Potential of a novel long-acting glucagon analogue, HM15136, for the treatment of obesity

Hanmi

766-P

Jae Hyuk Choi[1], Jung Kuk Kim[1], Jin Young Kim[1], Sun Myung Lee[1], CheongByeol Shin[1], Jong Soo Lee[1], Sang Hyun Lee[1], and In Young Choi[1]
[1]Hanmi Pharm. Co., Ltd, Seoul, South Korea

BACKGROUND

- Poor solubility and stability of native GCG limited its long-term pharmacologic investigation
- To overcome these limitation, HM15136 was developed, and its therapeutic potential in obesity was evaluated

Acute GCGR activation condition

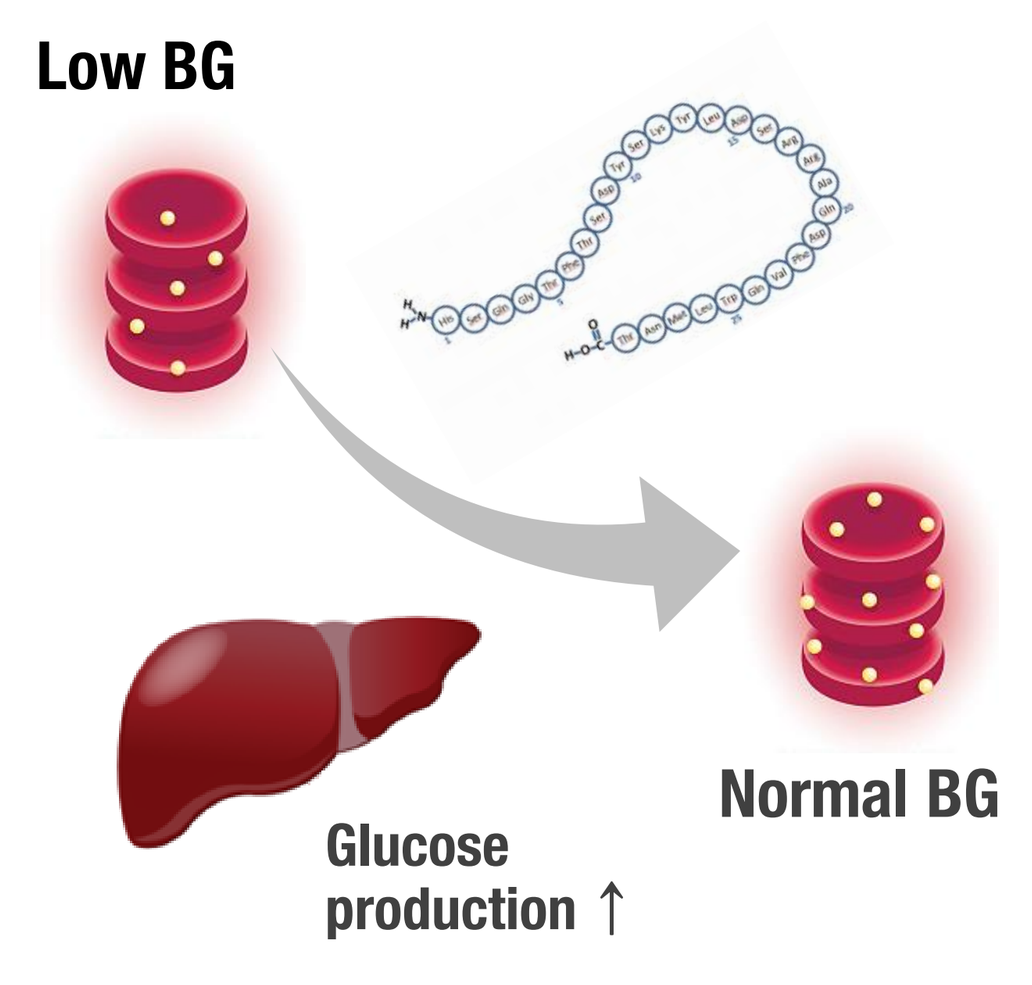
Low BG
Glucose production ↑
Normal BG

- **Hepatic glucose output ↑**
- Insufficient for
 : Lipid metabolism improvement
 : Energy expenditure induction

Sustained GCGR activation in obese condition

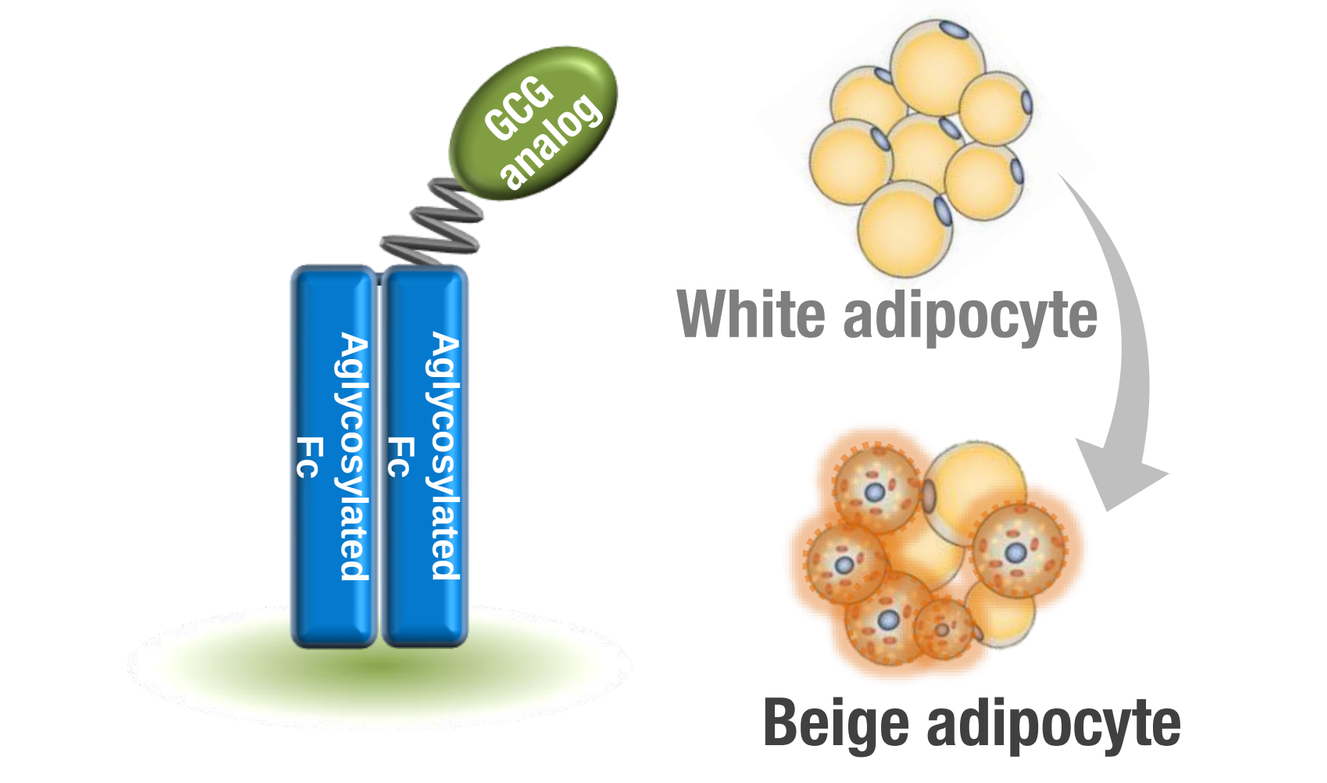
GCG analog
Aglycosylated Fc
Aglycosylated Fc
White adipocyte
Beige adipocyte

- Enhanced lipolysis & energy expenditure by fat browning → **Efficient BWL**
- Hepatic lipid clearance ↑
 → **Steatosis & blood lipotoxicity improvement**
- BG production sources ↓ (glycogen, lipid)
- INS sensitivity improvement
 → **BG normalization**

Optimized for
: ***Long-term use for obesity***

AIMS

- **We hypothesized that sustained GCG receptor (GCGR) activation could provide substantial BWL along with insulin resistance (IR) improvement and considerably reduced hyperglycemic risk**
- **To investigate our hypothesis, we developed HM15136, a novel long-acting GCG analog, with improved solubility and stability,**
- **The present study evaluated the PK/PD properties of HM15136.**

METHODS

- Solubility of HM15136 and its solution stability were evaluated using liquid chromatography analysis. The intrinsic activity of HM15136 was determined by measuring intracellular cAMP accumulation in CHO cells stably or transiently expressing GCGR from various species (*data not shown*). For PK assessments, blood samples were collected after single subcutaneous or intravenous administration of HM15136 in ICR mice, SD rats, and beagle dogs, followed by quantification of blood HM15136 concentration using an ELISA.
- To evaluate the potential therapeutic efficacy in obesity, diet-induced obesity (DIO) mice were chronically administered with HM15136, and BW, BG, and food intake (FI) were monitored.
- To investigate the FI inhibition-independent BWL mechanism, BW change in DIO mice was compared with liraglutide under pair-fed controlled condition.

RESULTS

Improved solubility & physical stability of HM15136

Table 1. Solubility of HM15136

Test article	Solubility at pH 7.0
Glucagon	0.03 mg/mL
HM15136	**≥ 150 mg/mL**

Figure 1. Physical stability of HM15136 in PBS (pH 7.0) at 25°C

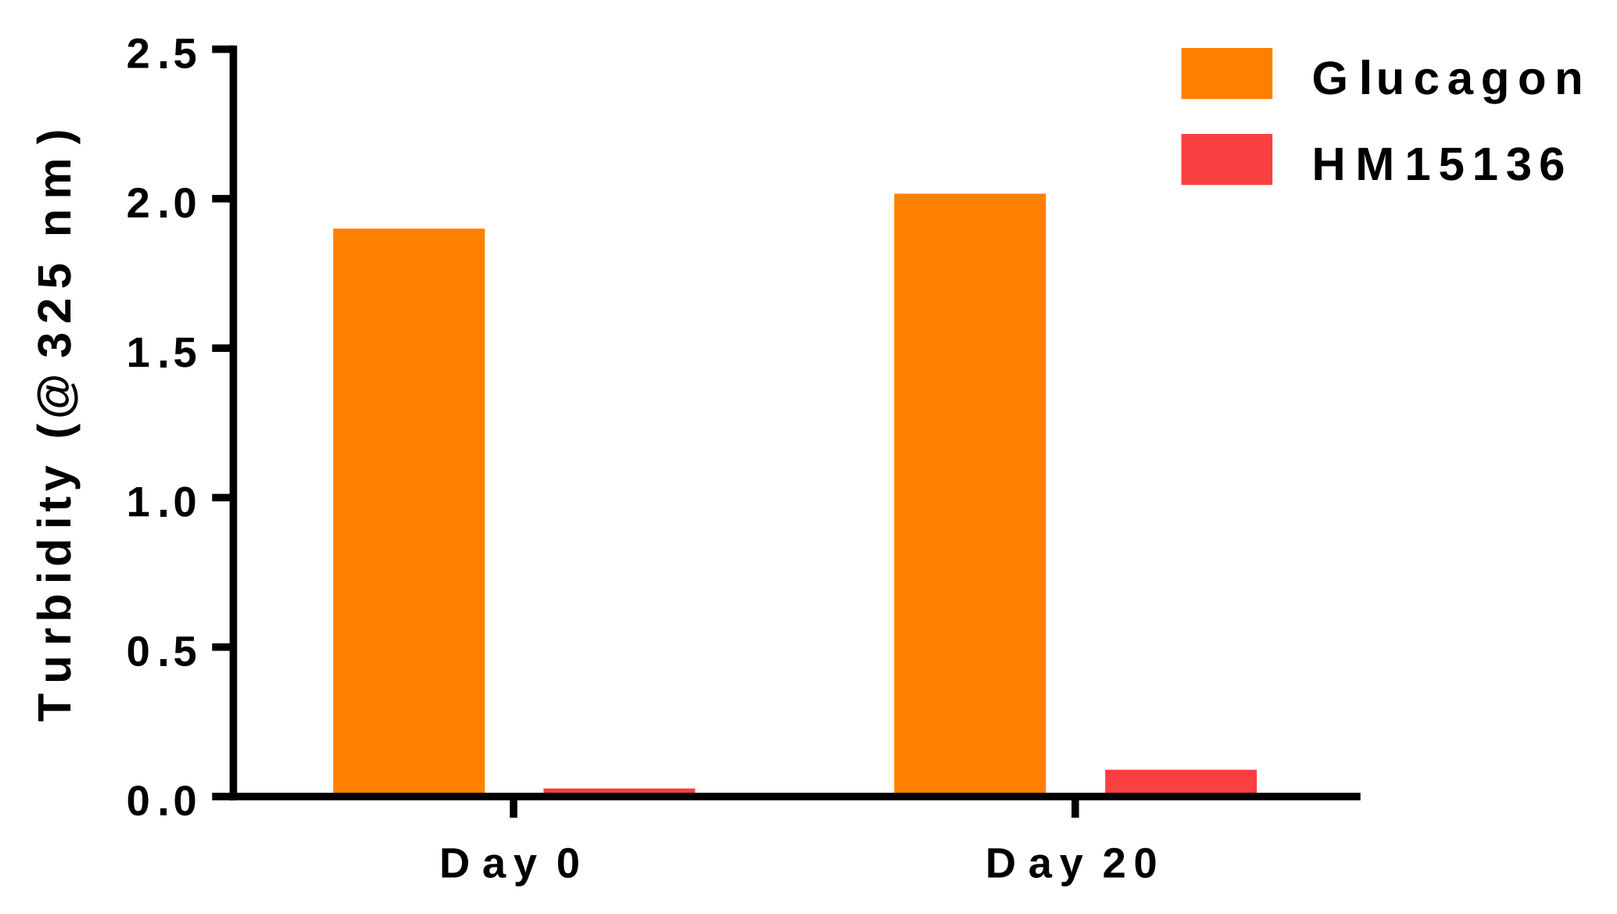
Turbidity (@ 325 nm)
2.5
2.0
1.5
1.0
0.5
0.0
Day 0
Day 20
Glucagon
HM15136

➢HM15136 shows improved solubility and physical stability at physiological pH

PK properties of HM15136

Figure 2. PK in various animal species (n=3), and human PK prediction

(a) ICR mice

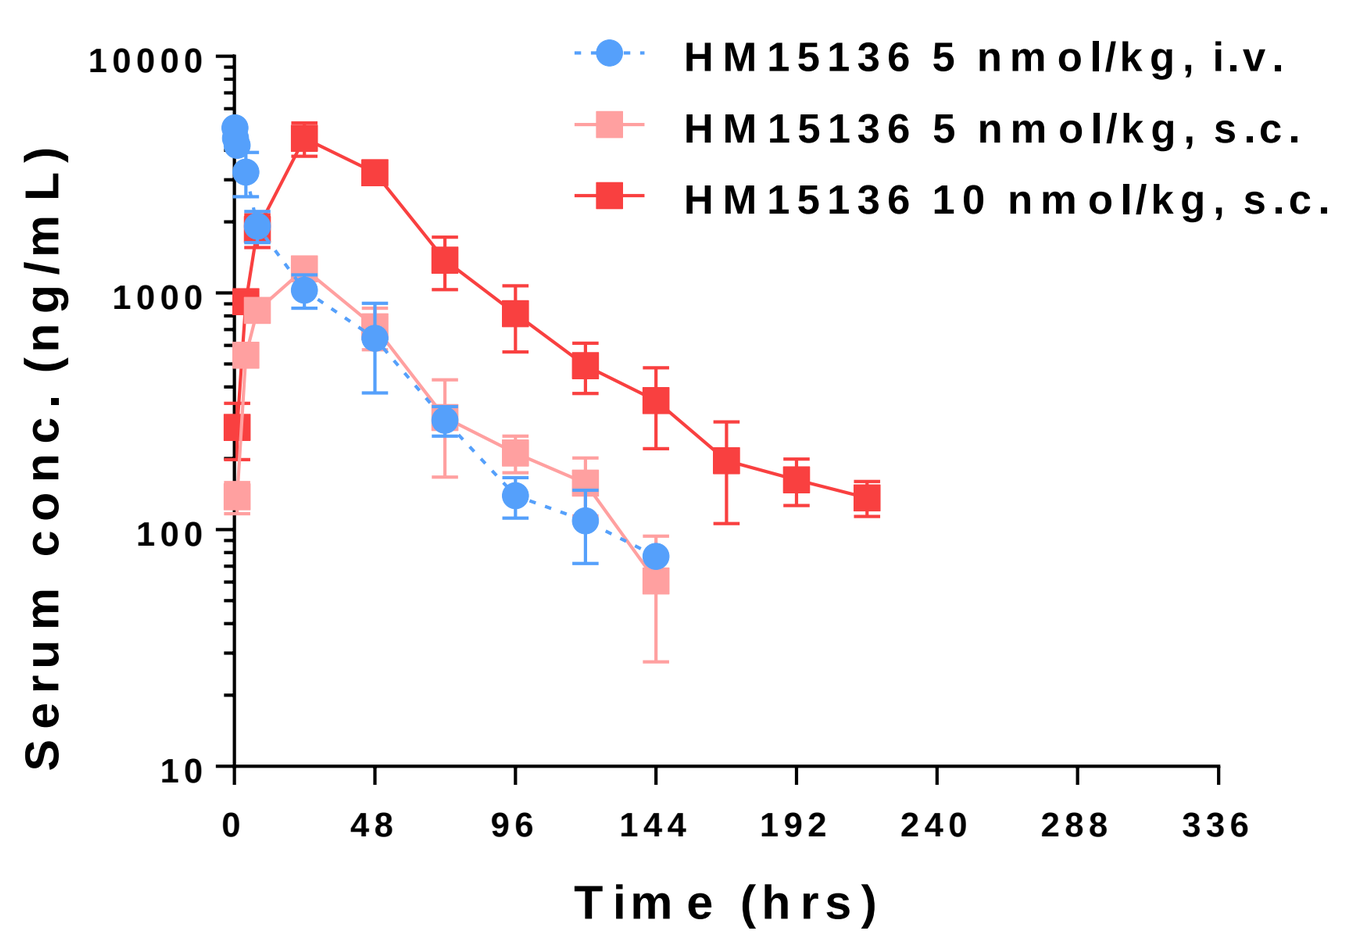
HM15136 5 nmol/kg, i.v.
HM15136 5 nmol/kg, s.c.
HM15136 10 nmol/kg, s.c.
Serum conc. (ng/mL)
Time (hrs)

(b) SD rats

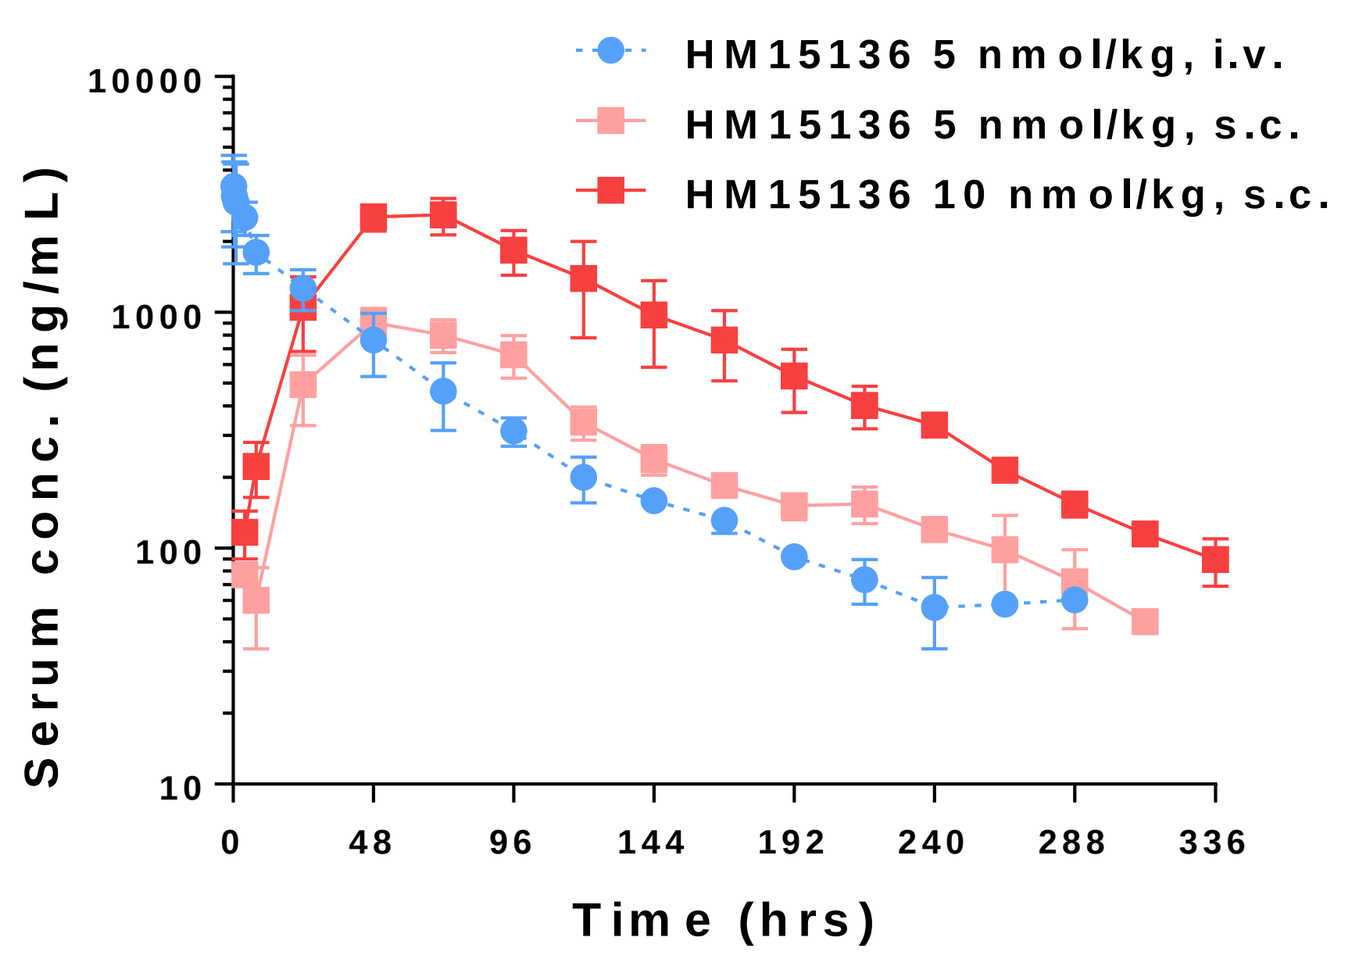
HM15136 5 nmol/kg, i.v.
HM15136 5 nmol/kg, s.c.
HM15136 10 nmol/kg, s.c.
Serum conc. (ng/mL)
Time (hrs)

(c) Beagle dogs

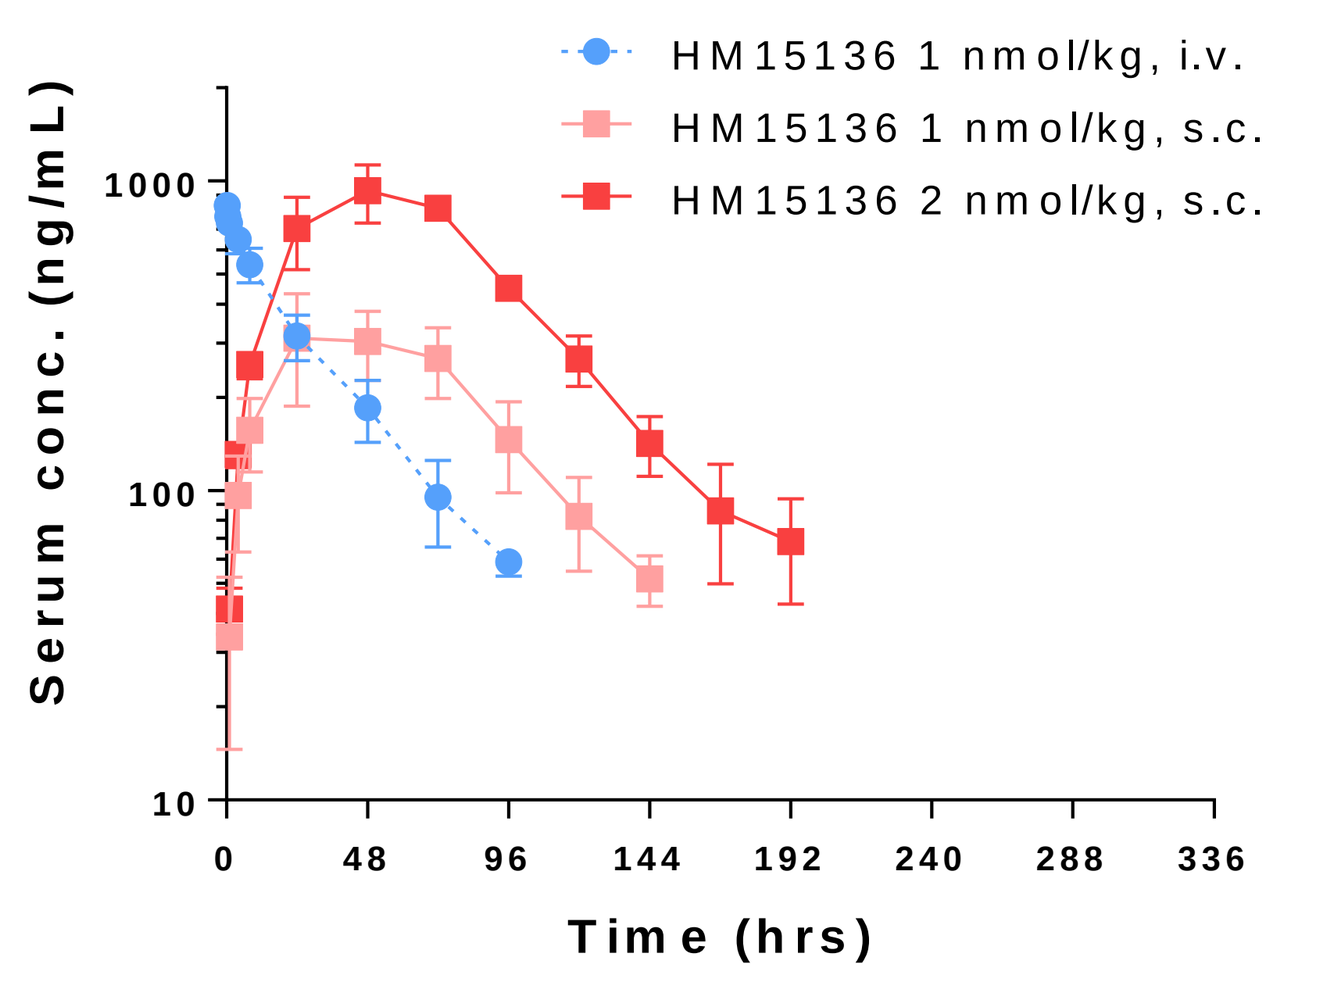
HM15136 1 nmol/kg, i.v.
HM15136 1 nmol/kg, s.c.
HM15136 2 nmol/kg, s.c.
Serum conc. (ng/mL)
Time (hrs)

(d) Human (prediction)

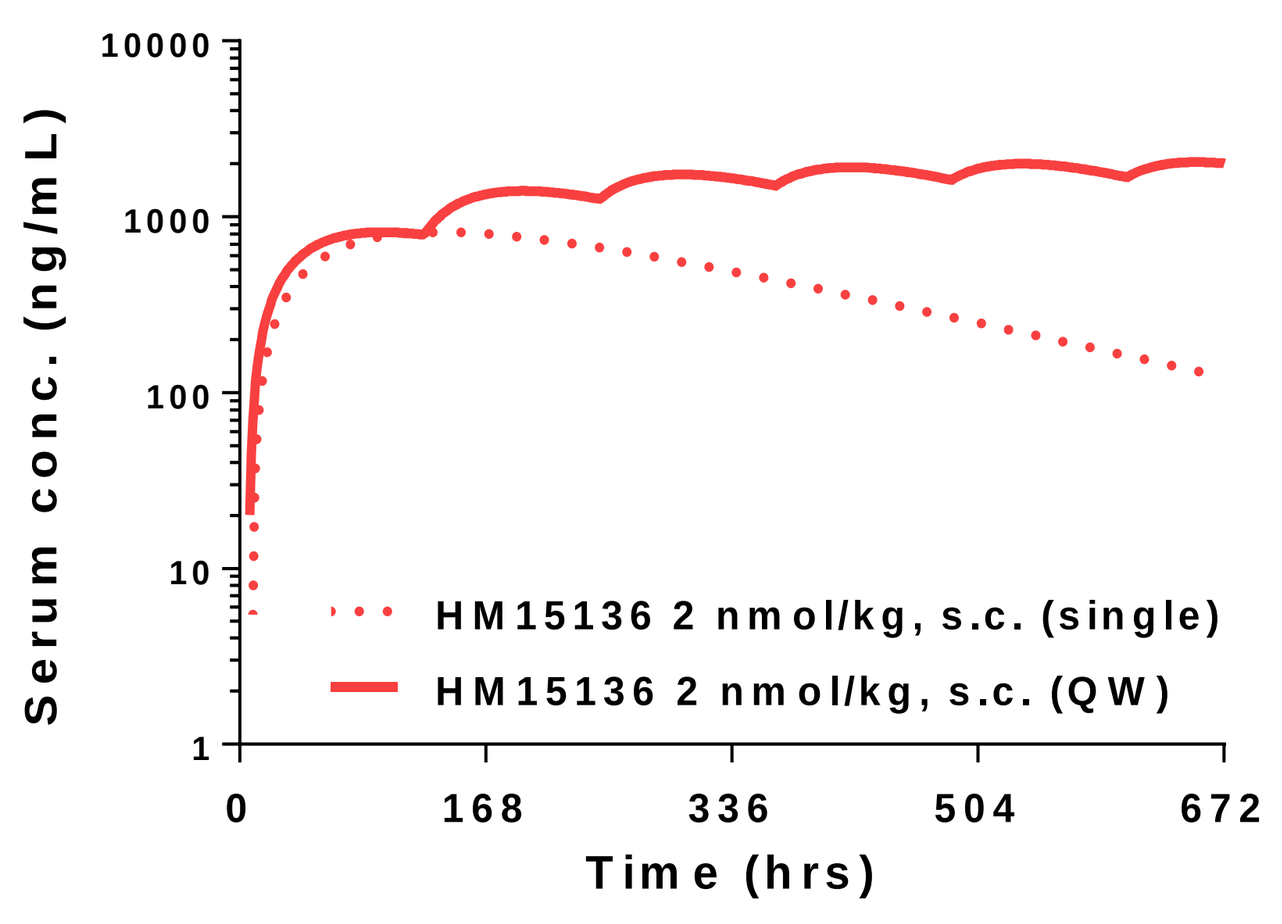
HM15136 2 nmol/kg, s.c. (single)
HM15136 2 nmol/kg, s.c. (QW)
Serum conc. (ng/mL)
Time (hrs)

Table 2. PK parameters of HM15136

	ICR mice	SD rats	Beagle dogs	Human (prediction)
T_{max} (hr)	24hr	48 ~ 64hr	32 ~ 64hr	129hr
$T_{1/2}$ (hr)	32.3 ~ 56.2hr	40.9 ~ 54.8hr	26.6 ~ 34.9hr	155.2hr
BA (%)	≥ 76.9%	≥ 89.2%	>100%	-

➢After LAPS-conjugation, HM15136 showed prolonged PK properties in various animal species. Simulated PK suggests the once-weekly dosing potential of HM15136 in human

In vivo efficacy in animal models of obesity

Figure 3. Effect of HM15136 on body weigh in DIO mice (n=7)

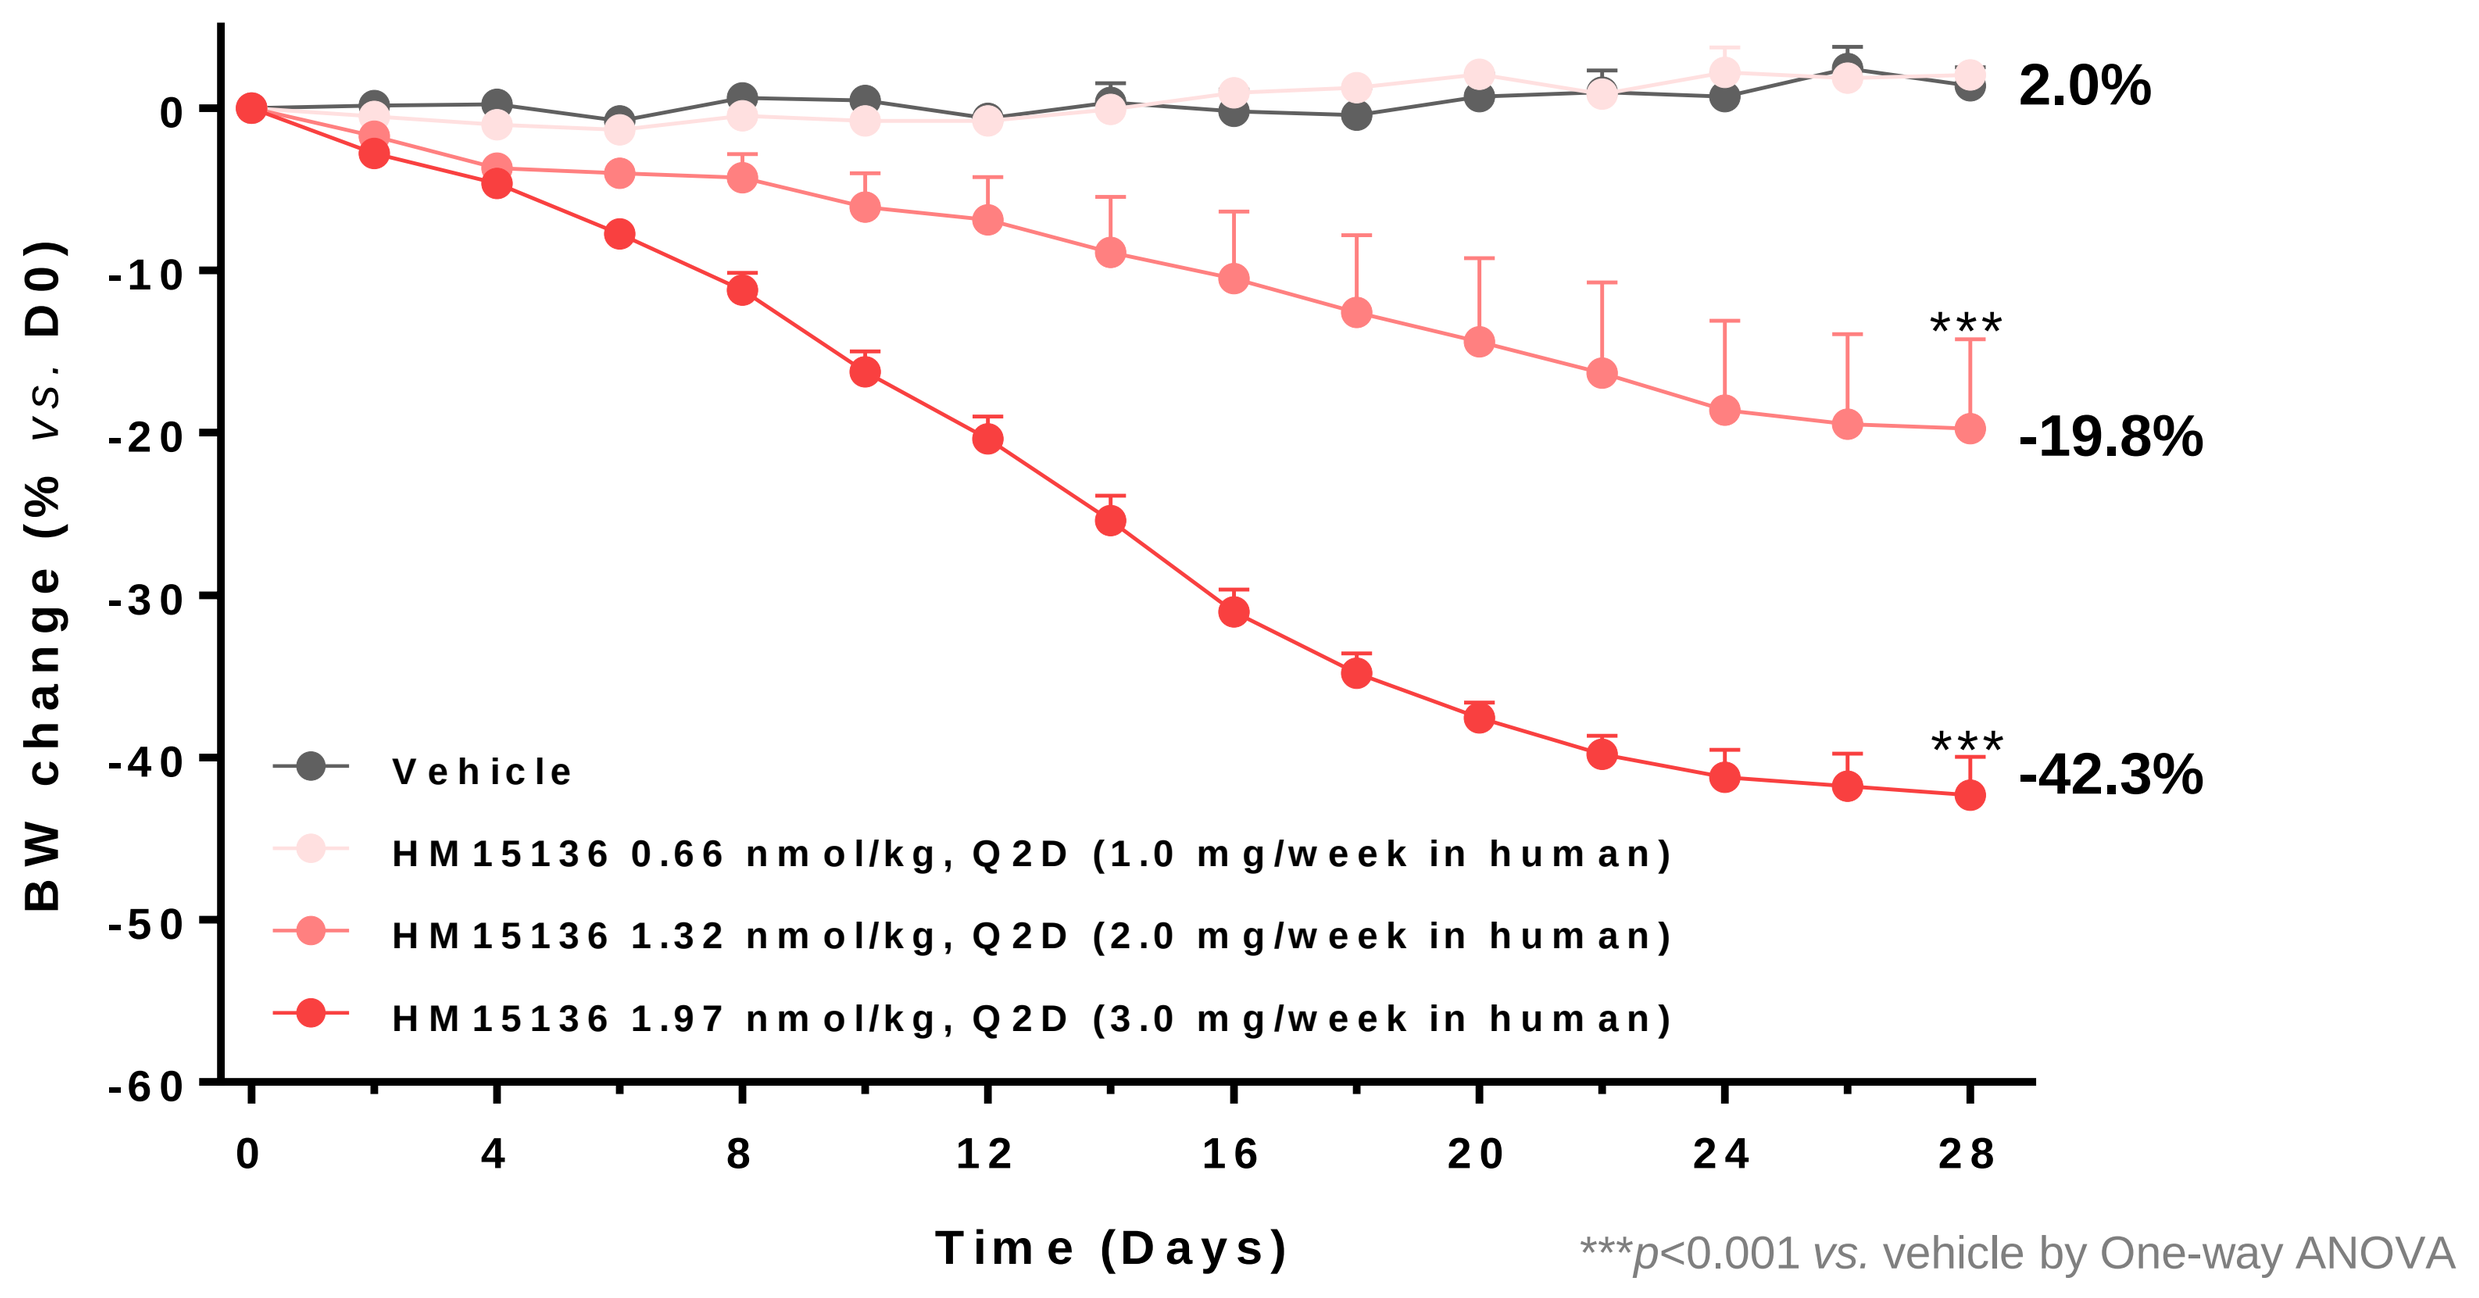
BW change (% vs. D0)
Time (Days)
Vehicle
HM15136 0.66 nmol/kg, Q2D (1.0 mg/week in human)
HM15136 1.32 nmol/kg, Q2D (2.0 mg/week in human)
HM15136 1.97 nmol/kg, Q2D (3.0 mg/week in human)
2.0%
-19.8%
-42.3%

***p<0.001 *vs.* vehicle by One-way ANOVA

➢Chronic treatment of HM15136 led to dose-dependent BWL in DIO mice, demonstrating the anti-obese effect of HM15136

Figure 4. Efficacy of HM15136 compared with liraglutide for BWL in DIO mice under with pair feeding (n=7)

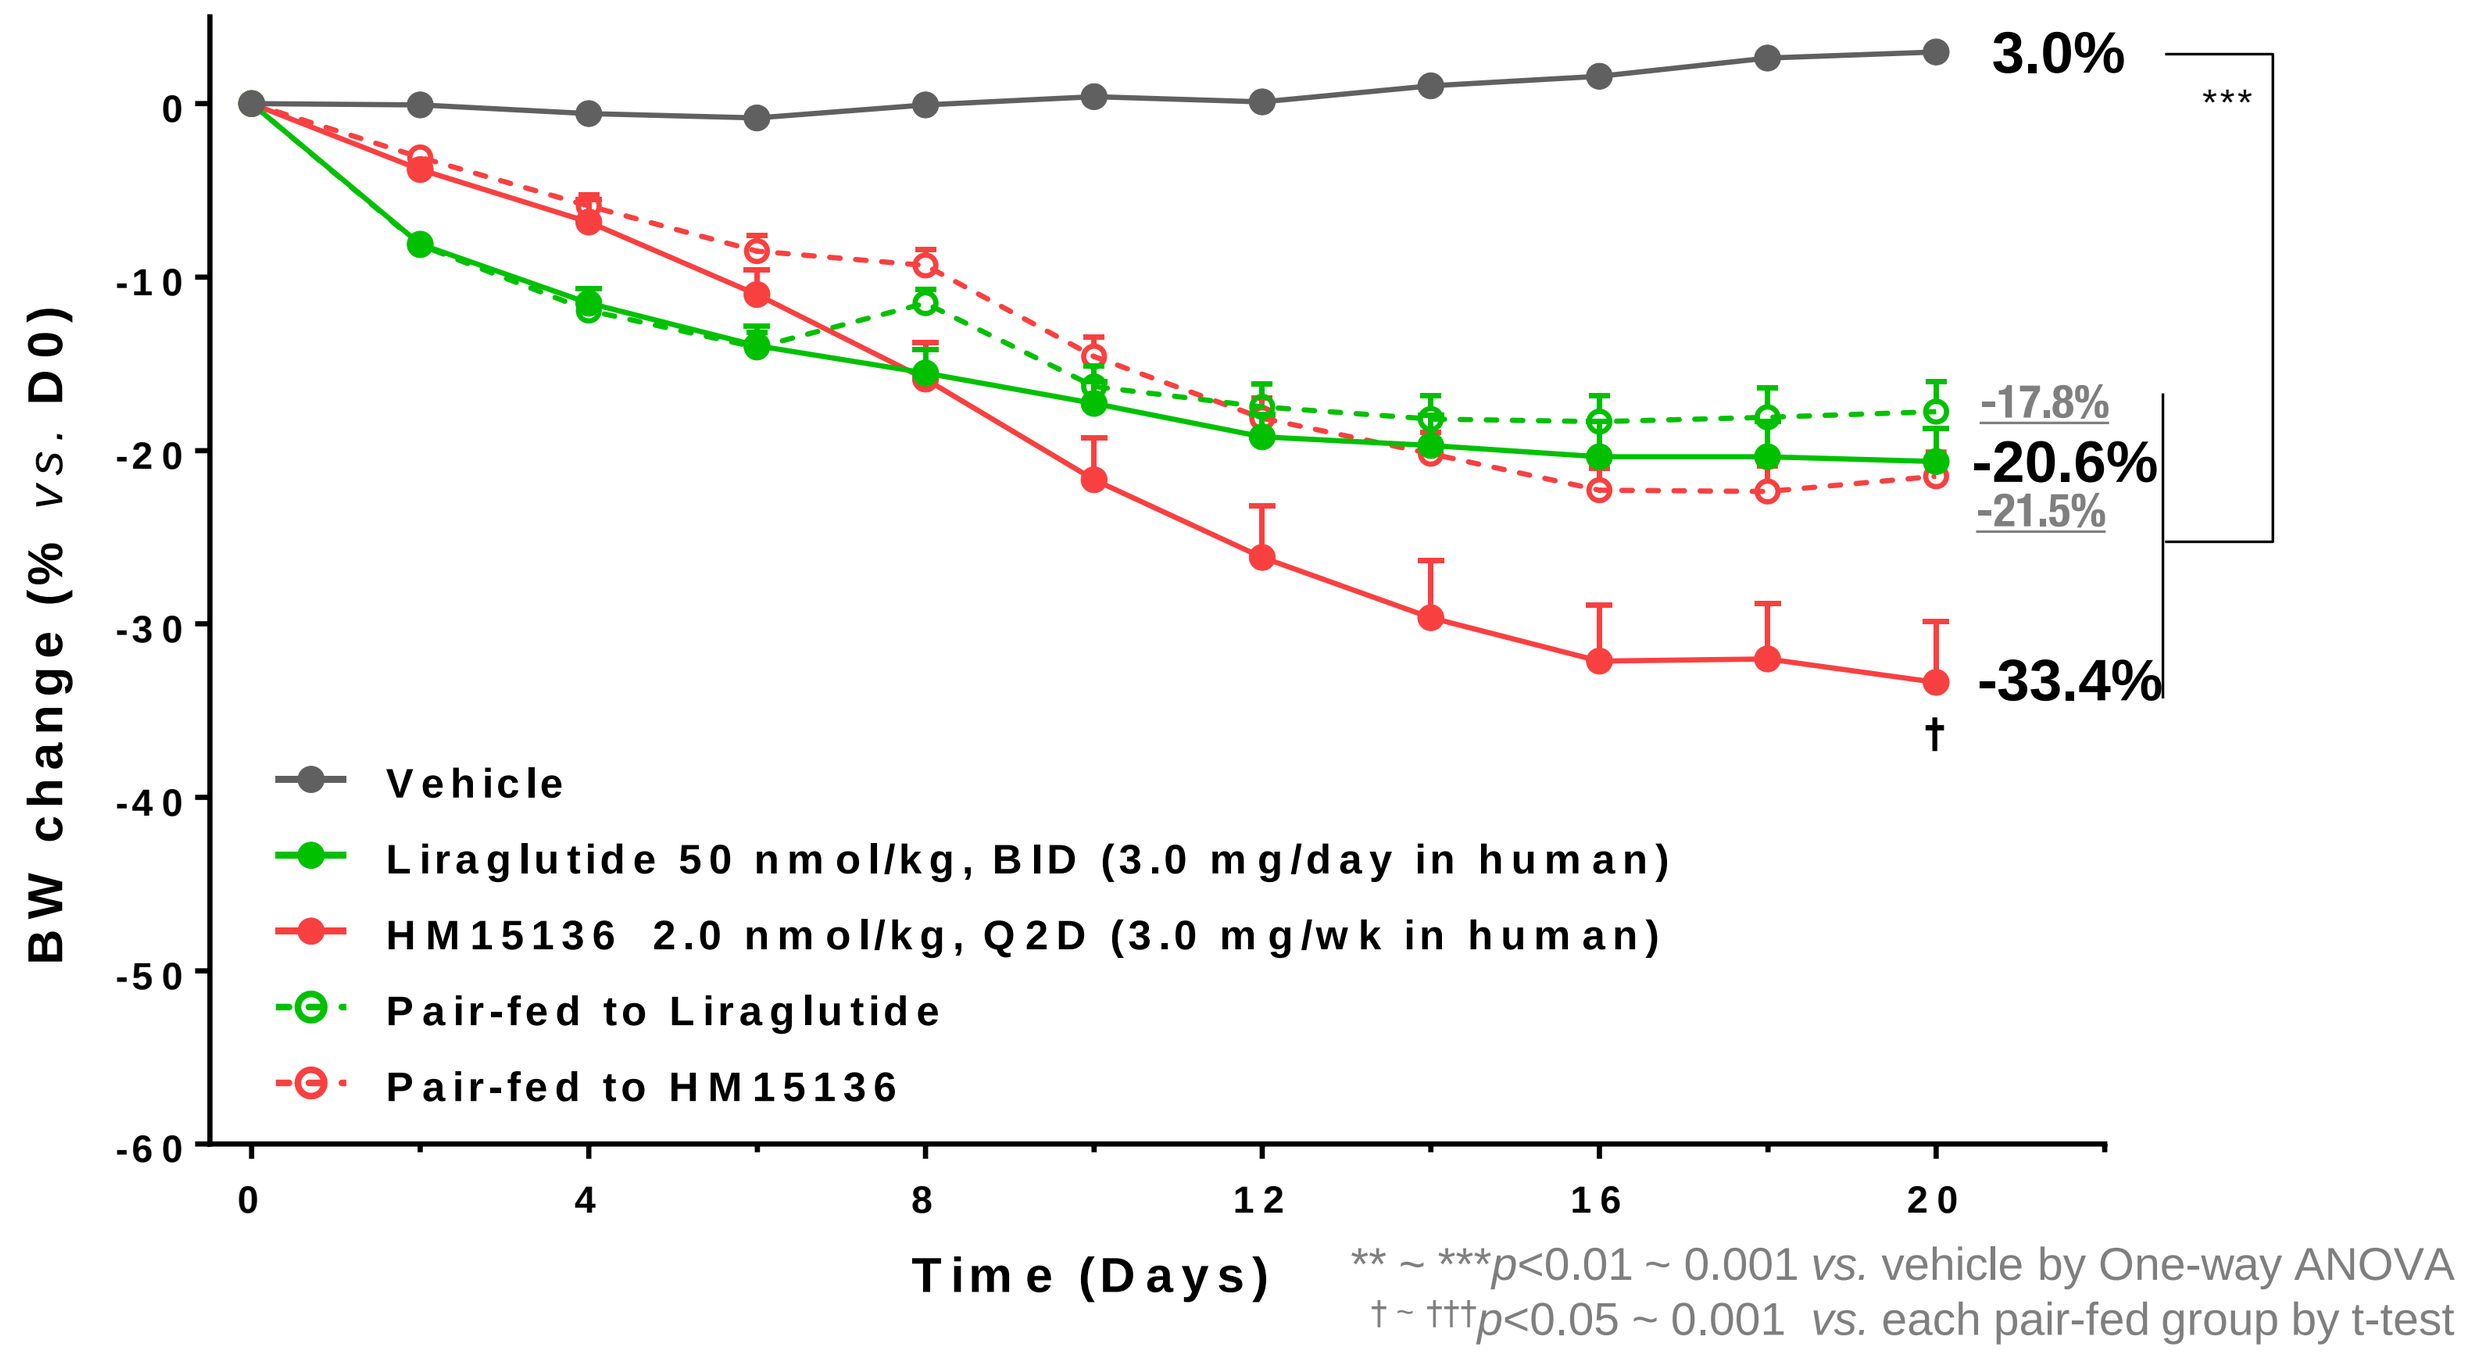
BW change (% vs. D0)
Time (Days)
Vehicle
Liraglutide 50 nmol/kg, BID (3.0 mg/day in human)
HM15136 2.0 nmol/kg, Q2D (3.0 mg/wk in human)
Pair-fed to Liraglutide
Pair-fed to HM15136
3.0%
-17.8%
-20.6%
-21.5%
-33.4%

†

** ~ ***p<0.01 ~ 0.001 *vs.* vehicle by One-way ANOVA
† ~ †††p<0.05 ~ 0.001 *vs.* each pair-fed group by t-test

➢ Unlike liraglutide, HM15136 provided more BWL than pair-fed group, suggesting satiety-independent BWL mechanism

Safety assessment: G/I tolerability

Figure 5. Effect of HM15136 on saccharin water preference in normal mice (n=6)

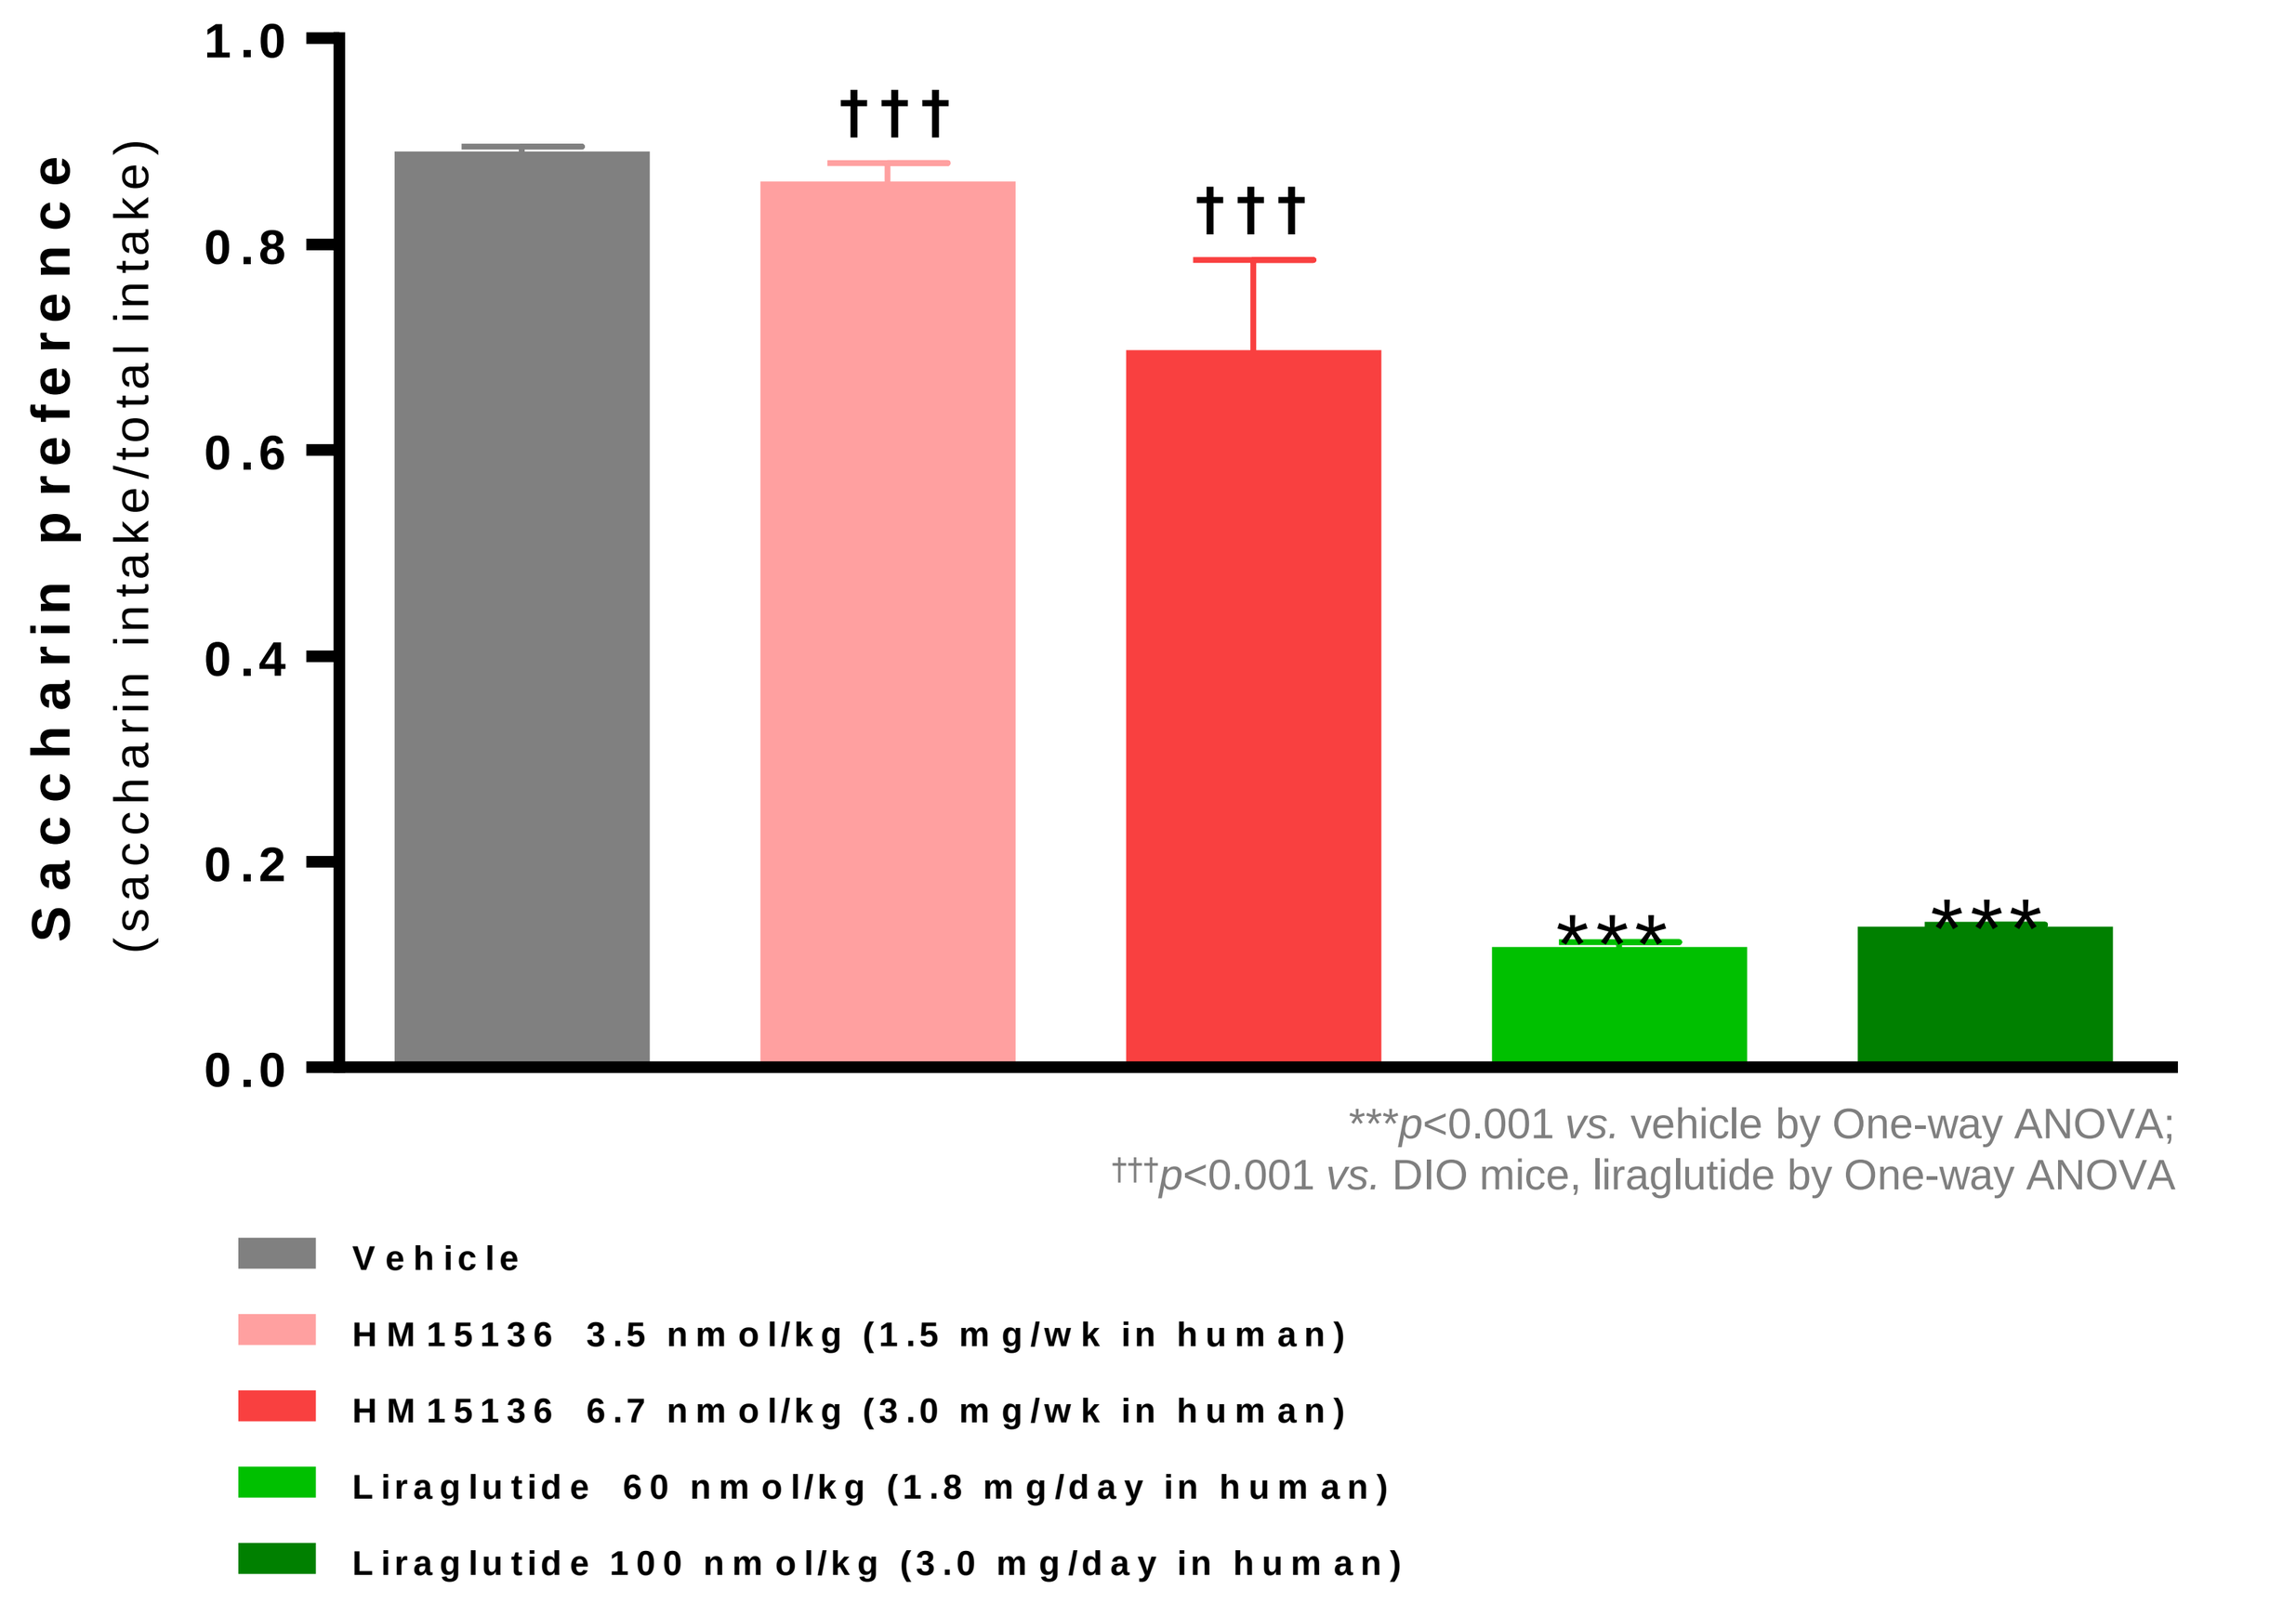
Saccharin preference
(saccharin intake/total intake)
†††
†††

Vehicle
HM15136 3.5 nmol/kg (1.5 mg/wk in human)
HM15136 6.7 nmol/kg (3.0 mg/wk in human)
Liraglutide 60 nmol/kg (1.8 mg/day in human)
Liraglutide 100 nmol/kg (3.0 mg/day in human)

***p<0.001 *vs.* vehicle by One-way ANOVA;
†††p<0.001 *vs.* DIO mice, liraglutide by One-way ANOVA

➢HM15136 showed more saccharin water preference than liraglutide, suggesting reduced nausea and vomiting risk

CONCLUSIONS

- **HM15136 is a novel long-acting glucagon analog with improved solubility and stability at physiological pH**
- **In DIO mice, HM15136 shows dose-dependent BWL. BG elevation is observed only during initial treatment phase, which is rapidly normalized (767-P)**
- **Unlike GLP-1RA, HM15136 shows food intake-independent BWL mechanism; WAT browning and enhanced energy expenditure might be involved (781-P)**
- **Compared to GLP-1RA, HM15136 shows more saccharin water preference in mice, suggesting mitigated nausea and vomiting incidence**
- **Therefore, HM15136 might be a novel therapeutic option for the treatment of obesity**

REFERENCES

- Pocai A *et al., Diabetes* **58**, 2258-66 (2009)
- Campbell JE and Drucker DJ., *Nat Rev Endocrinol.* **11**, 329-38 (2015)
- Muller TD *et al.*, *Physiol Rev.* **97**, 721-66 (2017)
- Kim T *et al.*, *Diabetes* **67**, 2157-66 (2018)

European Association for the Study of Diabetes (EASD) 55th Annual Meeting, Barcelona, Spain, 16 - 20 September 2019

Hanmi Pharm. Co., Ltd.