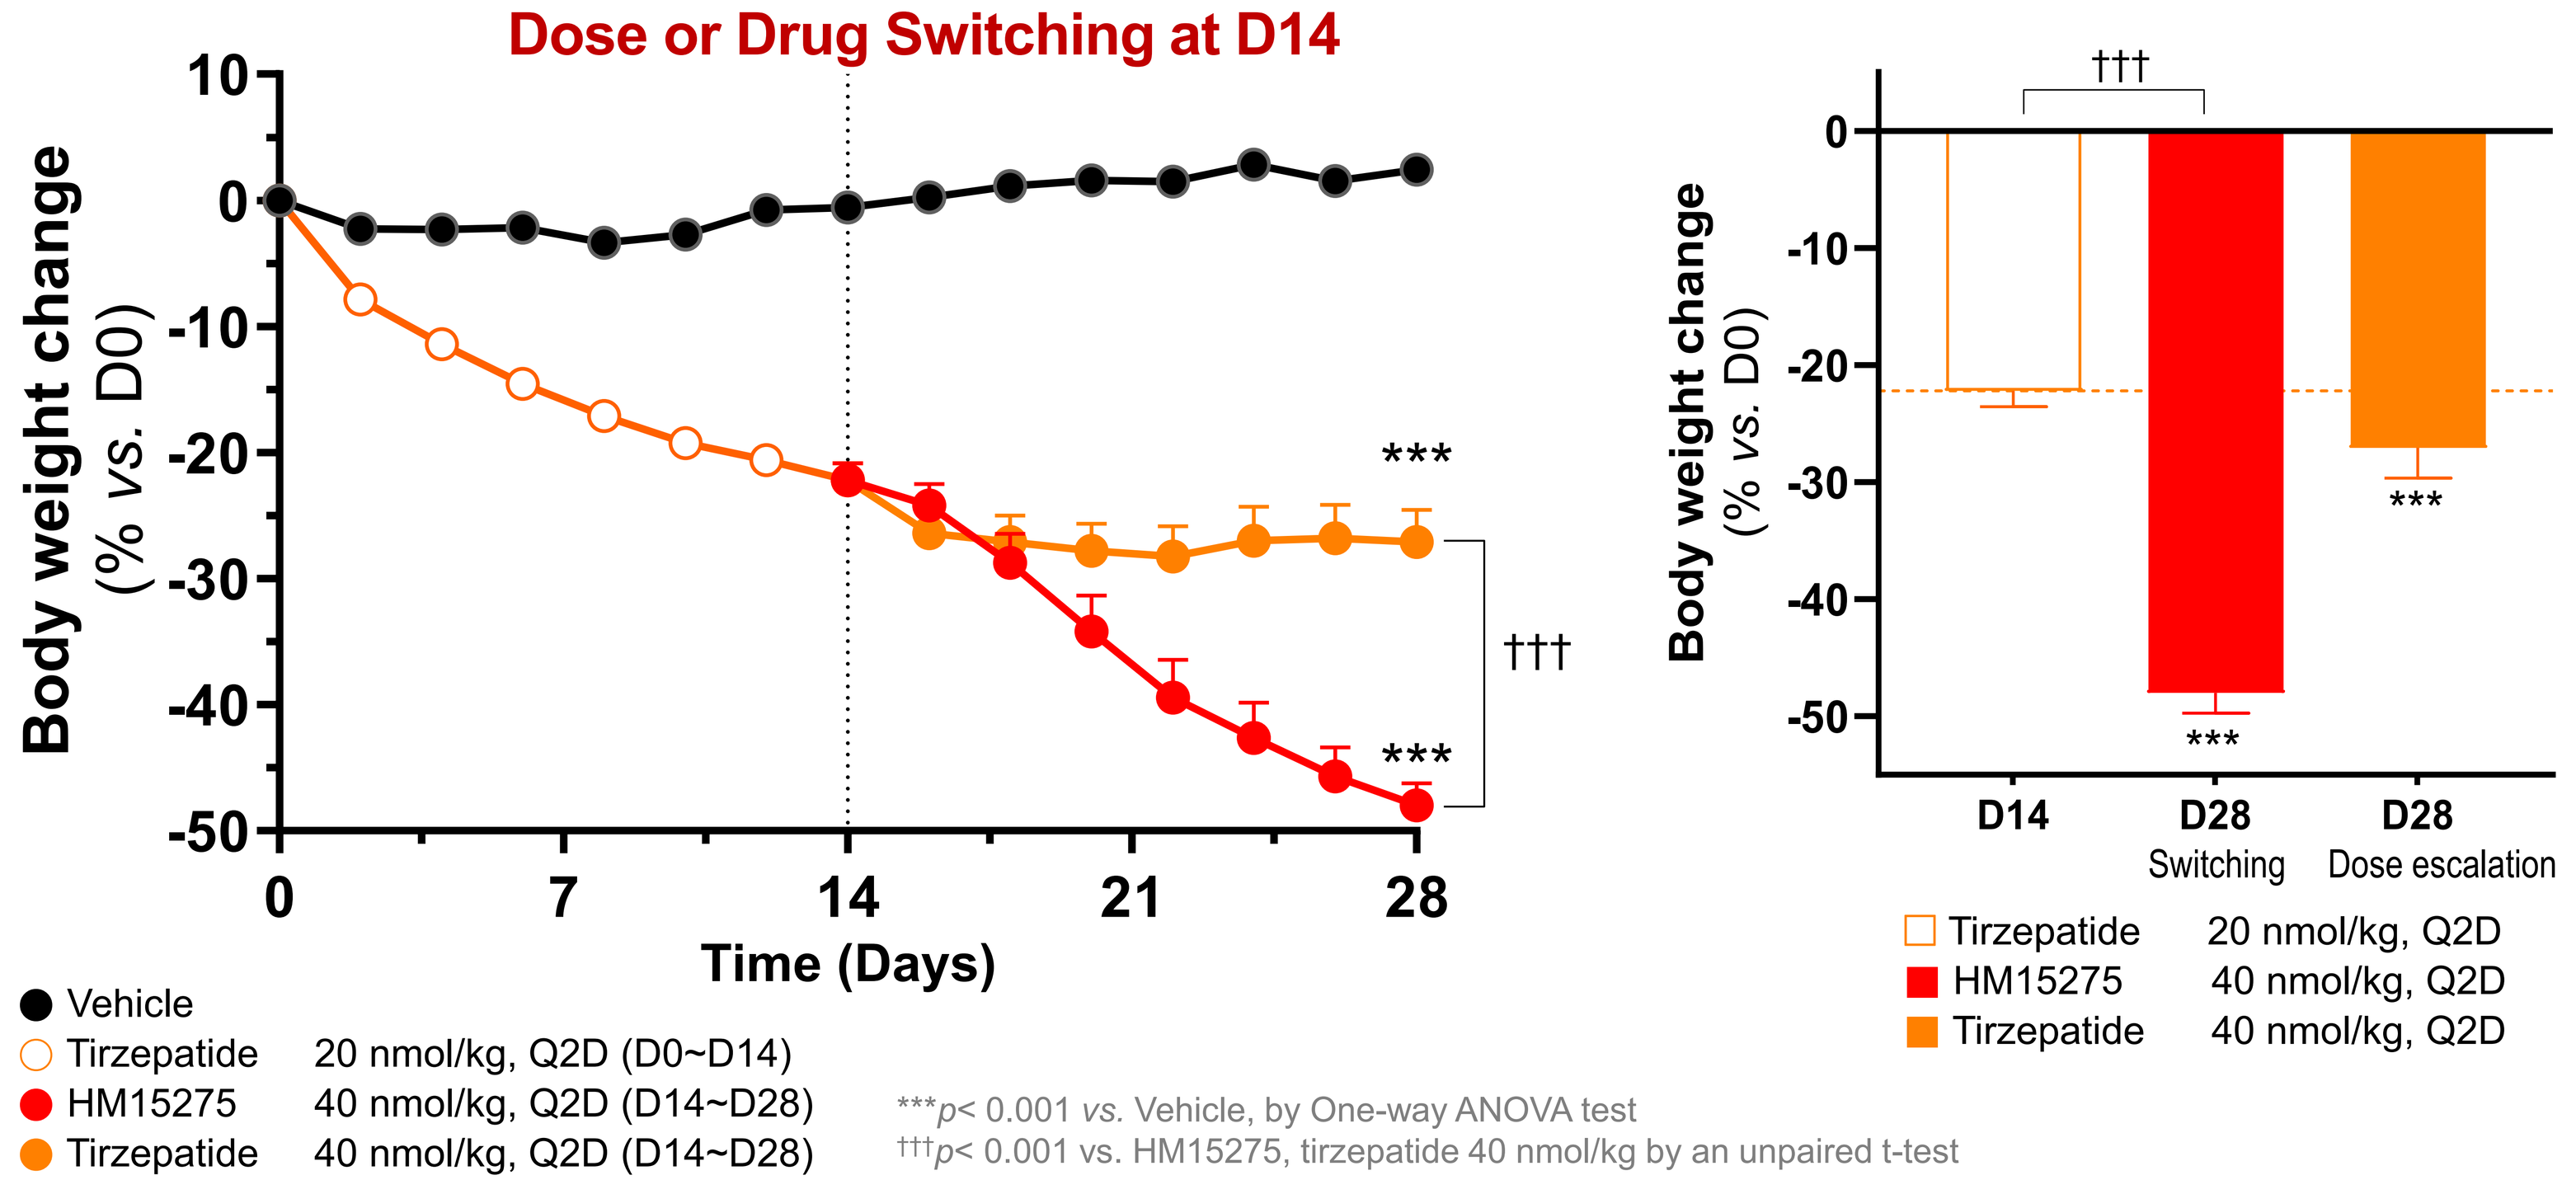
Dose or Drug Switching at D14
Body weight change (% vs. D0)
Time (Days)
D14
D28 Switching
D28 Dose escalation
Vehicle
Tirzepatide 20 nmol/kg, Q2D (D0~D14)
HM15275 40 nmol/kg, Q2D (D14~D28)
Tirzepatide 40 nmol/kg, Q2D (D14~D28)
***p< 0.001 vs. Vehicle, by One-way ANOVA test
†††p< 0.001 vs. HM15275, tirzepatide 40 nmol/kg by an unpaired t-test
Tirzepatide 20 nmol/kg, Q2D
HM15275 40 nmol/kg, Q2D
Tirzepatide 40 nmol/kg, Q2D

➢In DIO mice, additional weight loss was observed when tirzepatide treatment was switched to HM15275 at day 14, while dose escalation of tirzepatide only showed marginal additive effects.

Mode of action – Energy expenditure

Figure 3. Mechanism evaluation for body weight reduction by pair-fed DIO mice

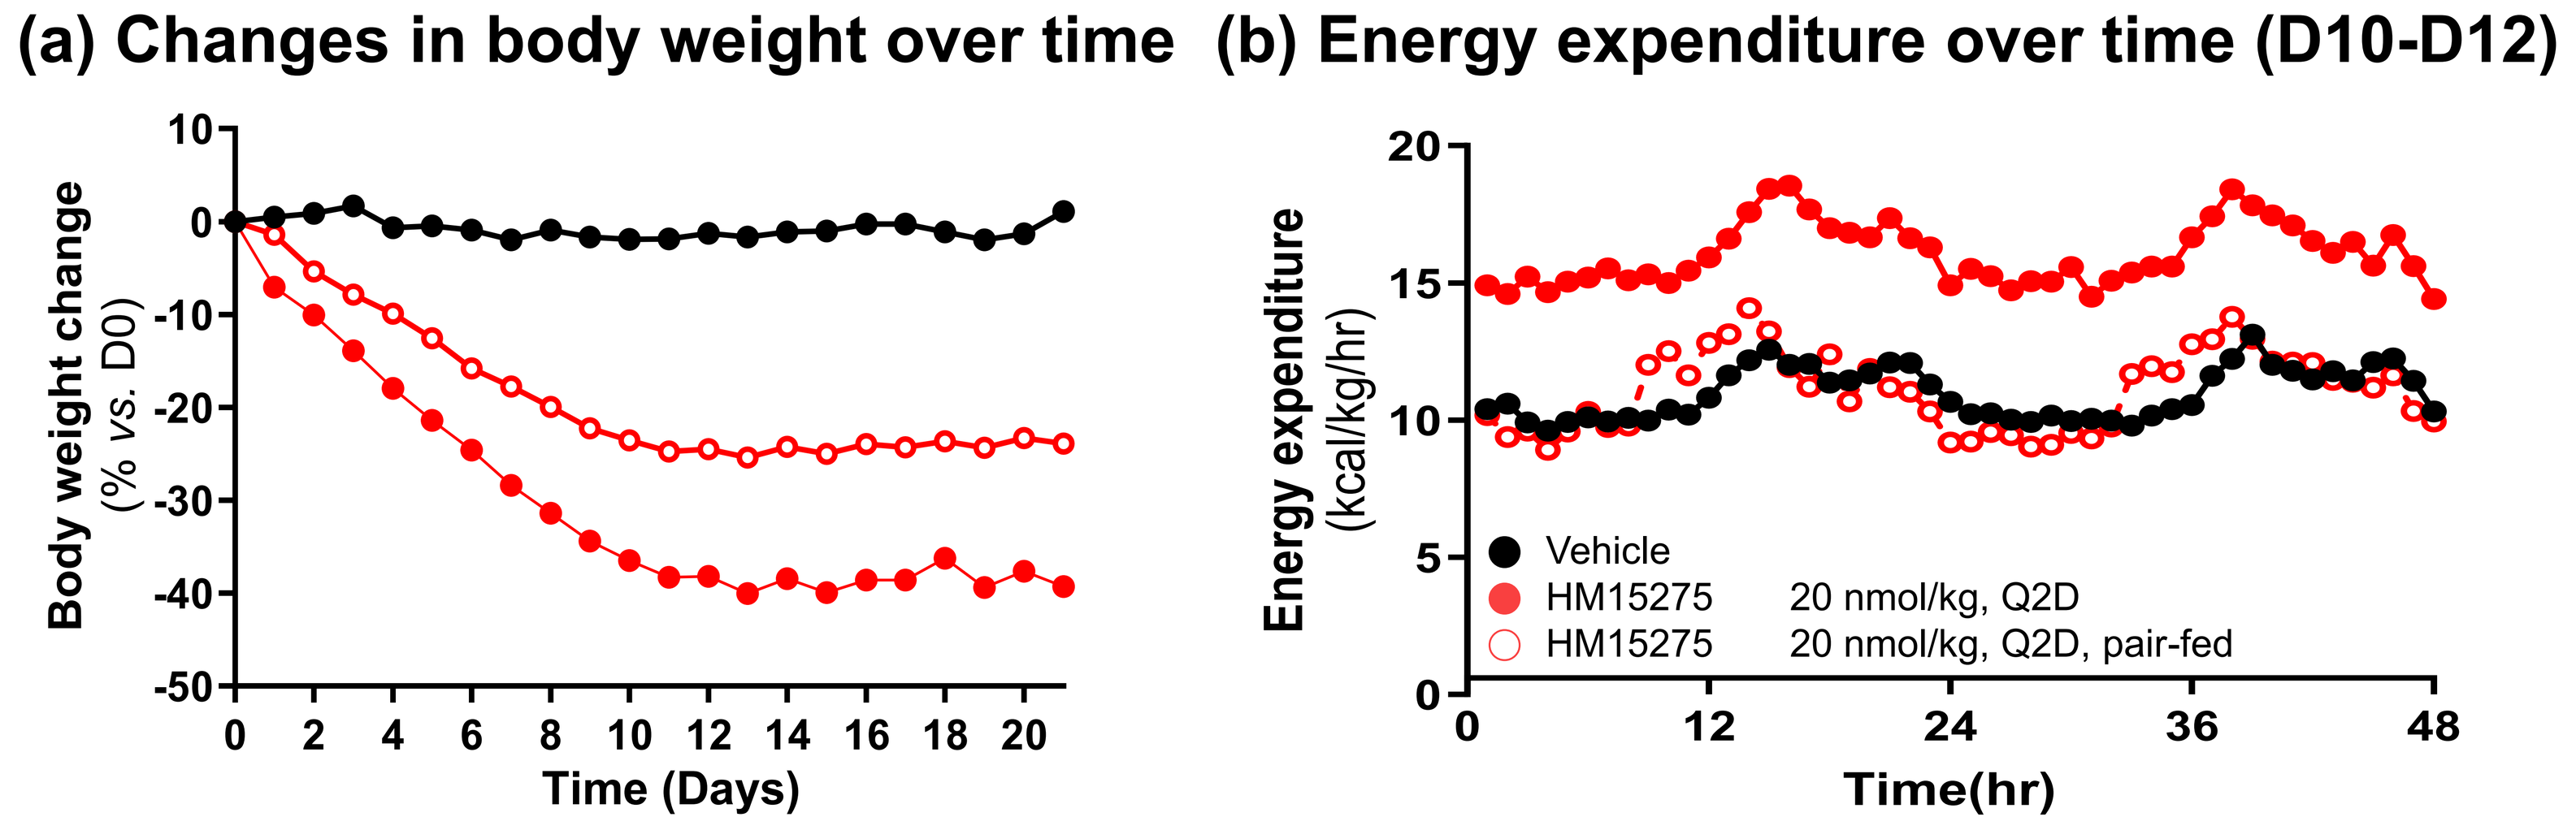
(a) Changes in body weight over time
(b) Energy expenditure over time (D10-D12)
Body weight change (% vs. D0)
Time (Days)
Energy expenditure (kcal/kg/hr)
Time(hr)
Vehicle
HM15275 20 nmol/kg, Q2D
HM15275 20 nmol/kg, Q2D, pair-fed

Figure 4. Immunohistochemistry and gene expression for energy expenditure

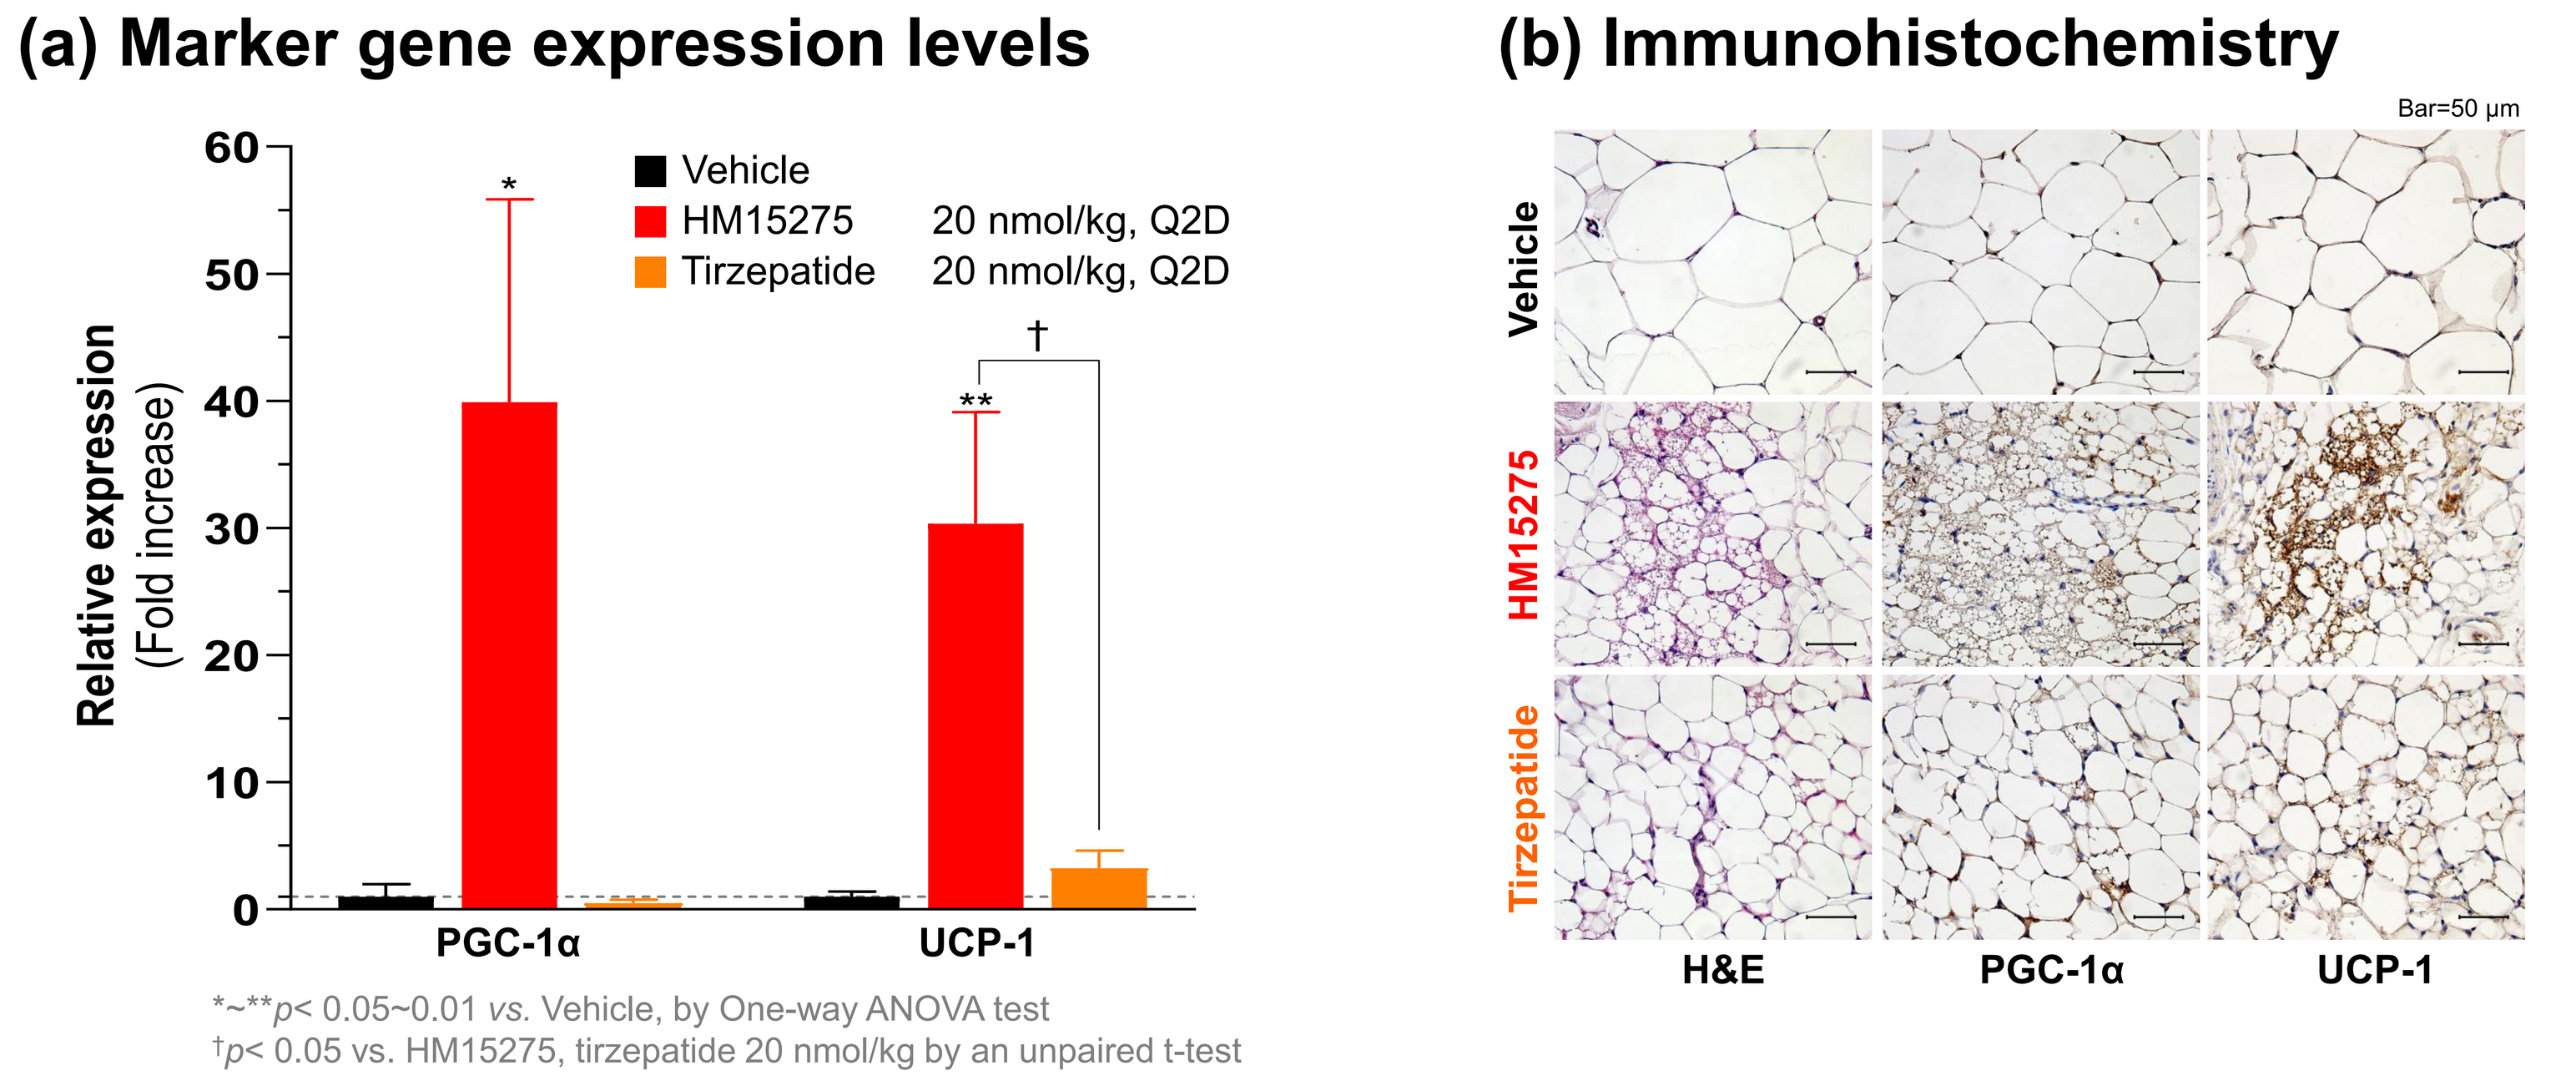
(a) Marker gene expression levels
(b) Immunohistochemistry
Relative expression (Fold increase)
Vehicle
HM15275 20 nmol/kg, Q2D
Tirzepatide 20 nmol/kg, Q2D
PGC-1α
UCP-1
*~**p< 0.05~0.01 vs. Vehicle, by One-way ANOVA test
†p< 0.05 vs. HM15275, tirzepatide 20 nmol/kg by an unpaired t-test
Bar=50 μm
Vehicle
HM15275
Tirzepatide
H&E
PGC-1α
UCP-1

➢ In DIO mice, HM15275 significantly decreased lipid droplet size in white adipose tissue. Also, increased expression and tissue staining of PGC-1α and UCP-1 suggest that HM15275 may exert a stronger fat browning effect compared to tirzepatide.

Long-term weight loss effect in DIO mice

Figure 5. Changes in body weight and body composition over time in DIO mice

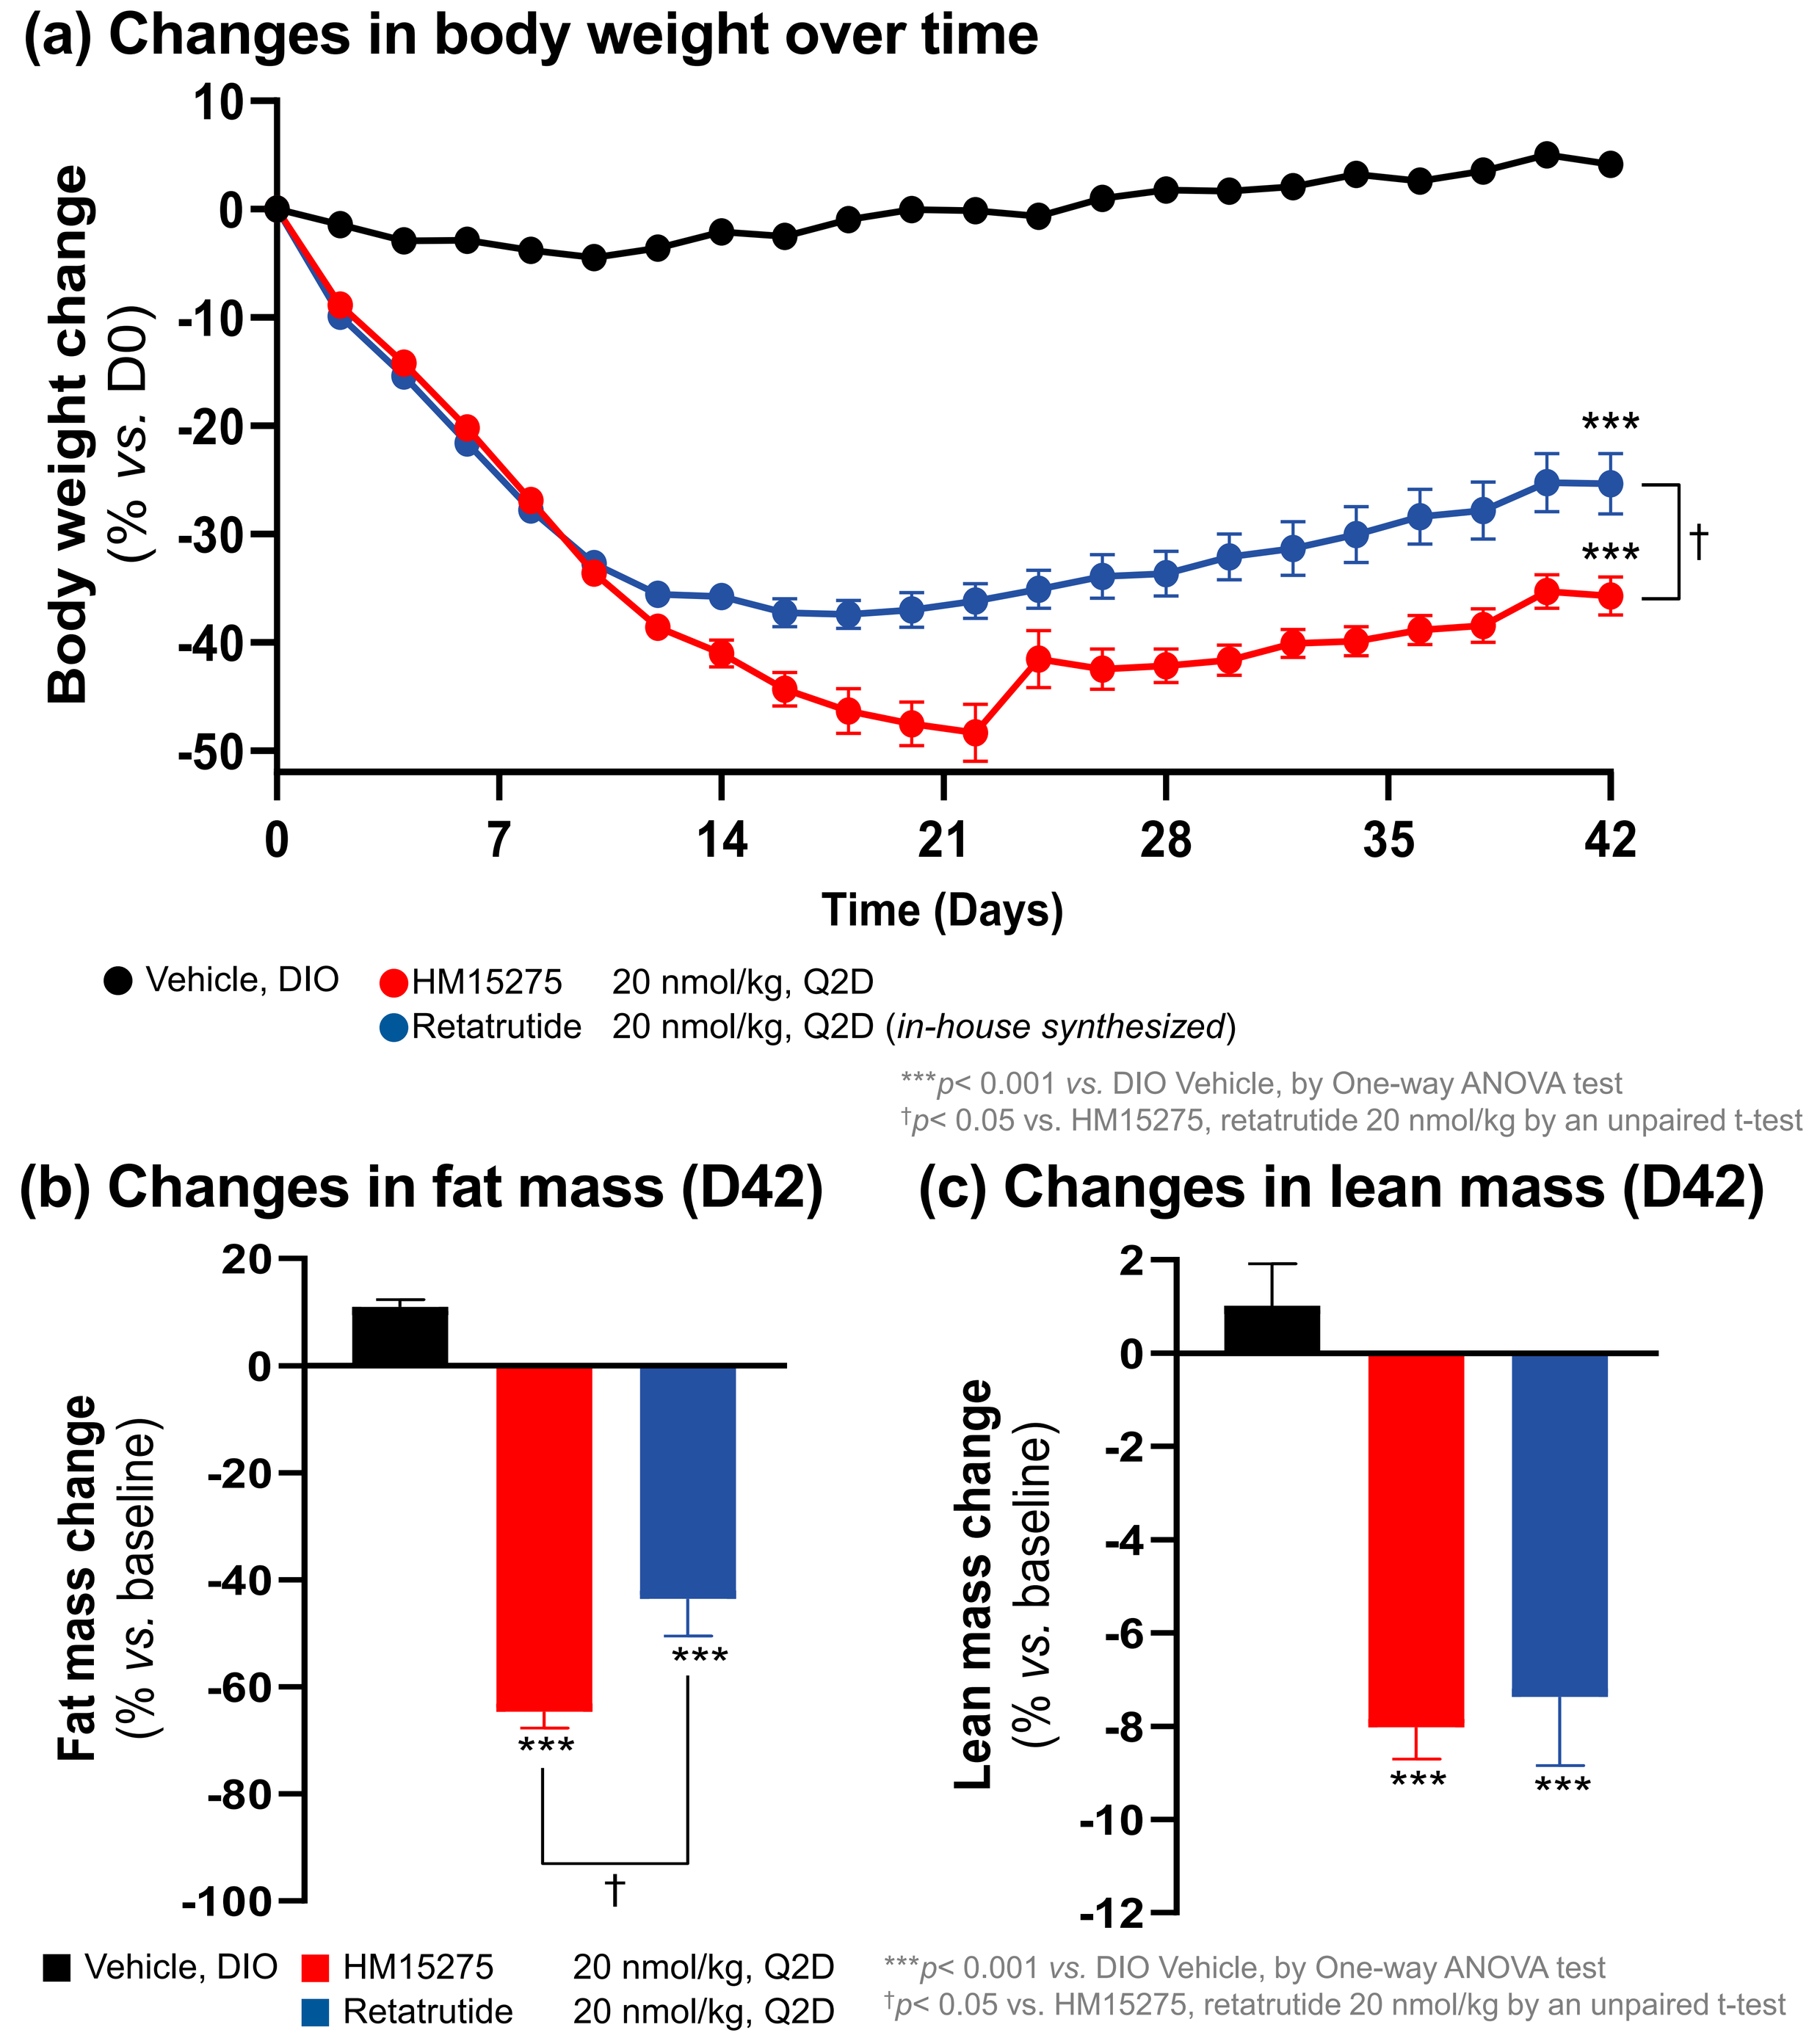
(a) Changes in body weight over time
Body weight change (% vs. D0)
Time (Days)
Vehicle, DIO
HM15275 20 nmol/kg, Q2D
Retatrutide 20 nmol/kg, Q2D (in-house synthesized)
***p< 0.001 vs. DIO Vehicle, by One-way ANOVA test
†p< 0.05 vs. HM15275, retatrutide 20 nmol/kg by an unpaired t-test
(b) Changes in fat mass (D42)
(c) Changes in lean mass (D42)
Fat mass change (% vs. baseline)
Lean mass change (% vs. baseline)
Vehicle, DIO
HM15275 20 nmol/kg, Q2D
Retatrutide 20 nmol/kg, Q2D
***p< 0.001 vs. DIO Vehicle, by One-way ANOVA test
†p< 0.05 vs. HM15275, retatrutide 20 nmol/kg by an unpaired t-test

➢In DIO mice, HM15275 significantly reduced body weight and enhanced fat-to-lean quality regulation with more therapeutic benefits compared to retatrutide.

Concluding Remarks

- HM15275, a novel long-acting triple agonist, is designed to treat obesity by optimally modulating GLP-1, GIP, and glucagon receptor activities.
- Previous *in-vitro* and PK studies clearly demonstrated the proper target engagement and long-acting profile (2024 ADA, 2024 ObesityWeek).
- In DIO mice, HM15275 demonstrated significant body weight reduction with superior efficacy and quality of weight loss compared to incretin-based drugs. The weight loss was driven by both reduced food intake and increased energy expenditure.
- Notably, additional weight loss benefit was observed after switching from tirzepatide to HM15275, indicating superior efficacy of HM15275.
- Long-term treatment also led to favorable changes in body weight and composition, outperforming another triple agonist candidate.
- Phase 1 obesity trial has been completed, and phase 2 trial is planned.
- Please also note additional posters presenting Hanmi's pipeline assets: HM15275 (755-P: Preclinical; 1980-LB: Phase 1 clinical) and HM17321 (842-P, 843-P, and 886-P).

American Diabetes Association. American Diabetes Association (ADA), June 20-23, 2025 Hanmi Hanmi Pharmaceutical Co., Ltd.